290 Tandem Meetings | Transplantation & Cellular Therapy Meetings of ASTCT™ & CIBMTR® | 15–19 February 2023 | Orlando, FL

Response to Lifileucel Tumor-Infiltrating Lymphocyte (TIL) Cell Therapy After ICI Resistance Regardless of Definition: An Analysis of the C-144-01 Trial in Patients With Advanced Melanoma

Amod Sarnaik,[1] Jason Chesney,[2] Theresa Medina,[3] Omid Hamid,[4] Sajeve Thomas,[5] Martin Wermke,[6] Viktoria Gontcharova,[7] Behzad Damirchi,[7] Joseph Dean,[7] Emma Masteller,[7] Xiao Wu,[7] Wen Shi,[7] Harriet Kluger[8]

[1]H. Lee Moffitt Cancer Center, Tampa, FL, USA; [2]UofL Health – Brown Cancer Center, University of Louisville, Louisville, KY, USA; [3]University of Colorado Cancer Center—Anschutz Medical Campus, Aurora, CO, USA; [4]The Angeles Clinic and Research Institute, a Cedars–Sinai Affiliate, Los Angeles, CA, USA; [5]Orlando Health Cancer Institute, Orlando, FL, USA; [6]Technical University Dresden – NCT/UCC Early Clinical Trial Unit, Dresden, Germany; [7]Iovance Biotherapeutics Inc, San Carlos, CA, USA; [8]Yale University School of Medicine, Smilow Cancer Center, New Haven Hospital, New Haven, CT, USA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASTCT™ & CIBMTR® and the author of this poster
Corresponding Author: Wen Shi (wen.shi@iovance.com)

Background

- ICI are important cornerstones of the current standard of care for advanced melanoma; however, 40%–65%[1-3] of patients have disease that is primary resistant to ICI, and 30%–40%[3-5] of patients have secondary-resistant disease using varying definitions of resistance
- Lifileucel, an investigational autologous TIL cell therapy, demonstrated encouraging activity in 153 patients with advanced melanoma who progressed after ICI and targeted therapy, if appropriate, in the C-144-01 trial, with an ORR of 31.4%[6]
 - In a previous subanalysis of C-144-01, lifileucel produced an ORR of 31.3% in patients with disease primary refractory to anti–PD-1/PD-L1 therapy using the study definition[6]
- The SITC Immunotherapy Resistance Taskforce recently developed an expert-consensus definition of resistance to anti–PD-1/PD-L1[7]
- Given the distinct mechanisms of action of TIL cell therapy and ICI, we hypothesized that subgroups identified based on resistance to anti–PD-1/PD-L1, irrespective of definition, would have similar outcomes after lifileucel TIL cell therapy

Objective

- In this post hoc analysis of the Phase 2 prospective, multicenter C-144-01 trial, we investigated outcomes in patients with disease primary-resistant or primary-refractory to prior anti–PD-1/PD-L1 treatment, with a focus on the SITC definition of resistance to anti–PD-1/PD-L1
 - We also explored association of translational biology features with primary resistance to anti–PD-1/PD-L1

Methods

Figure 1. C-144-01 (NCT0236057) Study Design

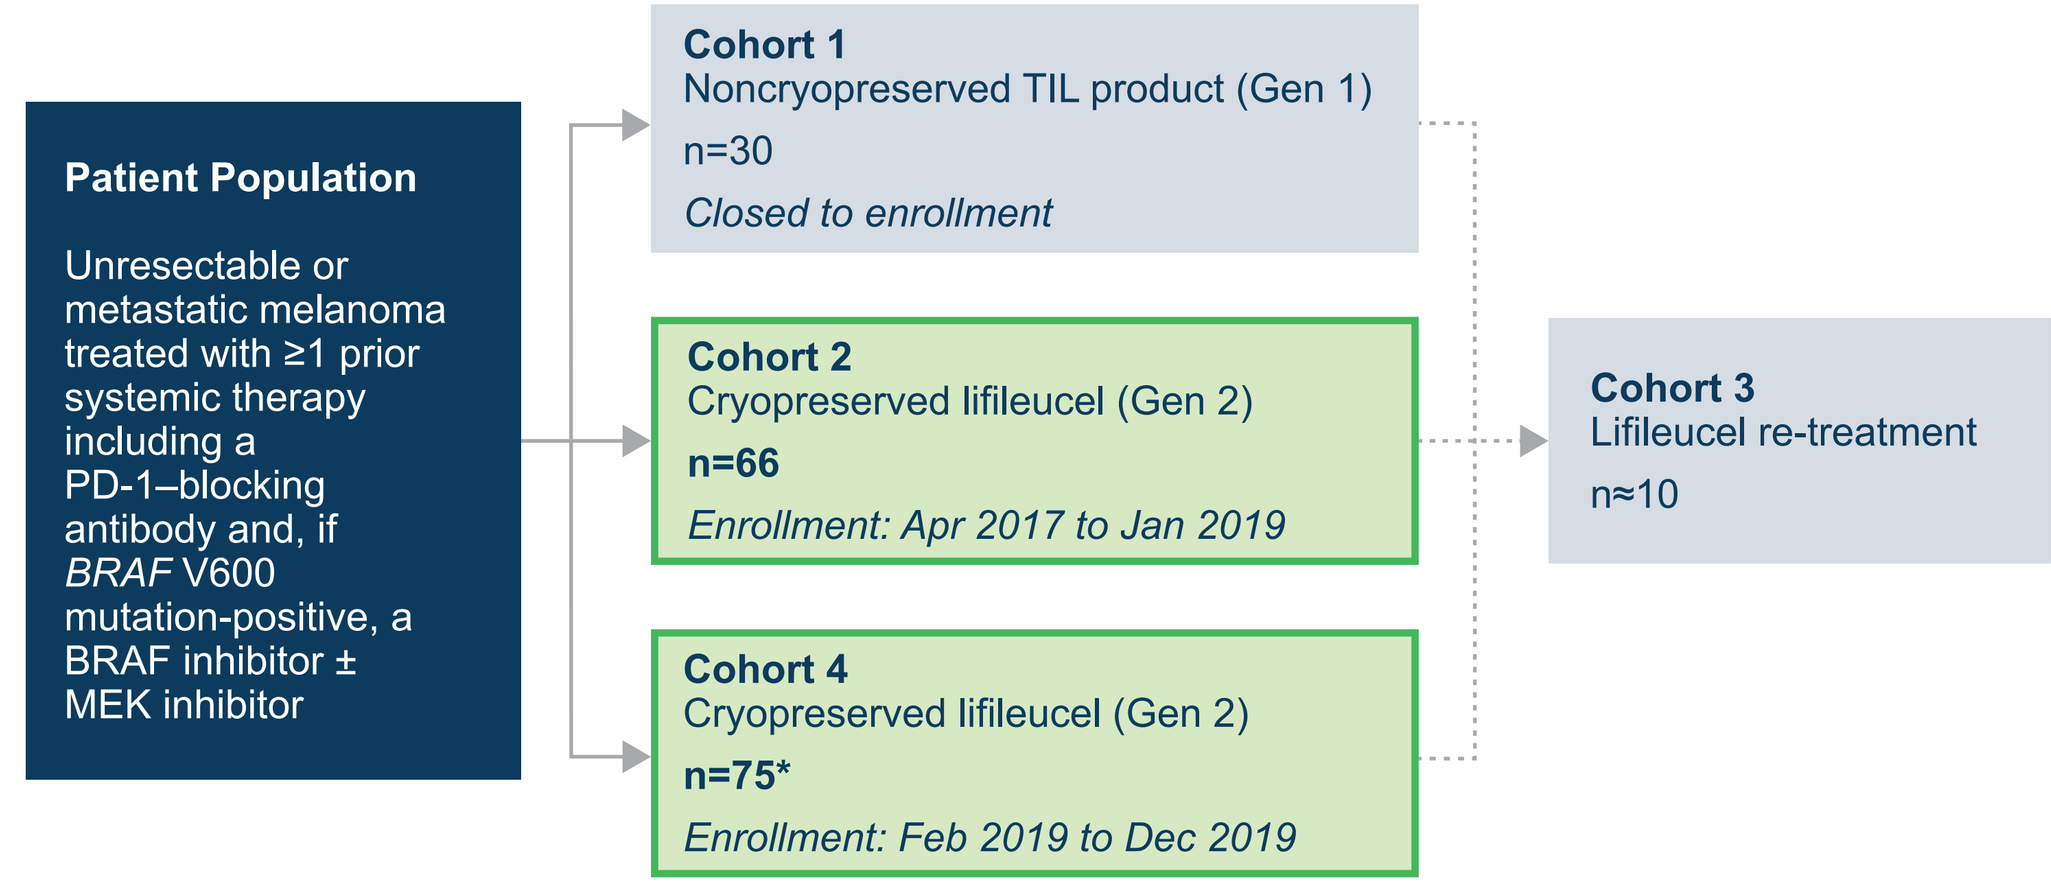
Patient Population
Unresectable or metastatic melanoma treated with ≥1 prior systemic therapy including a PD-1–blocking antibody and, if BRAF V600 mutation-positive, a BRAF inhibitor ± MEK inhibitor
Cohort 1
Noncryopreserved TIL product (Gen 1)
n=30
Closed to enrollment
Cohort 2
Cryopreserved lifileucel (Gen 2)
n=66
Enrollment: Apr 2017 to Jan 2019
Cohort 4
Cryopreserved lifileucel (Gen 2)
n=75*
Enrollment: Feb 2019 to Dec 2019
Cohort 3
Lifileucel re-treatment
n≈10

*The planned sample size for Cohort 4 was 75 per statistical plan, but the Full Analysis Set, defined as patients who received lifileucel that met specification, consisted of 87 patients due to rapid enrollment.

Key Endpoints

- Primary: ORR (IRC-assessed using RECIST v1.1)
- Secondary: DOR, PFS, OS, TEAE incidence and severity

Key Eligibility Criteria

- ≥1 tumor lesion resectable for TIL generation (≥1.5 cm in diameter) and ≥1 target tumor lesion for response assessment
- Age ≥18 years at time of consent
- ECOG performance status 0–1
- No limit on number of prior therapies

Treatment Regimen

- Lifileucel, a cryopreserved TIL cell therapy product, was used in Cohorts 2 and 4 and manufactured using the same 22-day Gen 2 process
- All patients received NMA-LD, a single lifileucel infusion, and up to 6 doses of high-dose IL-2

Data cutoff date: 15 July 2022

This post hoc analysis explores outcomes in patients from Cohorts 2 and 4 classified as primary resistant or primary refractory to anti–PD-1/PD-L1 therapy

Figure 2. Definitions of Primary Resistant/Refractory to Prior Anti–PD-1/PD-L1

SITC Taskforce Definition of "Primary Resistant"[7,*]

- Best overall response of progressive disease (or stable disease for <6 months) to prior anti–PD-1/PD-L1
 - ≥6 weeks of anti–PD-1/PD-L1 exposure
 - Confirmatory scan required ≥4 weeks after initial progression

C-144-01 Study Definition of "Primary Refractory"[6]

- Best overall response of progressive disease to prior anti–PD-1/PD-L1

*Definition for the advanced disease setting. Additional details available in Kluger H, et al.[7]

Application of the definitions

- Primary refractory
 - The first anti–PD-1/PD-L1 therapy with documented response was used in patients with multiline anti–PD-1/PD-L1
- Primary resistant
 - The first anti–PD-1/PD-L1 therapy in the metastatic setting was used in patients with multiline anti–PD-1/PD-L1; for patients who received only adjuvant anti–PD-1 therapy, early relapse was considered primary resistance
 - Because this analysis was performed retrospectively, criteria requiring confirmatory scans for progression could not be applied

Methods

Assessments

Clinical Assessments

- Response to lifileucel (ORR and DOR) was assessed by IRC (RECIST v1.1)

Translational Assessments

- Samples from 77 FFPE tumors resected for lifileucel manufacturing and 150 final TIL infusion products were available for testing

TCR Repertoire

- Analyzed using TCRvβ RNA sequencing data obtained from resected FFPE tumor, TIL infusion products, and PBMC
 - uCDR3 sequences (clonotypes): contribution to the total TCR repertoire
 - Simpson Clonality Index: values ranging from 0 (evenly distributed, polyclonal sample) to 1 (monoclonal sample)

TMB

- Measured using the ImmunoID NeXT Platform™ (Personalis®)
 - Whole exome sequencing
 - SNV and short indel calling: using Personalis®'s proprietary methods
 - TMB was calculated based on SNVs and indels

IFNγ Gene Signature

- A single gene set score for each gene set and patient calculated using the z-score method in the GSVA R package[8] (log_2[TPM counts + 1])

Tumor Mutations

- Somatic SNVs and CNAs were identified by Sentieon®, MuTect, VarDict, and Personalis® tools
- Gene variant effects were predicted using SnpEff[9]
- SNVs and CNAs were filtered by variant allelic frequency, effect on protein function, and clinical impact

Results

Table 1. Baseline Characteristics

Characteristic	Primary Resistant* (n=109)	Primary Refractory (n=83)	All Patients (N=153)
Median age (range), years	56.0 (20, 79)	55.0 (20,77)	56.0 (20, 79)
Sex, n (%)			
Male	63 (57.8)	48 (57.8)	83 (54.2)
Female	46 (42.2)	35 (42.2)	70 (45.8)
Screening ECOG performance status, n (%)			
0	75 (68.8)	57 (68.7)	104 (68.0)
1	34 (31.2)	26 (31.3)	49 (32.0)
Melanoma subtype,† n (%)			
Cutaneous	53 (48.6)	41 (49.4)	83 (54.2)
Mucosal	11 (10.1)	10 (12.0)	12 (7.8)
Acral	10 (9.2)	6 (7.2)	10 (6.5)
BRAF V600-mutated, n (%)	32 (29.4)	25 (30.1)	41 (26.8)
PD-L1 status,‡ n (%)			
TPS ≥1%	56 (51.4)	43 (51.8)	76 (49.7)
TPS <1%	22 (20.2)	18 (21.7)	32 (20.9)
Liver and/or brain lesions by IRC, n (%)	48 (44.0)	39 (47.0)	72 (47.1)
Median target lesion SOD (range), mm	100.4 (15.7, 552.9)	107.7 (15.7, 552.9)	101.1 (13.5, 552.9)
Baseline lesions in ≥3 anatomic sites, n (%)	75 (68.8)	60 (72.3)	109 (71.2)
Baseline target and nontarget lesions,§ n (%)			
>3	79 (72.5)	63 (75.9)	116 (75.8)
LDH, n (%)			
≤ULN	48 (44.0)	40 (48.2)	70 (45.8)
1–2 × ULN	38 (34.9)	27 (32.5)	54 (35.3)
>2 × ULN	23 (21.1)	16 (19.3)	29 (19.0)
Median number of prior therapies (range)	3.0 (1, 8)	3.0 (1, 8)	3.0 (1, 9)

*Using SITC Taskforce Criteria.[7]
†In the overall population, 47 patients (31%) had melanoma of other subtype (including unknown primary subtype or insufficient information).
‡In the overall population, 45 patients had missing PD-L1 status.
§One patient in the overall population had missing data on number of baseline target and nontarget lesions.

- Except for 1 patient, all primary refractory patients were categorized as primary resistant per SITC definition
- Baseline patient and disease characteristics were generally similar between the primary-resistant and primary-refractory groups and the overall population (**Table 1**)
- Patients were heavily pretreated and had high tumor burden at baseline

Table 2. Efficacy Outcomes

Characteristic	Primary Resistant* (n=109)	Primary Refractory (n=83)	All Patients (N=153)
ORR, n (%)	**36 (33.0)**	**26 (31.3)**	**48 (31.4)**
(95% CI)	(24.3, 42.7)	(21.6, 42.4)	(24.1, 39.4)
Best overall response, n (%)			
CR	8 (7.3)	7 (8.4)	9 (5.9)
PR	28 (25.7)	19 (22.9)	39 (25.5)
SD	47 (43.1)	39 (47.0)	71 (46.4)
Non-CR/non-PD†	1 (0.9)	1 (1.2)	1 (0.7)
PD	19 (17.4)	13 (15.7)	27 (17.6)
Nonevaluable‡	6 (5.5)	4 (4.8)	6 (3.9)

*Using SITC Taskforce Criteria.[7]
†Patient did not have measurable target lesions by IRC and had best overall response of non-CR/non-PD per IRC assessment.
‡Six patients were nonevaluable for response (5 due to early death; 1 due to new anticancer therapy).

- ORR and BOR were comparable between the primary-resistant and primary-refractory groups, and the overall population (**Table 2**)
- Further analyses in this presentation focus on the primary-resistant population as defined by the SITC Taskforce criteria[7]

Results

Figure 3. Tumor Burden Reduction and Best Response to Lifileucel, by Primary Resistance to Anti–PD-1/PD-L1*

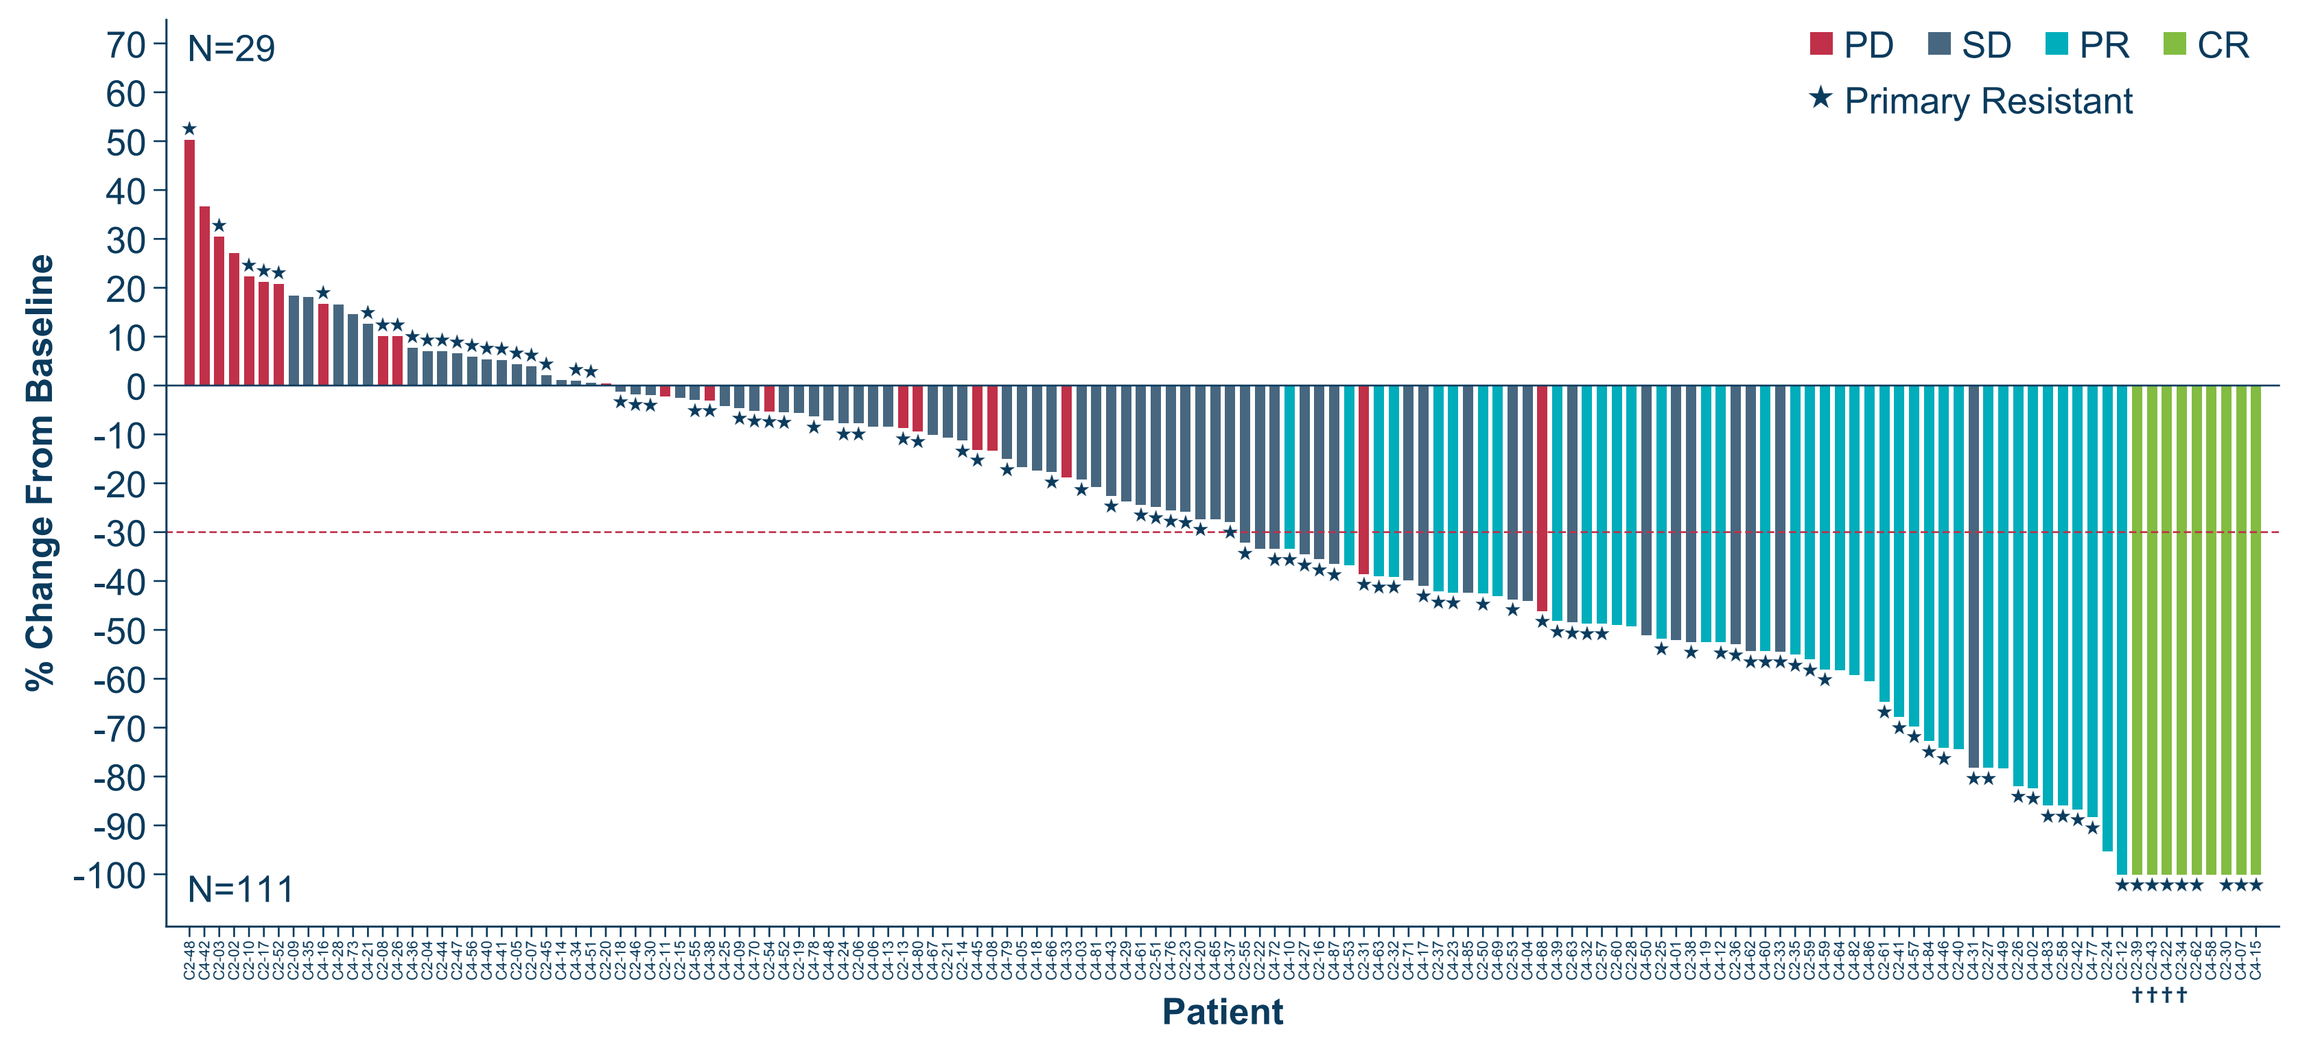
PD
SD
PR
CR
★ Primary Resistant
N=29
N=111
% Change From Baseline
Patient

*Using SITC Taskforce Criteria.[7]
†-100% change from baseline is presented for CR assessment that includes lymph node lesions.
13 patients in the Full Analysis Set are not included (best overall responses included NE [n=6], non-CR/non-PD [n=1], and PD [n=6]) for reasons including having no measurable lesions at baseline or no post-lifileucel target lesion SOD measurements.

- 78.6% (77/98) of patients primary resistant to anti–PD-1/PD-L1 and 79.3% (111/140) of all patients had a reduction in tumor burden (**Figure 3**)

Figure 4. Time to Response, DOR, and Time on Efficacy Assessment for Confirmed Responders (PR or Better), by Primary Resistance to Anti–PD-1/PD-L1*

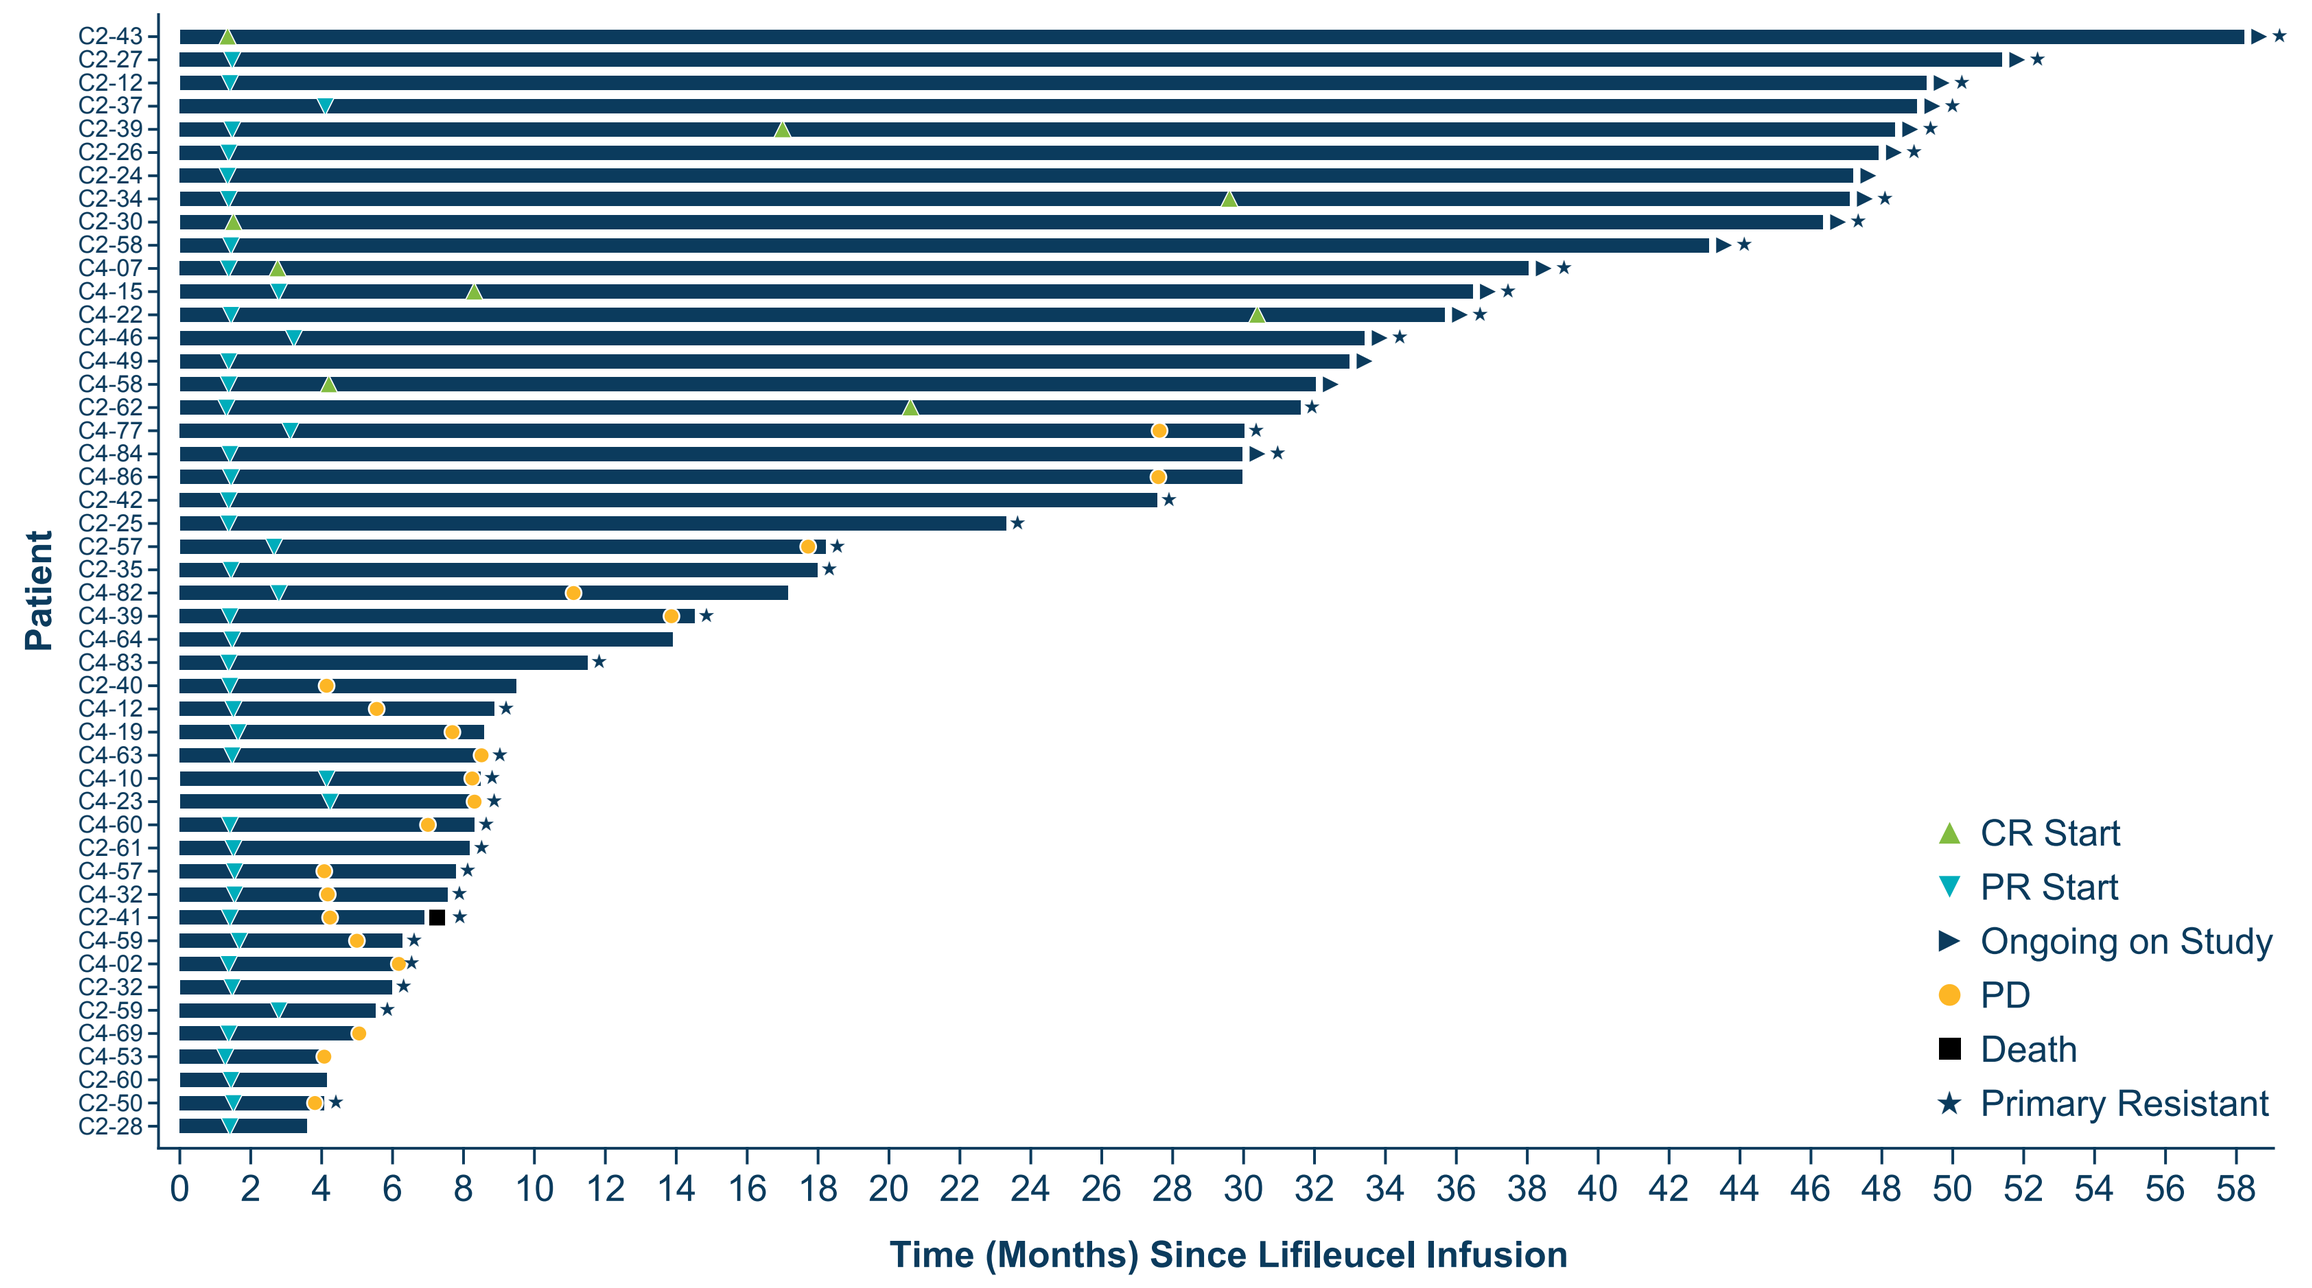
CR Start
PR Start
Ongoing on Study
PD
Death
★ Primary Resistant
Patient
Time (Months) Since Lifileucel Infusion

*Using SITC Taskforce Criteria.[7]

- 38.9% of responses in primary-resistant patients and 35.4% of responses in all patients were ongoing as of the data cutoff (**Figure 4**)

Table 3. DOR, by Primary Resistance to Anti–PD-1/PD-L1*

	Primary Resistant* (n=36)†	All Patients (N=48)†
Median DOR,‡ months	**NR**	**NR**
95% CI	(12.5, NR)	(8.3, NR)
Min, max (months)	1.4+, 54.1+	1.4+, 54.1+
DOR ≥12 months, n (%)	21 (58.3)	26 (54.2)
DOR ≥24 months, n (%)	16 (44.4)	20 (41.7)
Median study follow-up, months	40.1	36.5

*Using SITC Taskforce Criteria.[7]
†Includes sample size of responders.
‡Based on Kaplan-Meier estimate.

- At a median study follow up of 40.1 months, median DOR was not reached in primary-resistant patients (**Table 3**)
- Ongoing responses at ≥24 months were comparable between the primary-resistant patients and the overall population

Table 4. OS, by Primary Resistance to Anti–PD-1/PD-L1*

	Primary Resistant* (n=109)	All Patients (N=153)
Median OS,† months	14.1	13.9
95% CI	(9.7, 18.3)	(10.6, 17.8)
OS at 12 months (%)	53.3	54.0
95% CI	(43.4, 62.2)	(45.6, 61.6)

*Using SITC Taskforce Criteria.[7]
†Based on Kaplan-Meier estimate.

Table 5. Safety, by Primary Resistance to Anti–PD-1/PD-L1*

Non-Hematologic TEAEs in ≥30% of Patients (Either Group)†,‡

Preferred Term, n (%)	Primary Resistant* (n=111) Any Grade	Primary Resistant* (n=111) Grade 3/4	All Patients (N=156) Any Grade	All Patients (N=156) Grade 3/4
Chills	84 (75.7)	5 (4.5)	117 (75.0)	8 (5.1)
Pyrexia	54 (48.6)	10 (9.0)	81 (51.9)	17 (10.9)
Febrile neutropenia	48 (43.2)	48 (43.2)	65 (41.7)	65 (41.7)
Hypophosphatemia	42 (37.8)	28 (25.2)	58 (37.2)	41 (26.3)
Hypotension	42 (37.8)	16 (14.4)	52 (33.3)	17 (10.9)
Fatigue	39 (35.1)	5 (4.5)	51 (32.7)	6 (3.8)
Diarrhea	35 (31.5)	2 (1.8)	48 (30.8)	2 (1.3)

Grade 3/4 Hematologic Lab Abnormalities†

Preferred Term, n (%)	Primary Resistant* (n=111) Grade 3/4	All Patients (N=156) Grade 3/4
Leukopenia	111 (100)	156 (100)
Lymphopenia	111 (100)	156 (100)
Neutropenia	111 (100)	156 (100)
Thrombocytopenia	103 (92.8)	147 (94.2)
Anemia	81 (73.0)	111 (71.2)

*Using SITC Taskforce Criteria.[7]
†Per CTCAE v4.03; Safety Analysis Set.
‡Grade 5 TEAEs included pneumonia (n=1), acute respiratory failure (n=1), arrhythmia (n=1), and intra-abdominal hemorrhage (n=1).

- Median number of IL-2 doses administered was 6 in both primary-resistant and all patients
- TEAEs were consistent with known safety profiles of NMA-LD and IL-2 and were similar in primary-resistant and all patients (**Table 5**)

Figure 5. TMB, by Primary Resistance to Anti–PD-1/PD-L1*

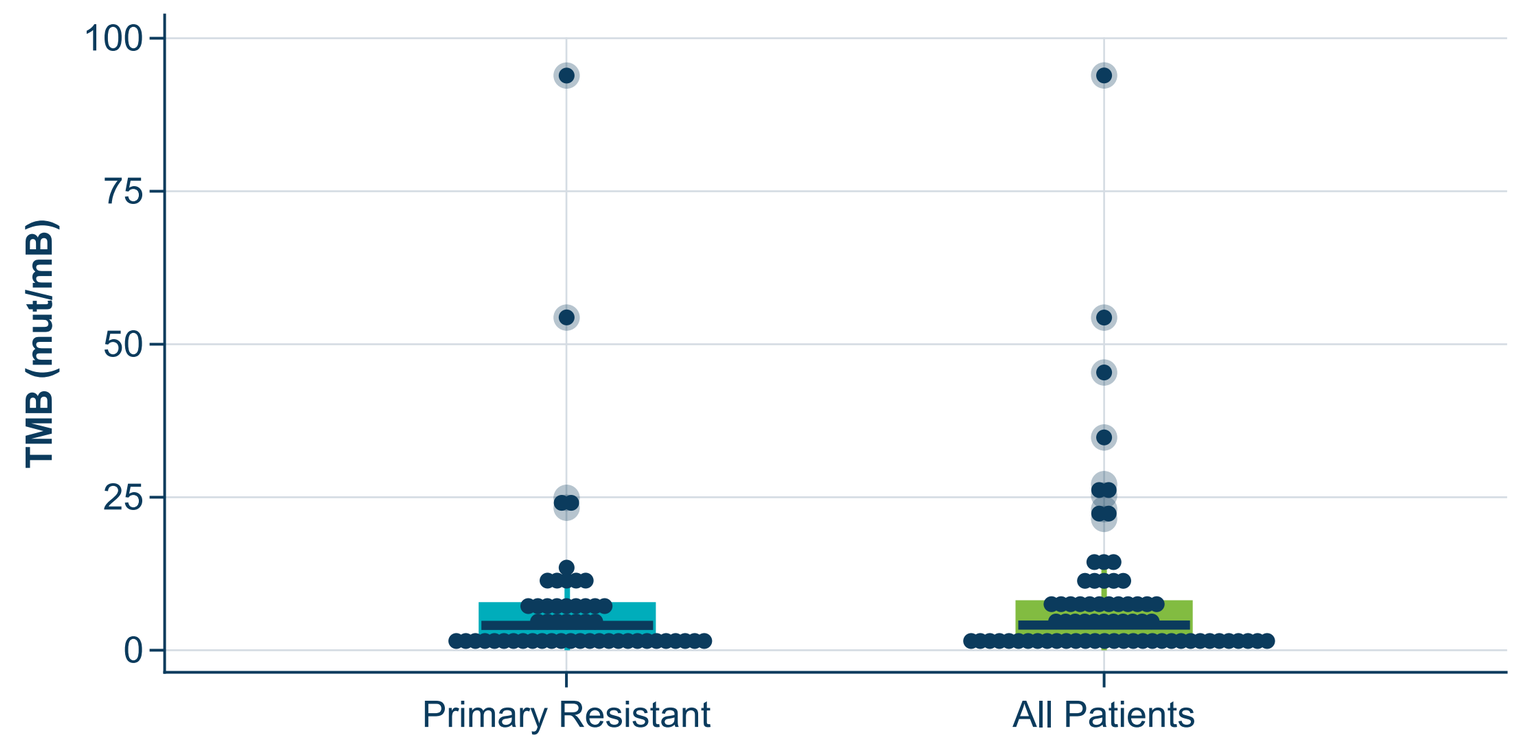
TMB (mut/mB)
Primary Resistant
All Patients

*Using SITC Taskforce Criteria.[7]

- TMB was similar regardless of primary resistance to anti–PD-1/PD-L1 (**Figure 5**)

Figure 6. Tumor Mutations, by Primary Resistance to Anti–PD-1/PD-L1*

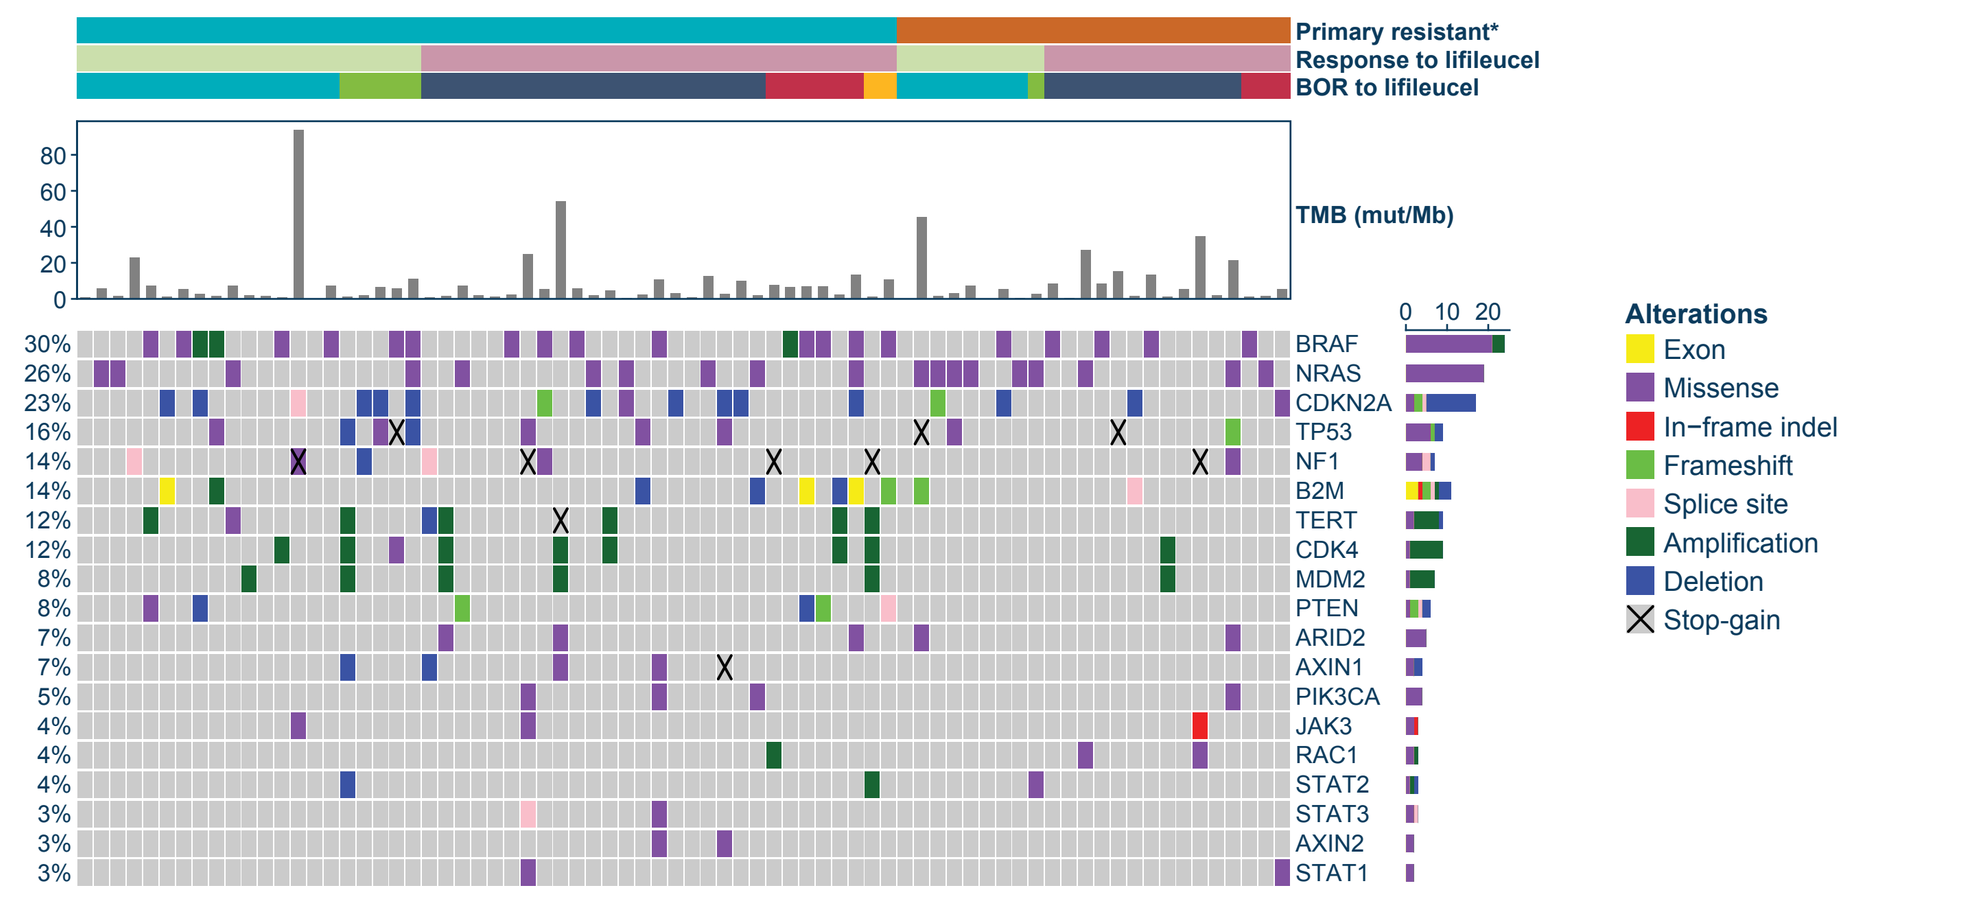
Primary resistant*
Response to lifileucel
BOR to lifileucel
TMB (mut/Mb)
BRAF
NRAS
CDKN2A
TP53
NF1
B2M
TERT
CDK4
MDM2
PTEN
ARID2
AXIN1
PIK3CA
JAK3
RAC1
STAT2
STAT3
AXIN2
STAT1
Alterations
Exon
Missense
In−frame indel
Frameshift
Splice site
Amplification
Deletion
Stop-gain
Primary resistant*
No
Yes
Response to lifileucel
Non−responder
Responder
BOR
CR
PR
SD
PD
NE

*Using SITC Taskforce Criteria.[7]

- No pattern was observed in tumor mutations in primary-resistant patients (**Figure 6**)

Figure 7. IFN-γ Gene Signature, by Primary Resistance to Anti–PD-1/PD-L1*

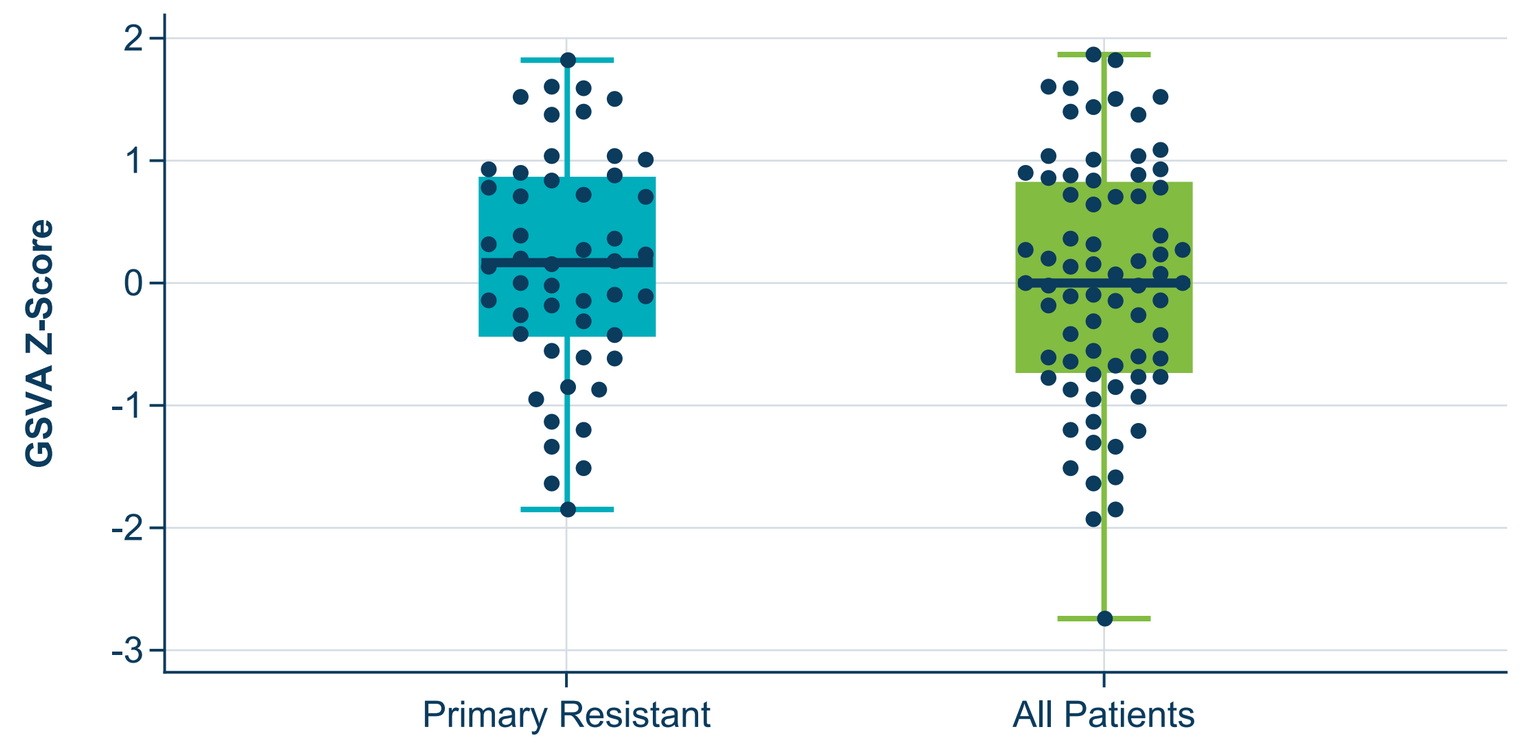
GSVA Z-Score
Primary Resistant
All Patients

*Using SITC Taskforce Criteria.[7]

- IFN-γ gene expression signature[10] was similar between the primary-resistant population and all patients (**Figure 7**)
- Other immune-related gene signatures were also analyzed, but no trend was observed

Figure 8. TCR Clonality, by Primary Resistance to Anti–PD-1/PD-L1*

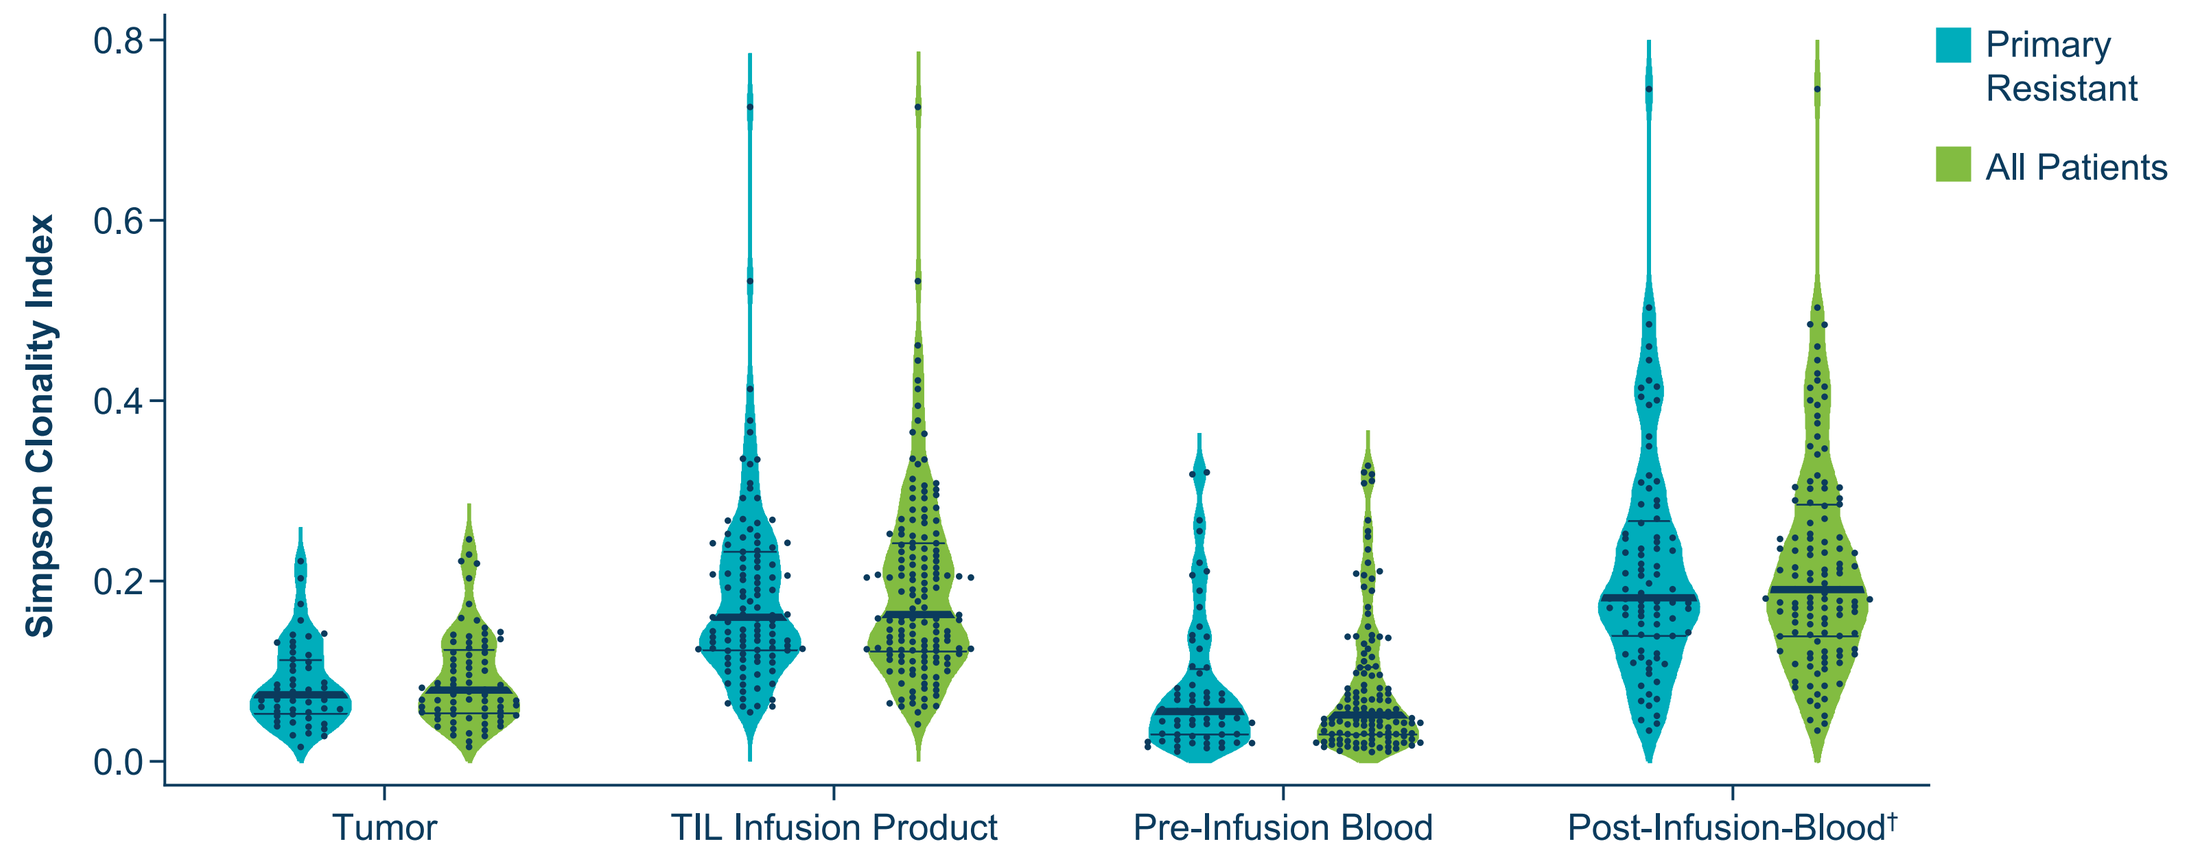
Primary Resistant
All Patients
Simpson Clonality Index
Tumor
TIL Infusion Product
Pre-Infusion Blood
Post-Infusion-Blood†

*Using SITC Taskforce Criteria.[7]
†Day 42 visit.

- The primary-resistant population had similar TCR clonality as the overall population in all samples assessed (**Figure 8**)

Figure 9. TCR Clonal Expansion and Persistence After Lifileucel Infusion, by Primary Resistance to Anti–PD-1/PD-L1*

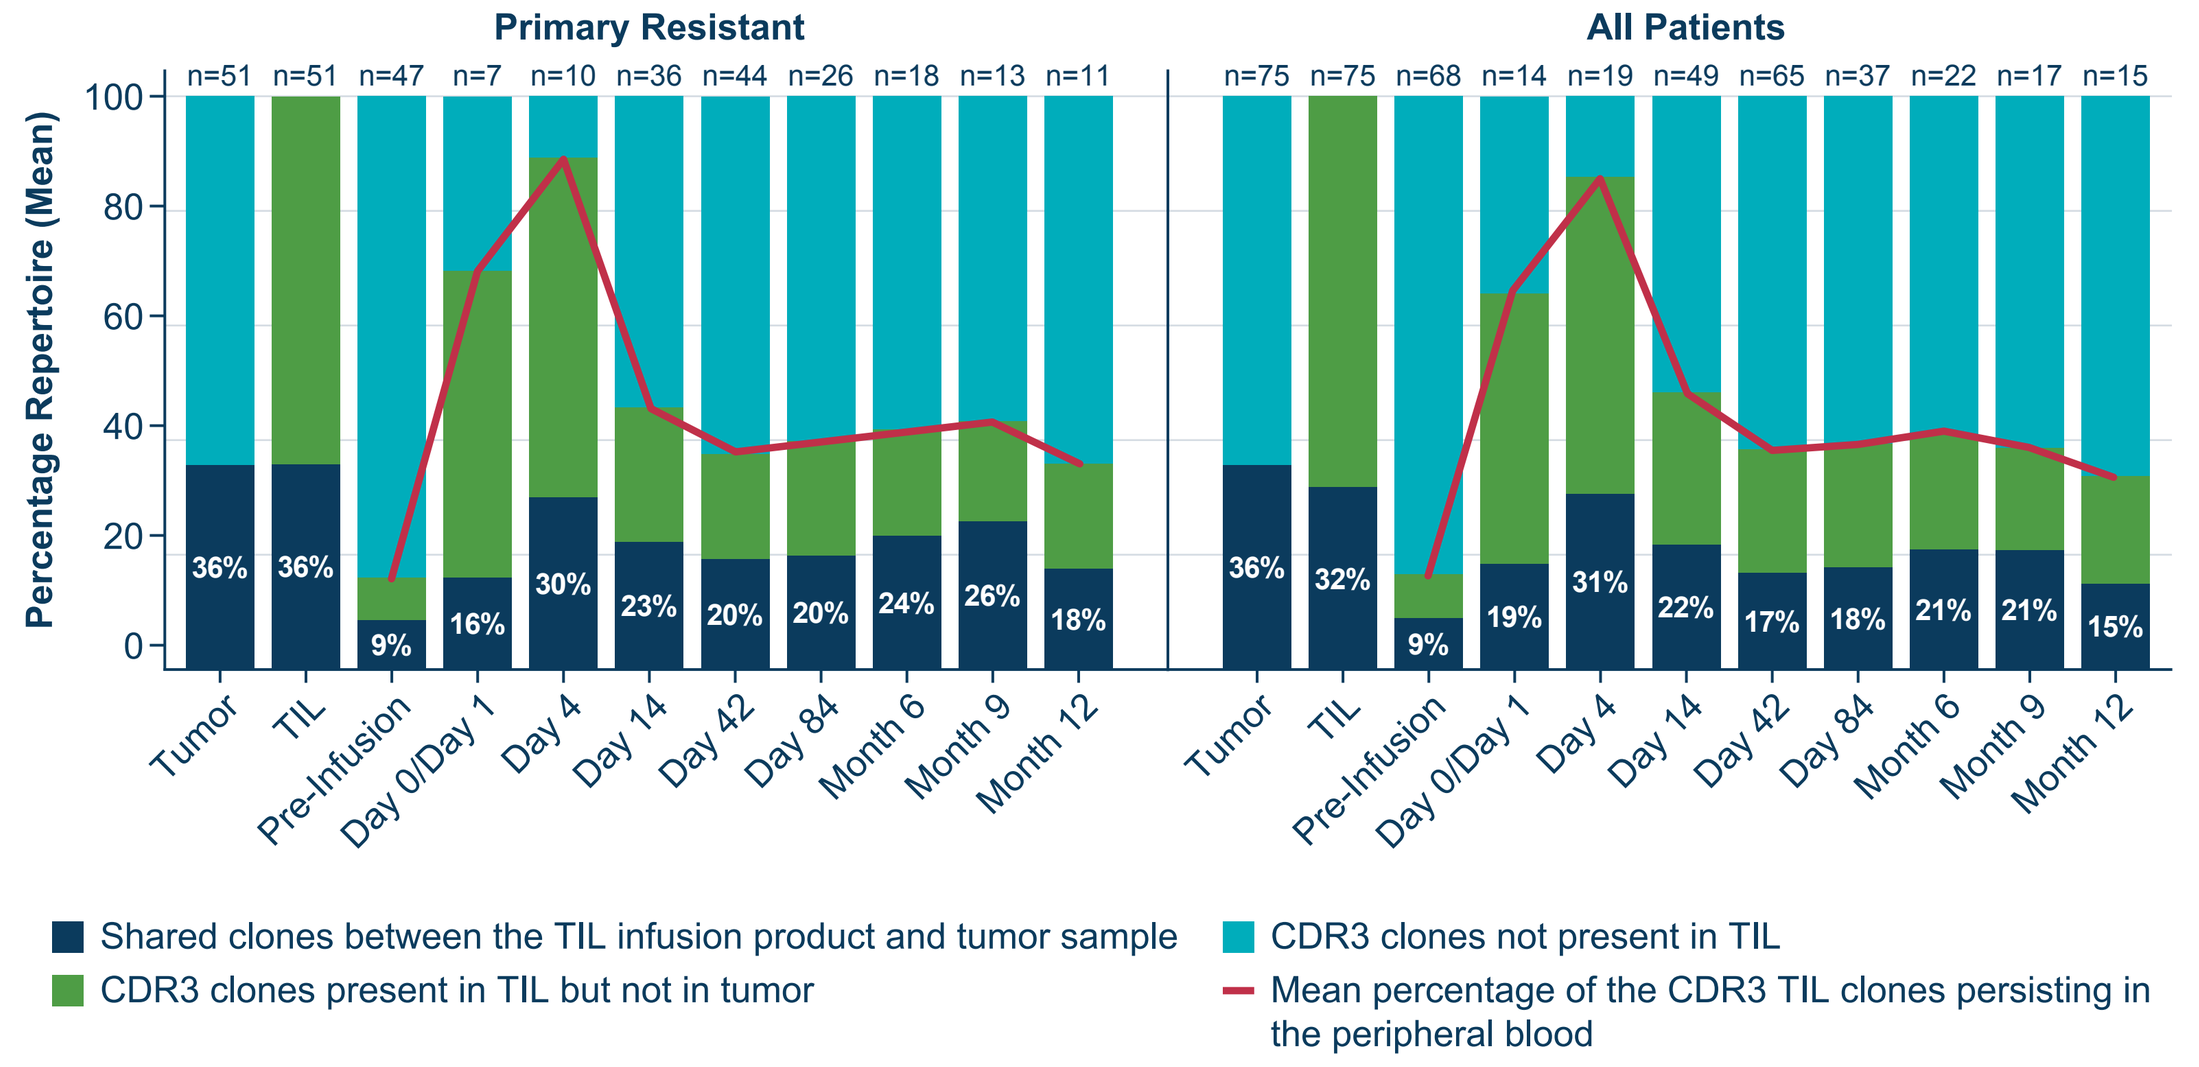
Primary Resistant
All Patients
Percentage Repertoire (Mean)
n=51 n=51 n=47 n=7 n=10 n=36 n=44 n=26 n=18 n=13 n=11
n=75 n=75 n=68 n=14 n=19 n=49 n=65 n=37 n=22 n=17 n=15
36% 36% 9% 16% 30% 23% 20% 20% 24% 26% 18%
36% 32% 9% 19% 31% 22% 17% 18% 21% 21% 15%
Tumor
TIL
Pre-Infusion
Day 0/Day 1
Day 4
Day 14
Day 42
Day 84
Month 6
Month 9
Month 12
Shared clones between the TIL infusion product and tumor sample
CDR3 clones present in TIL but not in tumor
CDR3 clones not present in TIL
Mean percentage of the CDR3 TIL clones persisting in the peripheral blood

*Using SITC Taskforce Criteria.[7]

- TIL clones expanded and persisted to a similar degree regardless of primary resistance to anti–PD-1/PD-L1 (**Figure 9**)

Conclusions

- In patients with advanced melanoma and prior anti–PD-1/PD-L1 therapy, response to lifileucel was not associated with primary resistance to anti–PD-1/PD-L1, regardless of definition
 - Efficacy in primary-resistant patients was clinically meaningful and durable, similar to that in the overall study population
 - Safety profile in patients with anti–PD-1/PD-L1 primary-resistant disease was expected and manageable and did not differ from that of the overall study population
- Translational analyses of tumor (TMB, tumor mutations, and immune-related gene signatures) and TIL infusion product (clonality, expansion, and persistence) did not reveal unique biological profiles of primary resistance
- These data are consistent with the observed potential benefit of lifileucel treatment across a broad spectrum of patients with melanoma that progressed on or after standard-of-care frontline therapy

References

1. Long GV, et al. *Clin Cancer Res*. 2021;27(19):5280-5288.
2. Larkin J, et al. *N Engl J Med*. 2015;373(1):23-34.
3. Mooradian MJ, et al. *Oncology (Williston Park)*. 2019;33(4):141-148.
4. Hamid O, et al. *Ann Oncol*. 2019;30(4):582-588.
5. Wolchok JD, et al. *N Engl J Med*. 2017;377(14):1345-1356.
6. Chesney JA, et al. *J ImmunoTher Cancer*. 2022;10:e005755.
7. Kluger H, et al. *J Immunother Cancer*. 2020;8(1):e000398.
8. Cingolani P, et al. *Fly (Austin)*. 2012;6(2):80-92.
9. Lee E, et al. *PLOS Comput Biol*. 2008;4(11):e1000217.
10. Ayers M, et al. *J Clin Invest*. 2017;127(8):2930-2940.

Abbreviations

AE, adverse event; APM, antigen-processing machinery; BOR, best overall response; CNA, copy number alteration; CR, complete response; CTCAE, Common Terminology Criteria for Adverse Events; DOR, duration of response; ECOG, Eastern Cooperative Oncology Group; FAS, Full Analysis Set; FFPE, formalin-fixed paraffin-embedded; Gen 1, Generation 1; Gen 2, Generation 2; GSVA, Gene Set Variation Analysis; ICI, immune checkpoint inhibitors; IFNγ, interferon γ; IL-2, interleukin 2; indel, insertion and deletion; IRC, Independent Review Committee; LDH, lactate dehydrogenase; NE, nonevaluable; NMA-LD, nonmyeloablative lymphodepletion; NR, not reached; ORR, objective response rate; OS, overall survival; PBMC, peripheral blood mononuclear cells; PD, progressive disease; PD-1, programmed cell death protein 1; PD-L1, programmed death ligand 1; PFS, progression-free survival; PR, partial response; RECIST, Response Evaluation Criteria in Solid Tumors; SD, stable disease; SITC, Society for Immunotherapy of Cancer; SNV, single nucleotide variant; SOD, sum of diameters; TCR, T-cell receptor; TEAE, treatment-emergent adverse events; TGFβ, transforming growth factor β; TIL, tumor-infiltrating lymphocytes; TLS, tertiary lymphocyte structure; TMB, tumor mutation burden; TPM, transcripts per million; TPS, tumor proportion score; uCDR3, unique complementarity-determining region 3; ULN, upper limit of normal.

Acknowledgments

- The authors would like to thank the patients and their families, as well as the investigators and study site team members who are participating in the study
- The authors would also like to thank Rongsu Qi for her contributions to gene mutational analysis
- This study is sponsored by Iovance Biotherapeutics, Inc. (San Carlos, CA)
- Editorial support was provided by Second City Science and funded by Iovance Biotherapeutics

Disclosures

AS: Honoraria and consulting or advisory role with Iovance Biotherapeutics, Guidepoint, Defined Health, Huron Consulting Group, KeyQuest Health, Istari, Gerson Lehrman Group, Physicians' Educational Resource, Medscape and MedStar Health; patents, royalties, or other intellectual property rights with Moffitt Cancer Center, Iovance Biotherapeutics, and Provectus
JC: Consulting or advisory role with Amgen and research funding from Replimune, Amgen, Iovance Biotherapeutics, and Fate Therapeutics
TM: Consulting or advisory role with Merck, Bristol-Myers Squibb, Iovance Biotherapeutics, Moderna, Nektar, Regeneron, Exicure, Checkmate Pharmaceuticals, BioAtla, Xencor, Replimune, Day One Biopharmaceuticals, Pfizer, and Taiga
OH: Honoraria and consulting or advisory role with Aduro, Akeso, Alkermes, Amgen, BeiGene, BioAtla, Bristol Myers Squibb, Roche Genentech, GlaxoSmithKline, Immunocore, Idera, Incyte, InstilBio, Iovance Biotherapeutics, Janssen, Merck, NextCure, Novartis, Pfizer, Sanofi/Regeneron, Seagen, Tempus, and Zelluna; research funding from Arcus, Aduro, Akeso, Amgen, BioAtla, Bristol Myers Squibb, CytomX, Exelixis, Roche Genentech, GSK, Immunocore, Idera, Incyte, Iovance Biotherapeutics, Merck, Moderna, Merck-Serono, NextCure, Novartis, Pfizer, Rubius, Sanofi-Regeneron, Seagen, Taiga, Torque, and Zelluna; speaker's bureau for Bristol Myers Squibb, Novartis, Pfizer, and Sanofi Regeneron
ST: Honoraria, consulting or advisory role, research funding, speaker's bureau, and travel, accommodations, expenses with Bristol Myers Squibb, Merck, Pfizer, Ipsen, Amgen, Genentech, and Foundation One
MW: Honoraria from Novartis, Pfizer, and Roche; consulting or advisory role with Bristol Myers Squibb, Novartis, Pfizer, Cellex GmbH, Eli Lilly, Boehringer Ingelheim, ISA Pharmaceuticals, GEMoaB, Roche, MSD, AstraZeneca, Amgen, and Immatics; research funding from Roche; travel, accommodations, and expenses from Pfizer, Bristol Myers Squibb, AstraZeneca, Roche, Amgen, and GEMoaB
VG: Employee of Iovance Biotherapeutics and holds Iovance Biotherapeutics and Gilead stocks
BD, JD, EM, XW, and WS: Employees of Iovance Biotherapeutics holding Iovance stock and/or stock options
HK: Consulting or advisory role with Bristol Myers Squibb, Clinigen, Shionogi, ChemoCentryx, Calithera, Signatera, GigaGen, GI Reviewers, and Merck; research funding from Apexigen, Bristol Myers Squibb, and Merck

© 2023, Iovance Biotherapeutics