270 Tandem Meetings | Transplantation & Cellular Therapy Meetings of ASTCT™ & CIBMTR®, April 23–26, 2022; Salt Lake City, UT, USA

Decitabine Treatment of Tumor-Infiltrating Lymphocytes (TIL) During Ex Vivo Expansion Induces a More Memory-like Phenotype, Reduces Inhibitory Receptor Expression, and Increases Functionality

Rafael Cubas,[1] Andrew Yuhas,[1] Marcus Machin,[1] Yongliang Zhang[1]

[1]Iovance Biotherapeutics, Inc., San Carlos, CA, USA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASTCT™ & CIBMTR®, and the author of this poster

Corresponding Author:
Rafeal Cubas; rafael.cubas@iovance.com

Background

- Adoptive cell therapy using autologous tumor-infiltrating lymphocytes (TIL) has shown durable responses in patients with metastatic melanoma,[1] cervical carcinoma,[2] and other epithelial malignancies, such as metastatic head and neck squamous cell carcinoma and metastatic non-small cell lung cancer[3-5]
- Whereas TIL can be reactivated and expanded ex vivo, their epigenetic programming could be maintaining a more differentiated and less functional state
- De novo DNA methylation has been shown to promote and reinforce the development of T-cell exhaustion[6] and represents a cell-intrinsic barrier to T-cell reinvigoration
- Low-dose decitabine (DAC), a DNA hypomethylation agent, has been shown to confer some level of epigenetic reprogramming on exhausted T cells and endow chimeric antigen receptor (CAR) T cells with enhanced persistence, memory-like phenotype, and antitumor potential[7]
- In this study, we investigated whether low-dose DAC treatment during ex vivo TIL expansion could improve the quality and function of TIL by reducing their effector differentiation and maintaining a population of more memory-like cells

Methods

- Patient tumors (N = 8) from different tumor types (non-small cell lung, head & neck, ovarian, and breast cancers) were received, fragmented, and subjected to a 22-day expansion protocol for TIL generation
- Different concentrations (10 nM, 30 nM, and 100 nM) of DAC were added to the culture during ex vivo expansion either during the pre-REP and REP stages or during REP only (**Figure 1**)
- The expansion potential as well as the phenotypic and functional characteristics of TIL were evaluated in the final TIL product

Figure 1.

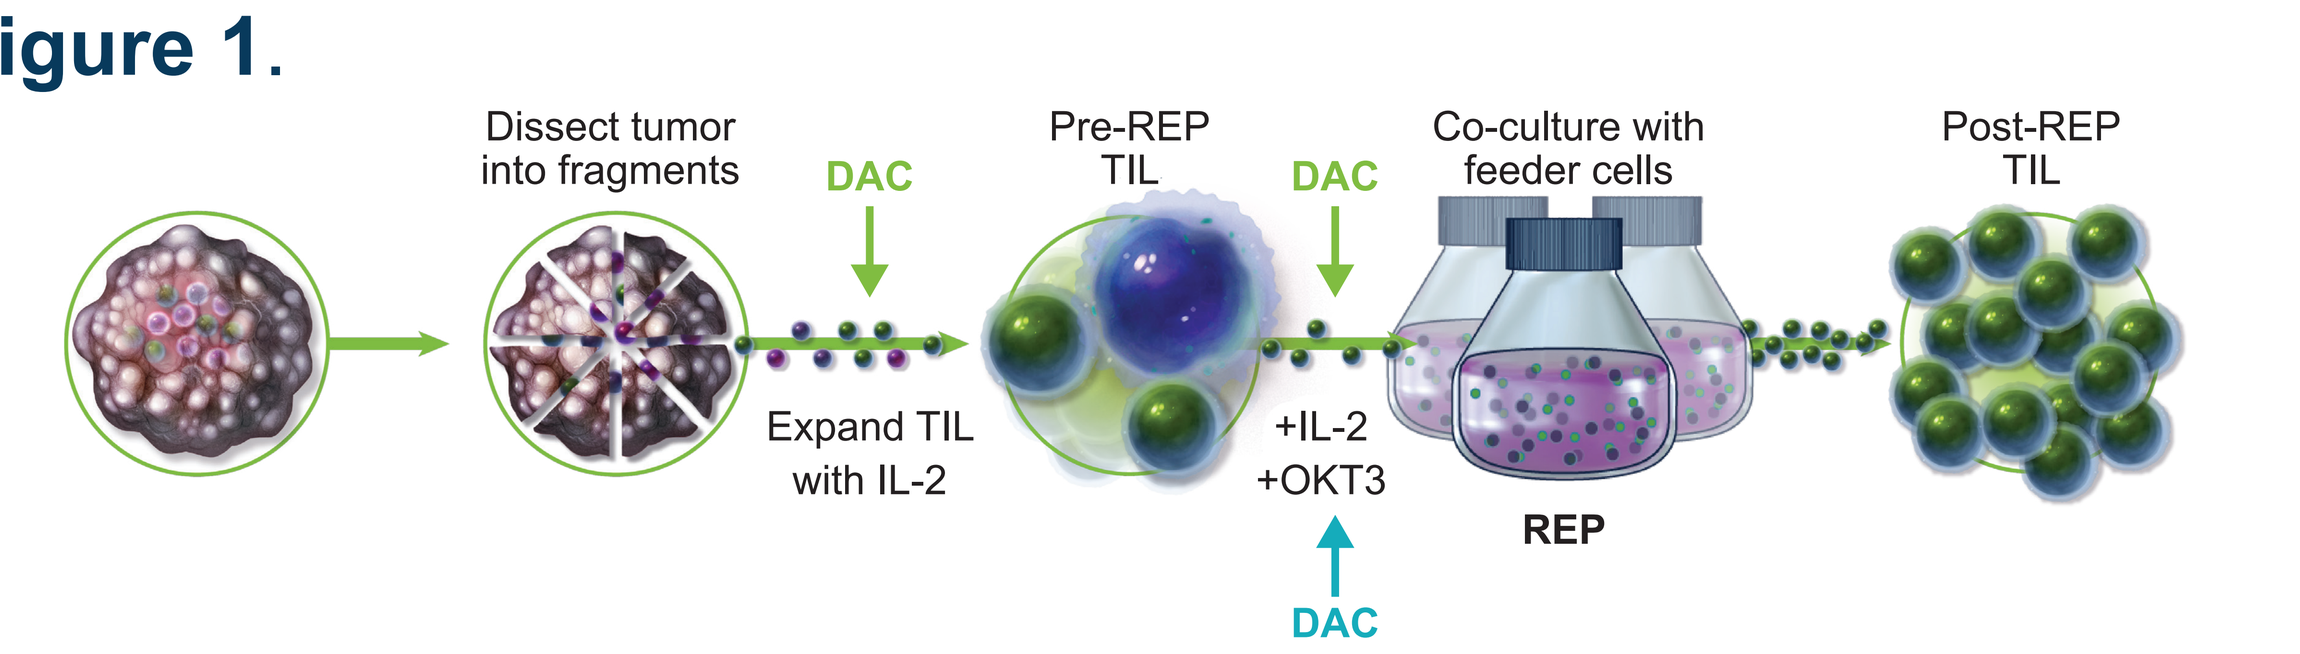
Dissect tumor into fragments
DAC
Pre-REP TIL
DAC
Co-culture with feeder cells
Post-REP TIL
Expand TIL with IL-2
+IL-2
+OKT3
REP
DAC

Results

Figure 2. DAC treatment maintained TIL viability, but decreased expansion while increasing the CD4+/CD8+ T-cell ratio

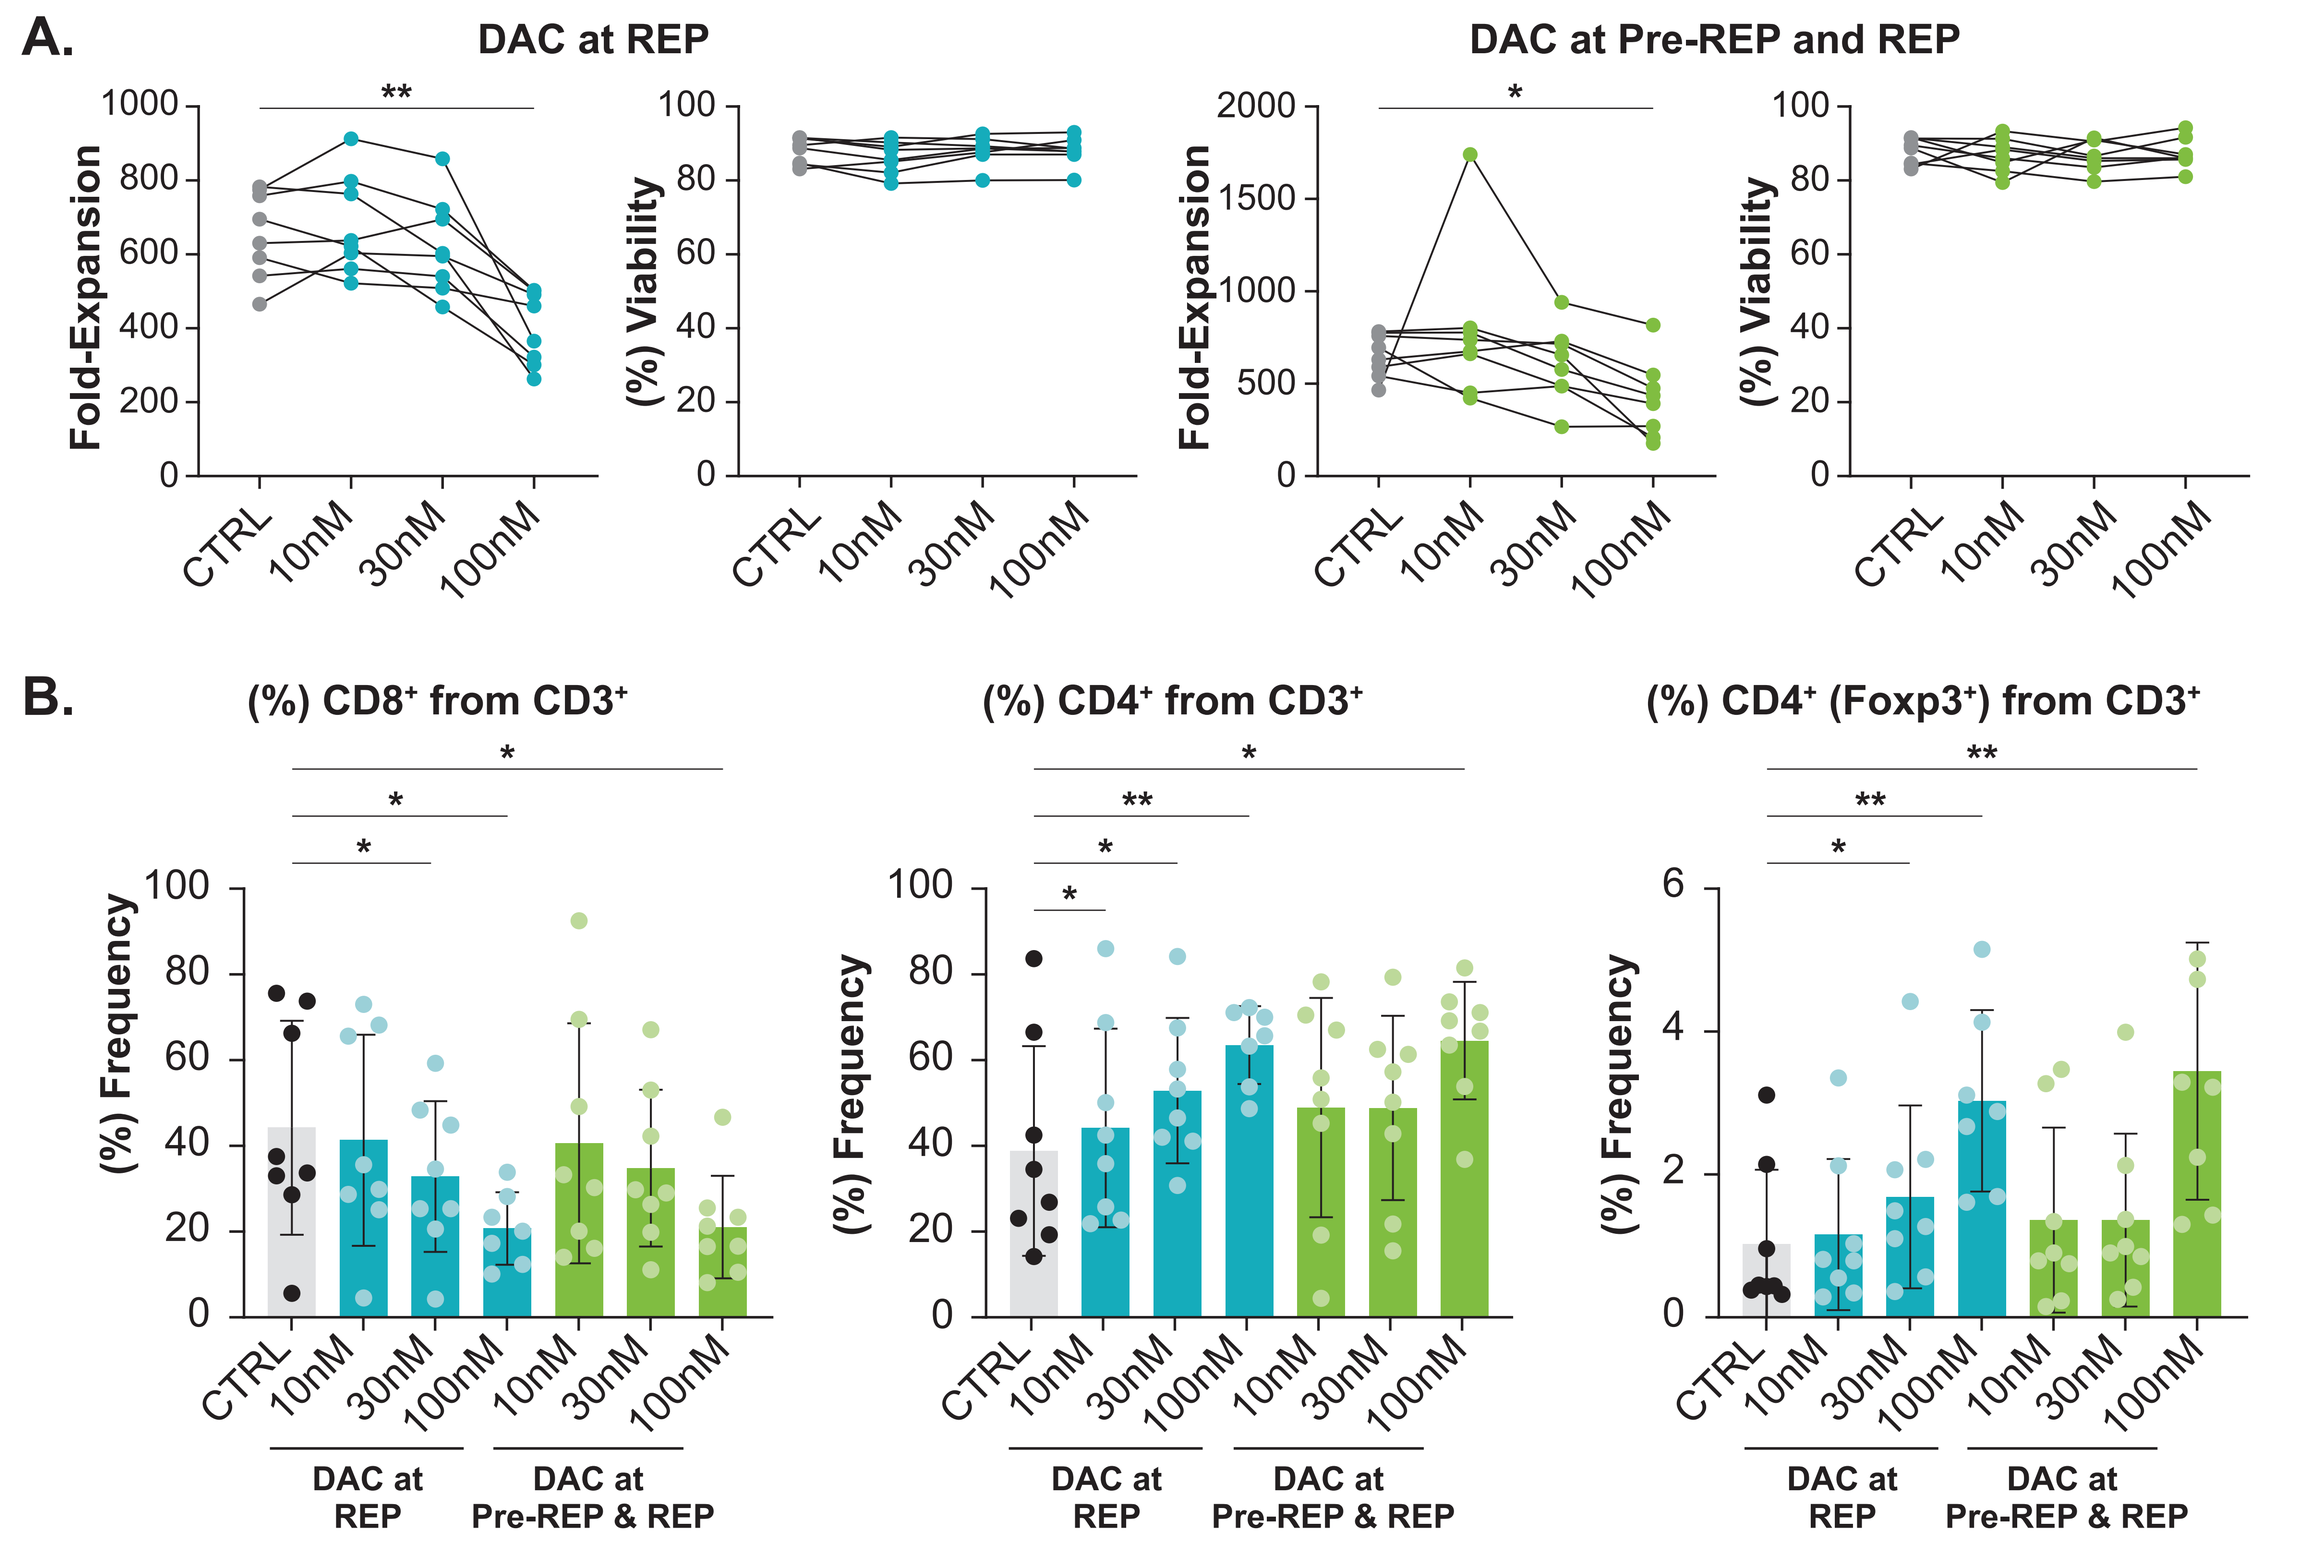
A.
DAC at REP
DAC at Pre-REP and REP
Fold-Expansion
(%) Viability
B.
(%) CD8+ from CD3+
(%) CD4+ from CD3+
(%) CD4+ (Foxp3+) from CD3+
DAC at REP
DAC at Pre-REP & REP

Expansion, viability, and T-cell distribution in control- and DAC-treated TIL. TIL were left untreated (CTRL, gray bars) or treated with increasing concentrations of DAC. Treatment was added either during the REP stage only (blue bars) or during both pre-REP and REP (green bars). **A.** Fold-expansion and viability of TIL at the end of the 22-day expansion process. **B.** Frequency of CD8+, CD4+, and CD4+ (Foxp3+) cells after the expansion process on cryopreserved cells. *$P < 0.05$, **$P < 0.01$.

Figure 3. DAC treatment at REP increased the frequency of Tcm-like cells in both CD8+ and CD4+ T cells

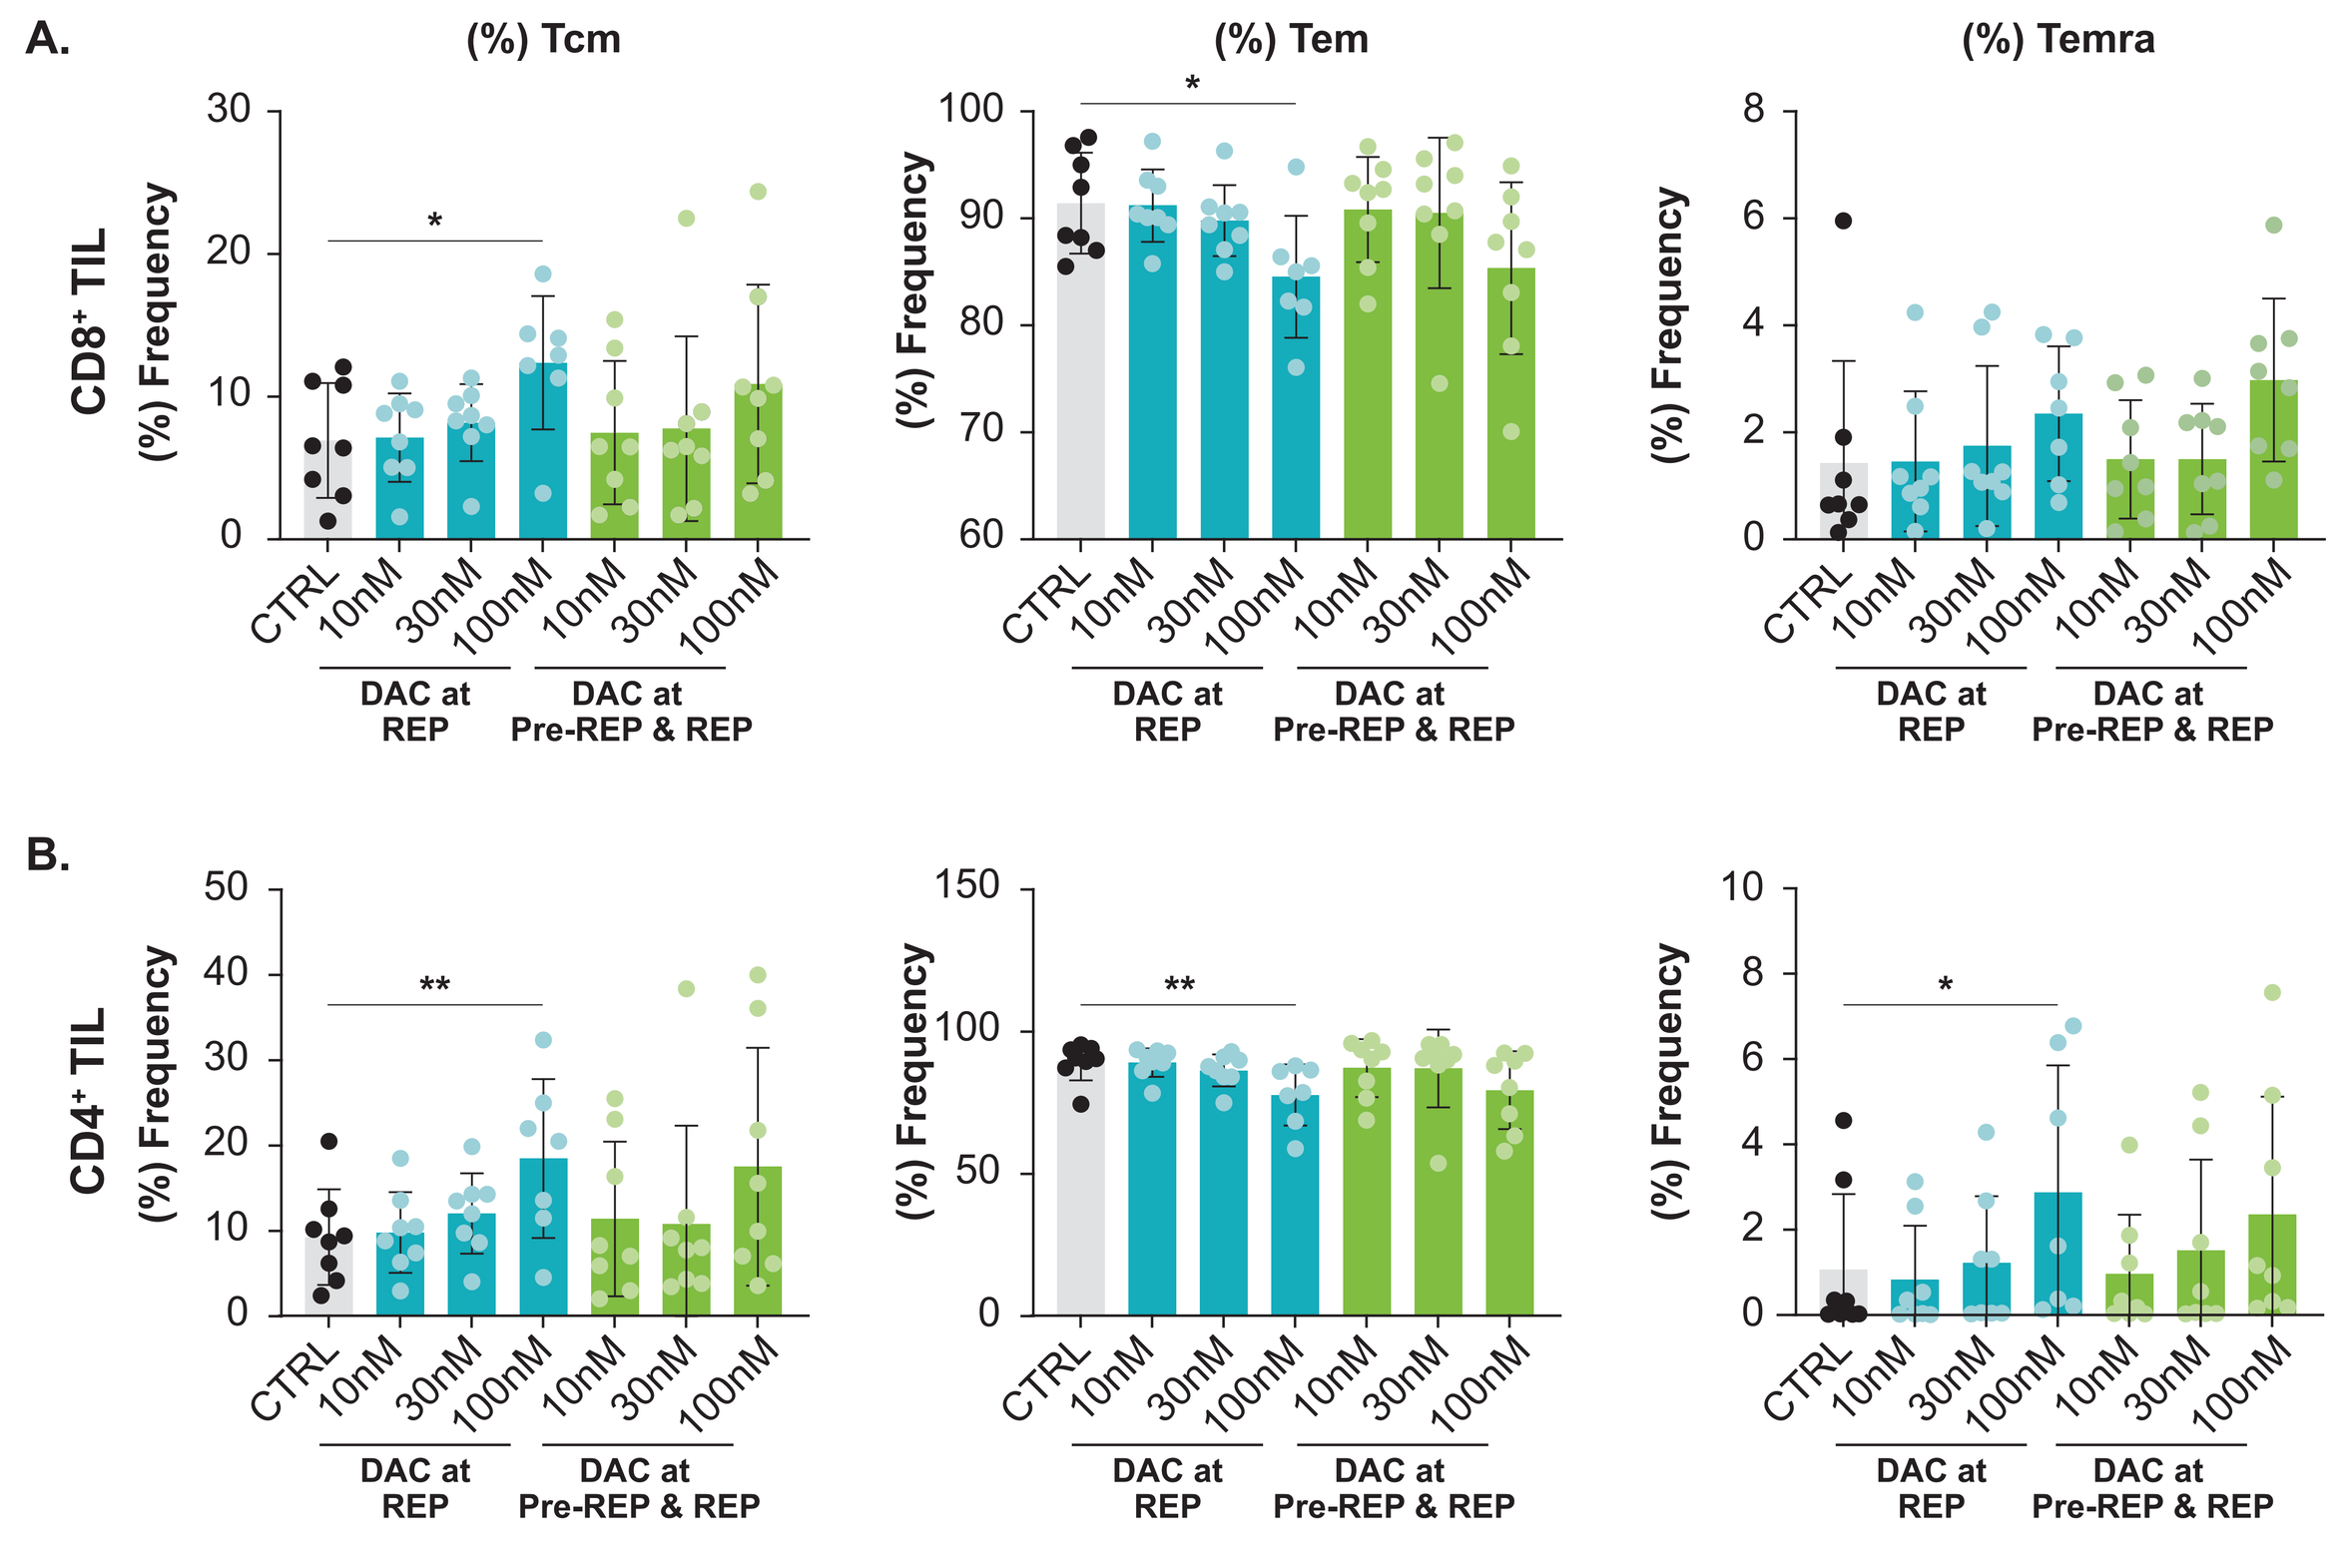
A.
(%) Tcm
(%) Tem
(%) Temra
CD8+ TIL
B.
CD4+ TIL
(%) Frequency
DAC at REP
DAC at Pre-REP & REP

T-cell subsets in control- and DAC-treated TIL. Frequency of Tcm (CD45RA-CCR7+), Tem (CD45RA-CCR7-), and Temra (CD45+CCR7-) cells in **A.** CD8+ and **B.** CD4+ TIL after expansion. *$P < 0.05$, **$P < 0.01$.

Figure 4. DAC treatment increased the frequency of co-stimulatory receptors while decreasing inhibitory receptor expression on CD8+ TIL

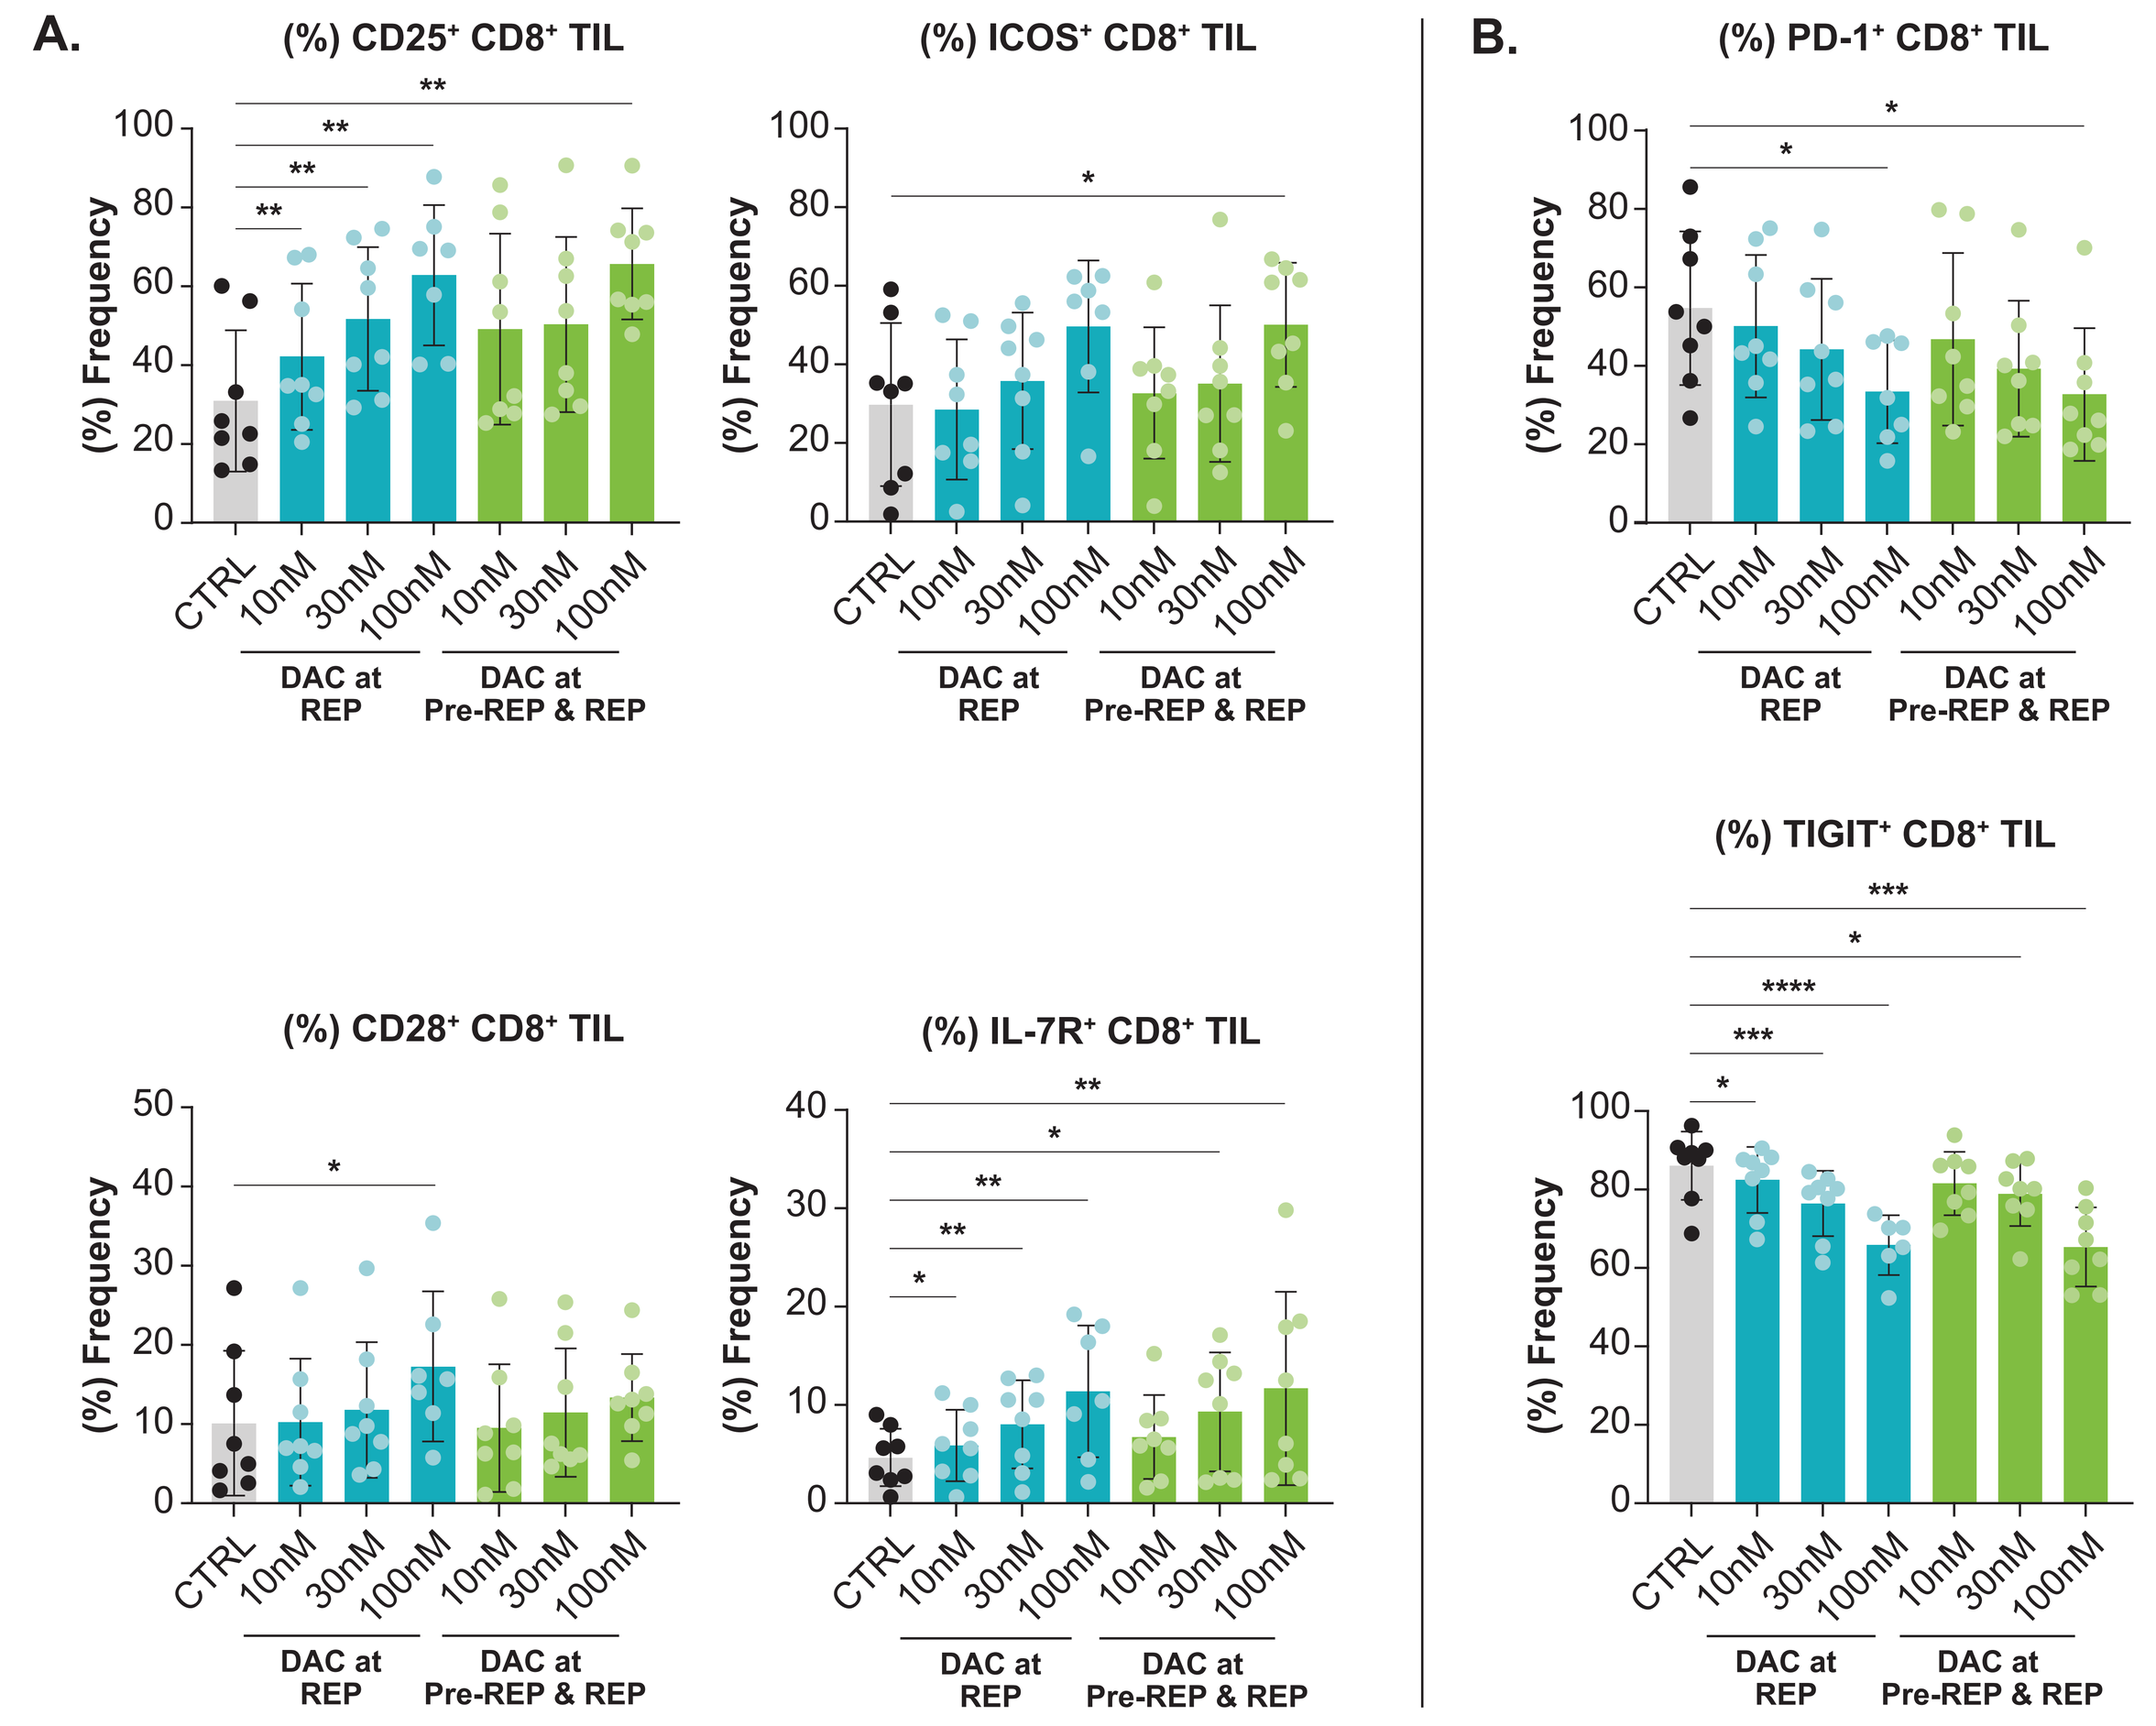
A.
(%) CD25+ CD8+ TIL
(%) ICOS+ CD8+ TIL
(%) CD28+ CD8+ TIL
(%) IL-7R+ CD8+ TIL
B.
(%) PD-1+ CD8+ TIL
(%) TIGIT+ CD8+ TIL
(%) Frequency
DAC at REP
DAC at Pre-REP & REP

Expression of surface markers on DAC-treated TIL. Control- or DAC-treated cryopreserved TIL were thawed and stained for flow cytometry analysis. **A.** Expression of CD25, ICOS, CD28, and IL-7R on CD8+ TIL. **B.** Expression of inhibitory receptors PD-1 and TIGIT on CD8+ TIL. Similar results were observed for CD4+ TIL. *$P < 0.05$, **$P < 0.01$, ***$P < 0.001$, ****$P < 0.0001$.

Figure 5. DAC treatment increased the expression of memory-associated transcription factors

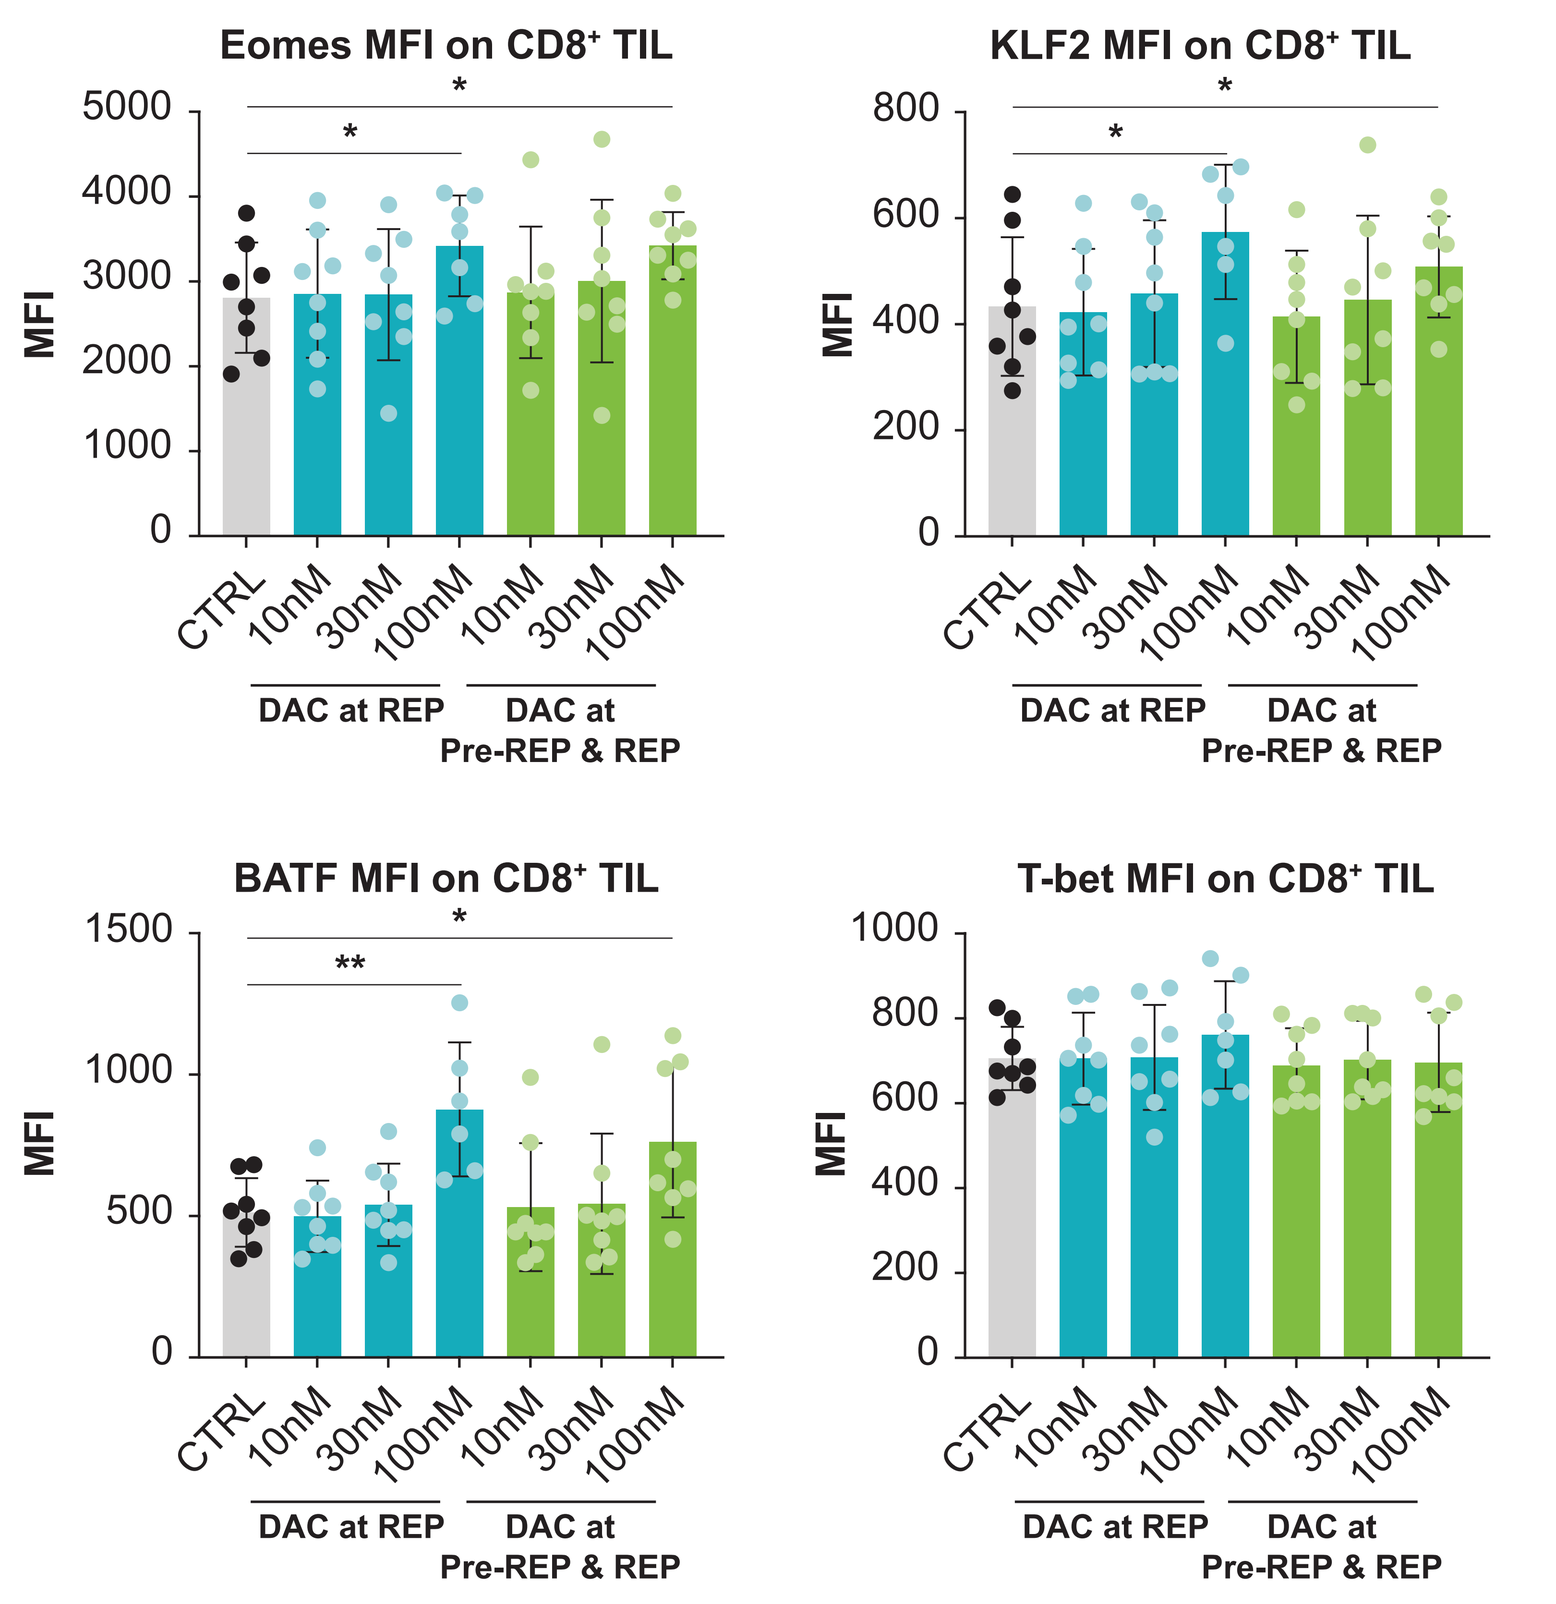
Eomes MFI on CD8+ TIL
KLF2 MFI on CD8+ TIL
BATF MFI on CD8+ TIL
T-bet MFI on CD8+ TIL
MFI
DAC at REP
DAC at Pre-REP & REP

Expression of transcription factors in DAC-treated TIL. Control- or DAC-treated cryopreserved TIL were thawed and stained for flow cytometry analysis. Expression of Eomes, KLF2, BATF, and T-bet on CD8+ TIL are shown. *$P < 0.05$, **$P < 0.01$.

Figure 6. DAC treatment increased the frequency of TNFα- and GZMB-expressing CD8+ TIL following stimulation

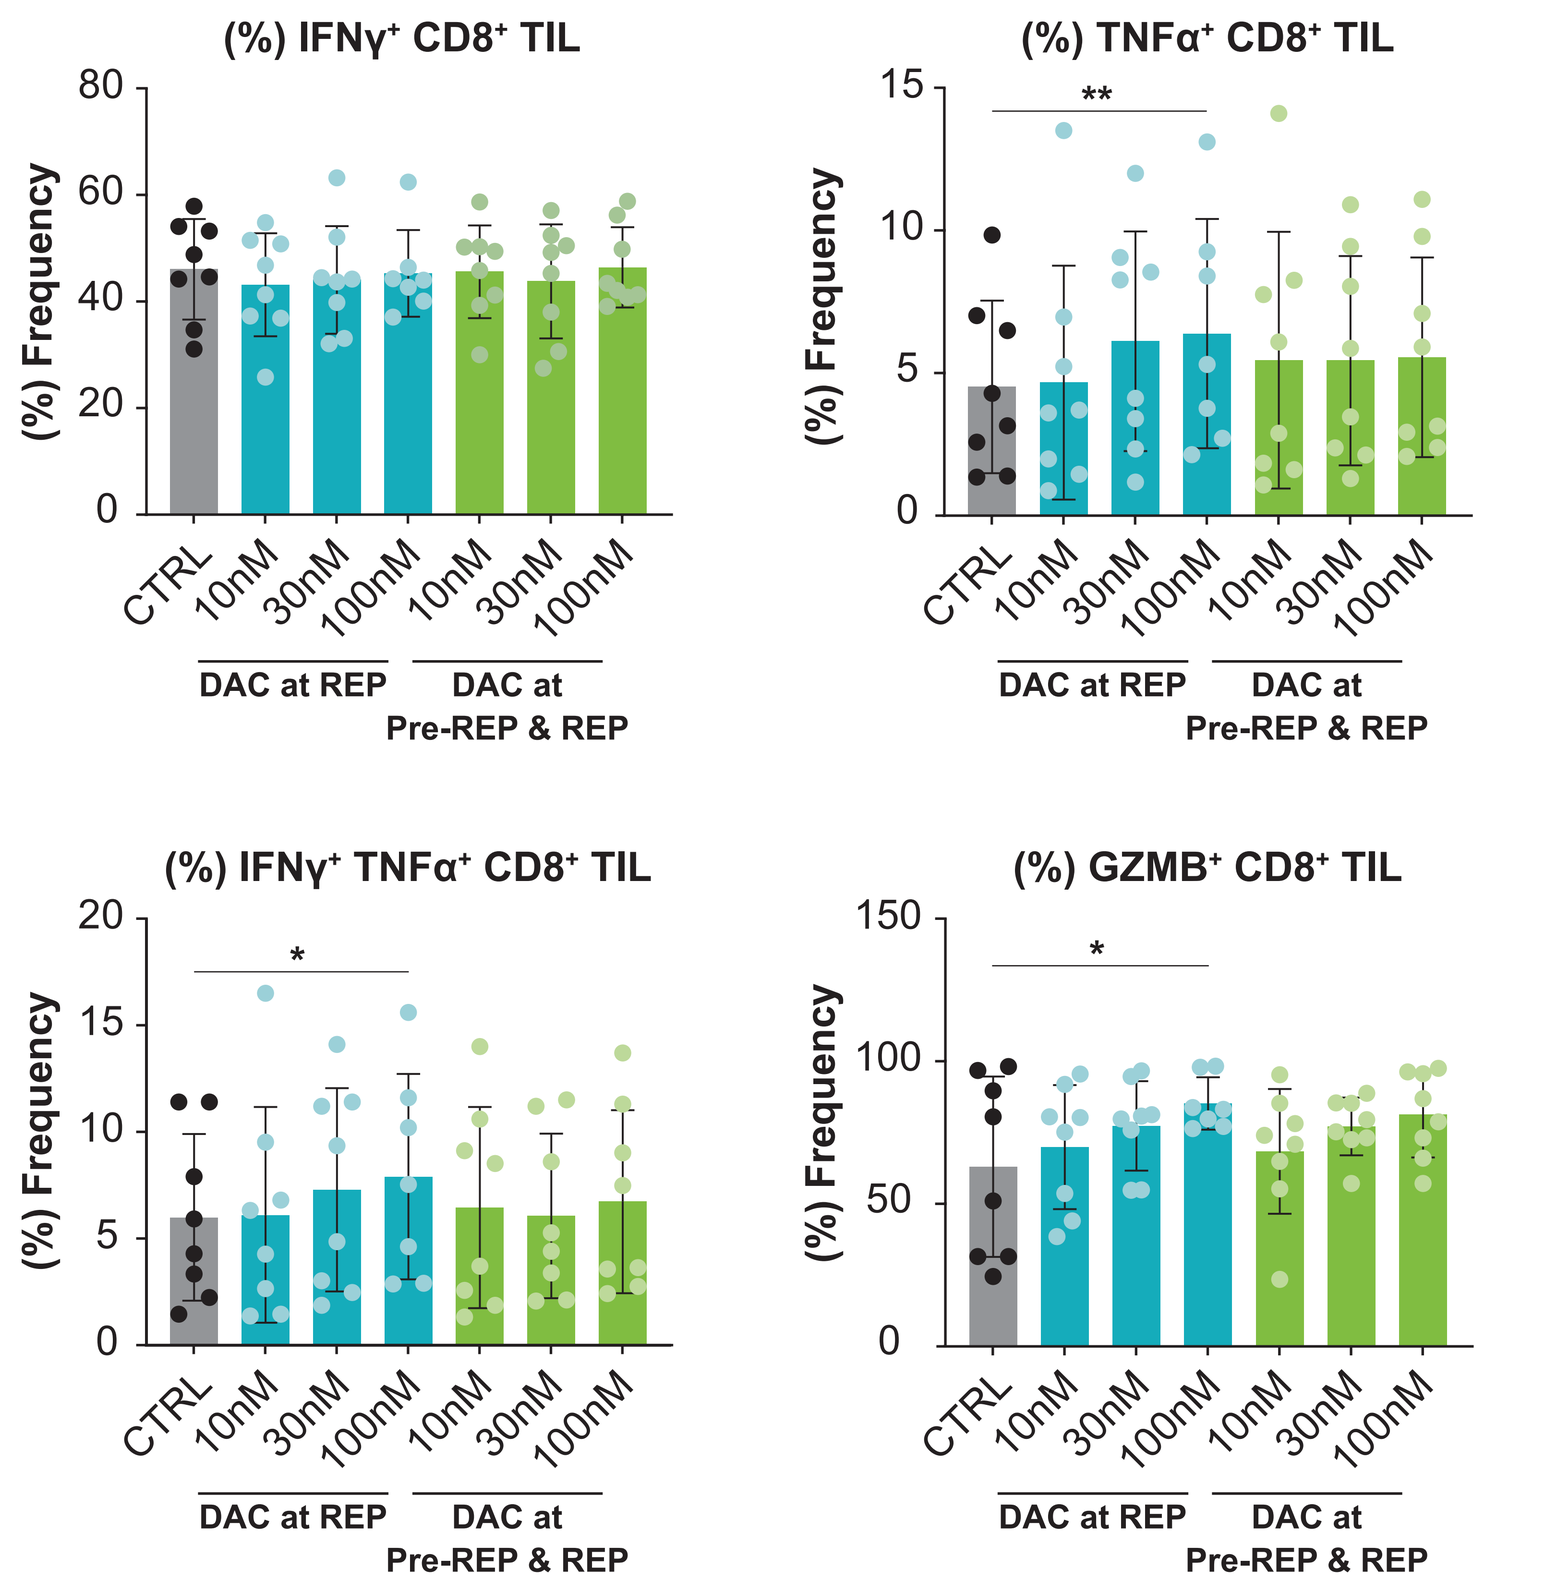
(%) IFNγ+ CD8+ TIL
(%) TNFα+ CD8+ TIL
(%) IFNγ+ TNFα+ CD8+ TIL
(%) GZMB+ CD8+ TIL
(%) Frequency
DAC at REP
DAC at Pre-REP & REP

Cytokine expression in control- or DAC-treated TIL following in vitro stimulation. Cryopreserved control- and DAC-treated TIL were stimulated overnight with anti-CD3/CD28 beads at a bead-to-cell ratio of 1:5. Expression of IFNγ, TNFα, and GZMB on CD8+ TIL are shown. *$P < 0.05$, **$P < 0.01$.

Figure 7. DAC-treated TIL showed increased cytotoxicity that was sustained after repeated stimulation

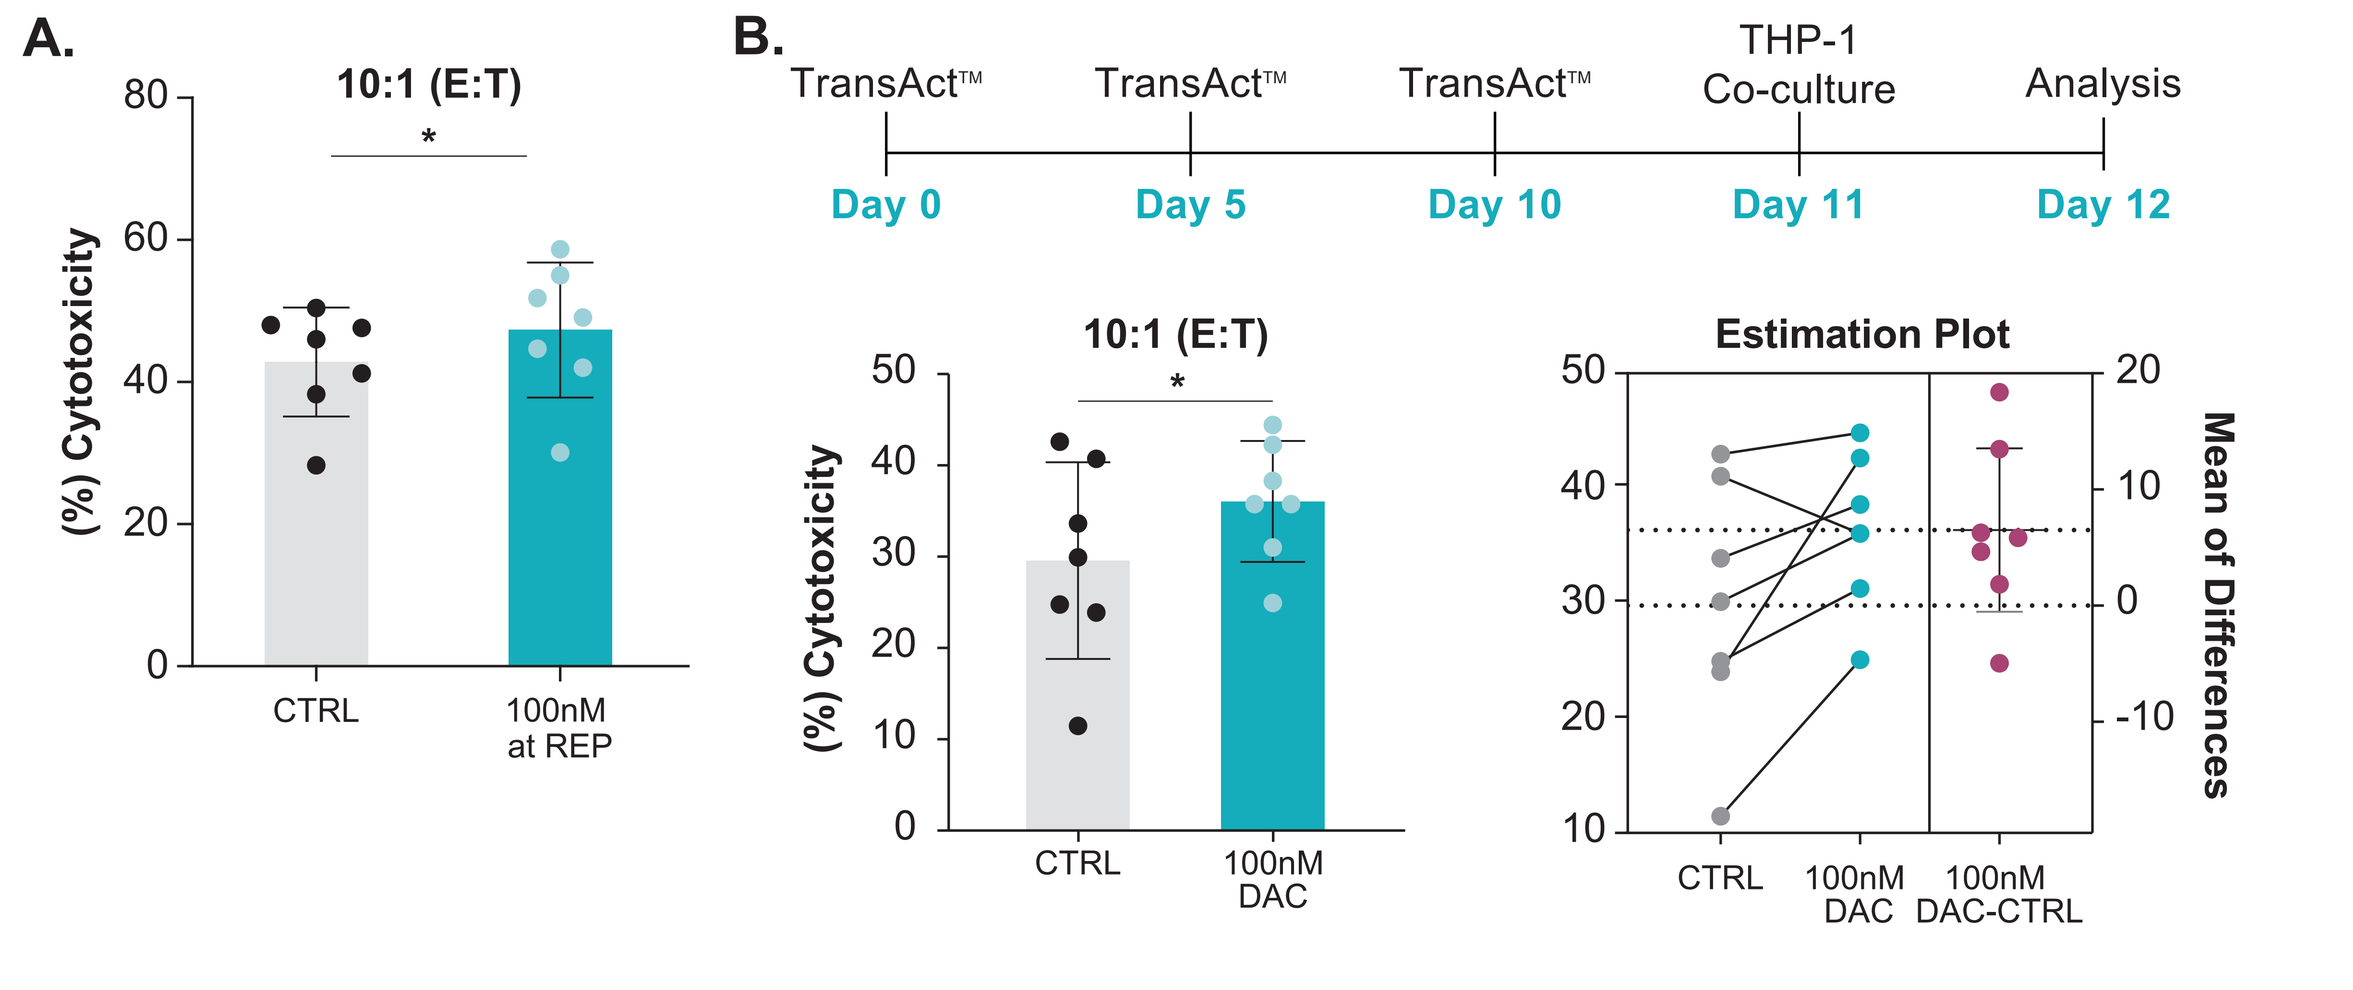
A.
10:1 (E:T)
(%) Cytotoxicity
CTRL
100nM at REP
B.
TransAct™
TransAct™
TransAct™
THP-1 Co-culture
Analysis
Day 0
Day 5
Day 10
Day 11
Day 12
10:1 (E:T)
Estimation Plot
Mean of Differences
CTRL
100nM DAC
100nM DAC-CTRL

Cytotoxicity of control- and DAC-treated TIL. **A.** Cryopreserved control and TIL treated at REP with 100 nM DAC were cocultured for 24 h with KILR® THP-1 cells (Eurofins DiscoverX, Fremont, CA, USA) at a 10:1 E:T cell ratio to measure cytotoxicity in an allogeneic setting. **B.** Control- and DAC-treated TIL were stimulated every 5 days with TransAct™ (Miltenyi Biotec, Germany). One day after the third stimulation, cells were washed and cocultured at a 10:1 effector-to-target cell ratio with KILR THP-1 cells for 24 h to measure cytotoxicity. *$P < 0.05$.

Figure 8. DAC-treated TIL showed reduced inhibitory receptor expression and lower levels of TOX while having increased IL-7R expression after repeated stimulation

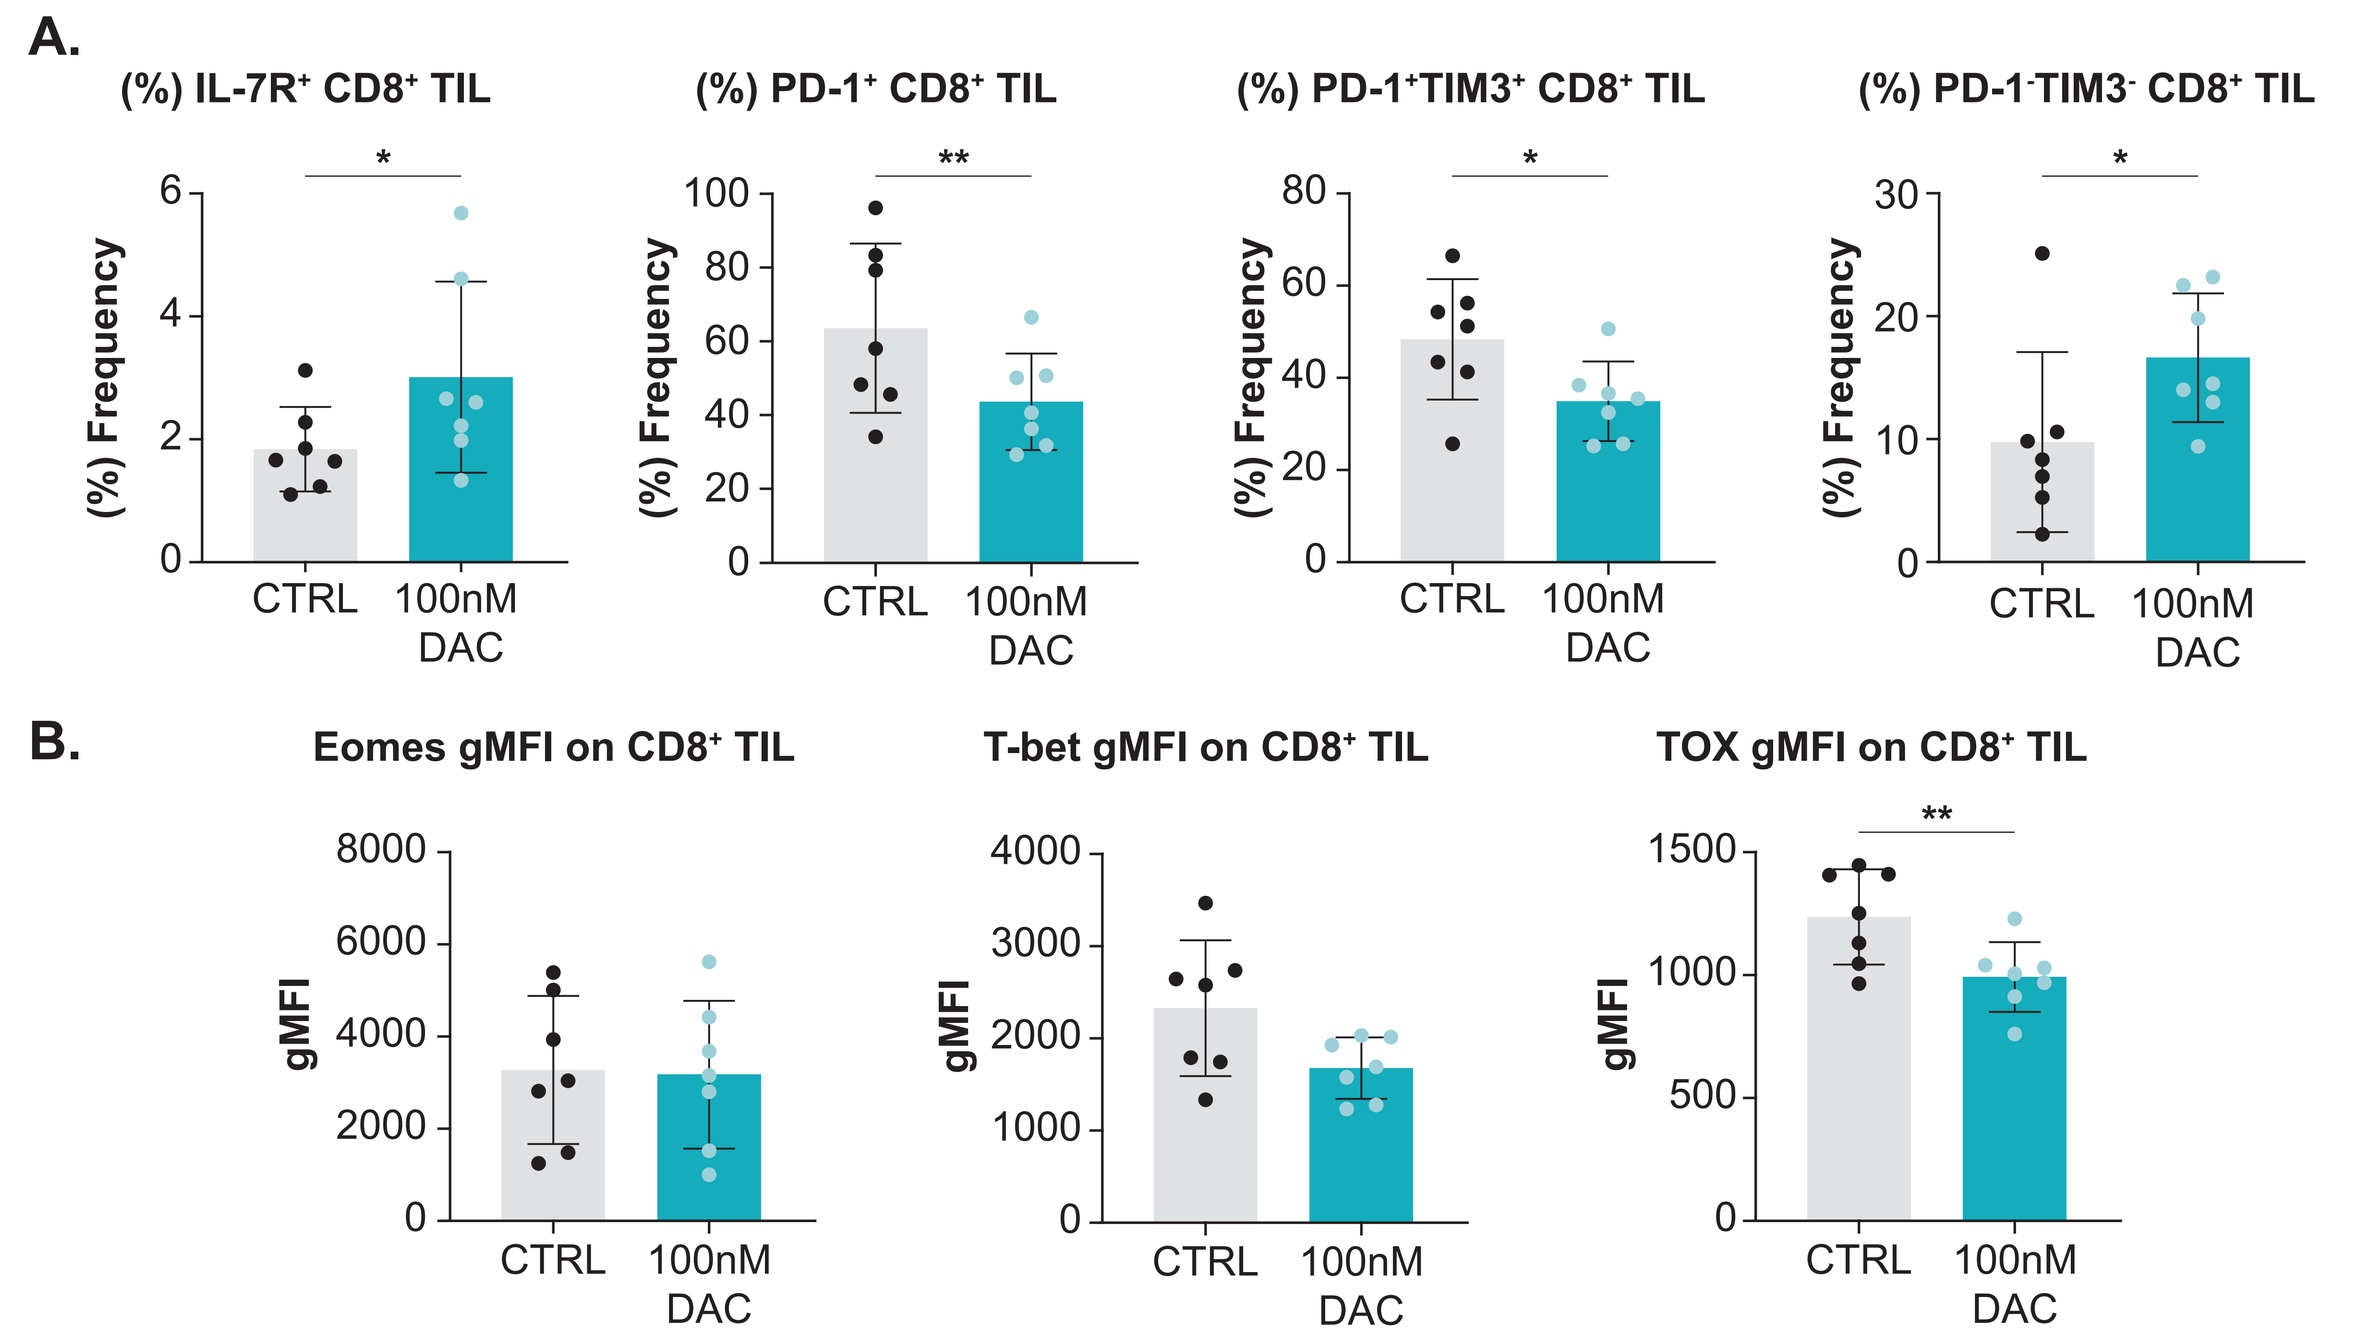
A.
(%) IL-7R+ CD8+ TIL
(%) PD-1+ CD8+ TIL
(%) PD-1+TIM3+ CD8+ TIL
(%) PD-1-TIM3- CD8+ TIL
B.
Eomes gMFI on CD8+ TIL
T-bet gMFI on CD8+ TIL
TOX gMFI on CD8+ TIL
CTRL
100nM DAC

Phenotype of control- and DAC-treated TIL after repeated stimulation. Control- and DAC-treated TIL were stimulated every 5 days with TransAct™ (Miltenyi Biotec, Germany). One day after the third stimulation, cells were washed and stained for flow cytometry analysis. **A.** Expression of IL-7R, PD-1, and TIM3 in TIL after repeated stimulation. **B.** Expression levels of transcription factors in TIL after repeated stimulation. *$P < 0.05$, **$P < 0.01$.

Conclusions

- DAC treatment during TIL expansion can shift the balance away from effector differentiation and toward a more memory-like phenotype
- DAC treatment at 100 nM in the REP stage only increased the expression of costimulatory receptors while reducing inhibitory receptor expression
- DAC treatment increased the frequency of TNFα+ and IFNγ+TNFα+ CD8+ TIL while conferring increased killing activity, which was sustained even after repeated stimulation
- DAC-treated TIL showed reduced TOX levels and lower frequency of PD1+TIM3+ CD8+ TIL following repeated stimulation
- Inhibiting DNA methylation programs during TIL expansion could represent a useful approach for modifying the epigenetic regulation of TIL to improve their therapeutic potential

Abbreviations

BATF, basic leucine zipper ATF-like transcription factor; CAR, chimeric antigen receptor; CTRL, control; DAC, decitabine; Eomes, eomesodermin; E:T, effector-to-target cell; gMFI, geometric mean fluorescence intensity; GZMB, Granzyme B; ICOS, inducible T-cell costimulatory; IFNγ, interferon-γ; IL-2, interleukin-2; IL-7R, interleukin-7 receptor; KLF2, Kruppel-like factor 2; MFI, mean fluorescence intensity; PD-1, programmed cell death protein-1; REP, rapid expansion protocol; Tcm, central memory T-cell subset; Tem, effector memory T-cell subset; Temra, effector memory RA+ T-cell subset; TIGIT, T cell immunoreceptor with Ig and ITIM domains; TIL, tumor-infiltrating lymphocytes; TIM3, T cell immunoglobulin and mucin domain-containing protein 3; TNFα, tumor necrosis factor-α; TOX, thymocyte selection-associated high mobility group box protein.

References

1. Sarnaik AA, et al. *J Clin Oncol*. 2021;39(24):2656-2666.
2. Jazaeri AA, et al. *J Clin Oncol*. 2019;37(Suppl 15):2538-2538.
3. Schoenfeld AJ, et al. *J Immunother Cancer*. 2021;9(Suppl 2): Abstract 458.
4. Creelan BC, et al. *Nat Med*. 2021;27:1410-1418.
5. O'Malley D, et al. *J Immunother Cancer*. 2021;9(Suppl 2):Abstract 492.
6. Ghoneim HE, et al. *Cell*. 2017;170(1):142-157.
7. Wang Y, et al. *Nat Commun*. 2021;12(1):409.

Acknowledgments

- Editorial support was provided by Second City Science (Chicago, IL, USA) and funded by Iovance Biotherapeutics

Disclosures

- All authors meet the criteria for authorship set forth by the International Committee of Medical Journal Editors
- All authors are employees of Iovance and may have stock options

© 2022, Iovance Biotherapeutics