403 Society for Immunotherapy of Cancer 38th Annual Meeting | November 3-5, 2023 | San Diego, CA

Tumor-infiltrating lymphocytes (TIL) with inducible and membrane-bound IL-12 exhibit superior anti-tumor activity *in vitro*

Yongliang Zhang,[1] Nathan Gilbert,[1] Patrick Innamarato,[1] Judy Fang,[1] Hequn Yin[1]

[1]Iovance Biotherapeutics, Inc., San Carlos, CA, USA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster

Corresponding Author: Hequn Yin; hequn.yin@iovance.com

Background

- Tumor-infiltrating lymphocyte (TIL) cell therapy has shown clinical benefit for patients with solid tumors[1,2]
 - However, an immunosuppressive tumor microenvironment (TME) may abrogate the full potential of TIL cell therapy[3]
- The proinflammatory cytokine IL-12, known to increase IFN-γ production and promote type 1 immune responses, reshapes the TME and has the potential to augment anti-tumor activity[4]
- Engineered TIL secreting IL-12 showed clinical benefit, although circulating IL-12–related AEs limited its further development[5]
 - Therefore, enabling IL-12 expression on the surface of therapeutic T cells has the potential to attenuate IL-12 toxicity[6]
- Here, we report on the development, characterization, and cytotoxic activity of genetically engineered TIL with inducible and membrane-bound IL-12, incorporated into a 22-day TIL manufacturing process

Methods

- Tissues from several solid tumor histologies including non-small cell lung cancer, breast cancer, head & neck cancer, and ovarian cancer were fragmented and cultured for 11 days (pre-rapid expansion protocol [pre-REP])
- The tumor tissue cultures were then transduced with a lentivirus containing a gene encoding membrane-bound IL-12 via an EF-1α promoter (TeIL-12; constitutively expressing TeIL-12 TIL) or an NFAT-promoter (NFAT-TeIL-12; inducible TeIL-12 TIL), and underwent a subsequent 11-day expansion period via a rapid expansion protocol (REP) (**Figure 1**).
 - Mock (without transduction) and pLenti-control (pLV ctrl) virally transduced TIL were simultaneously evaluated as controls

Figure 1. Generation of TeIL-12 genetically engineered TIL

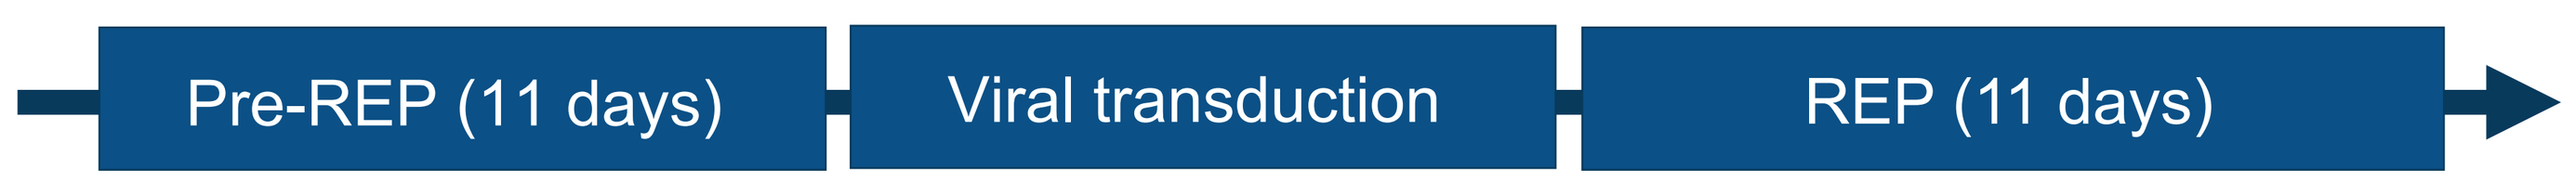
Pre-REP (11 days)
Viral transduction
REP (11 days)

- The expression, biological function, and shedding of IL-12 molecules were assessed *in vitro*
- TeIL-12 genetically engineered TILs were characterized by flow cytometry
- *In vitro* cytotoxic activity of TeIL-12 genetically engineered TIL was evaluated using the KILR®-THP-1 (Eurofins DiscoverX, Fremont, CA, USA) and xCELLigence® (Agilent Technologies, Santa Clara, CA) assays

Results

Figure 2. TeIL-12 is expressed on the cell surface of TIL and triggers downstream signaling in a contact-dependent manner

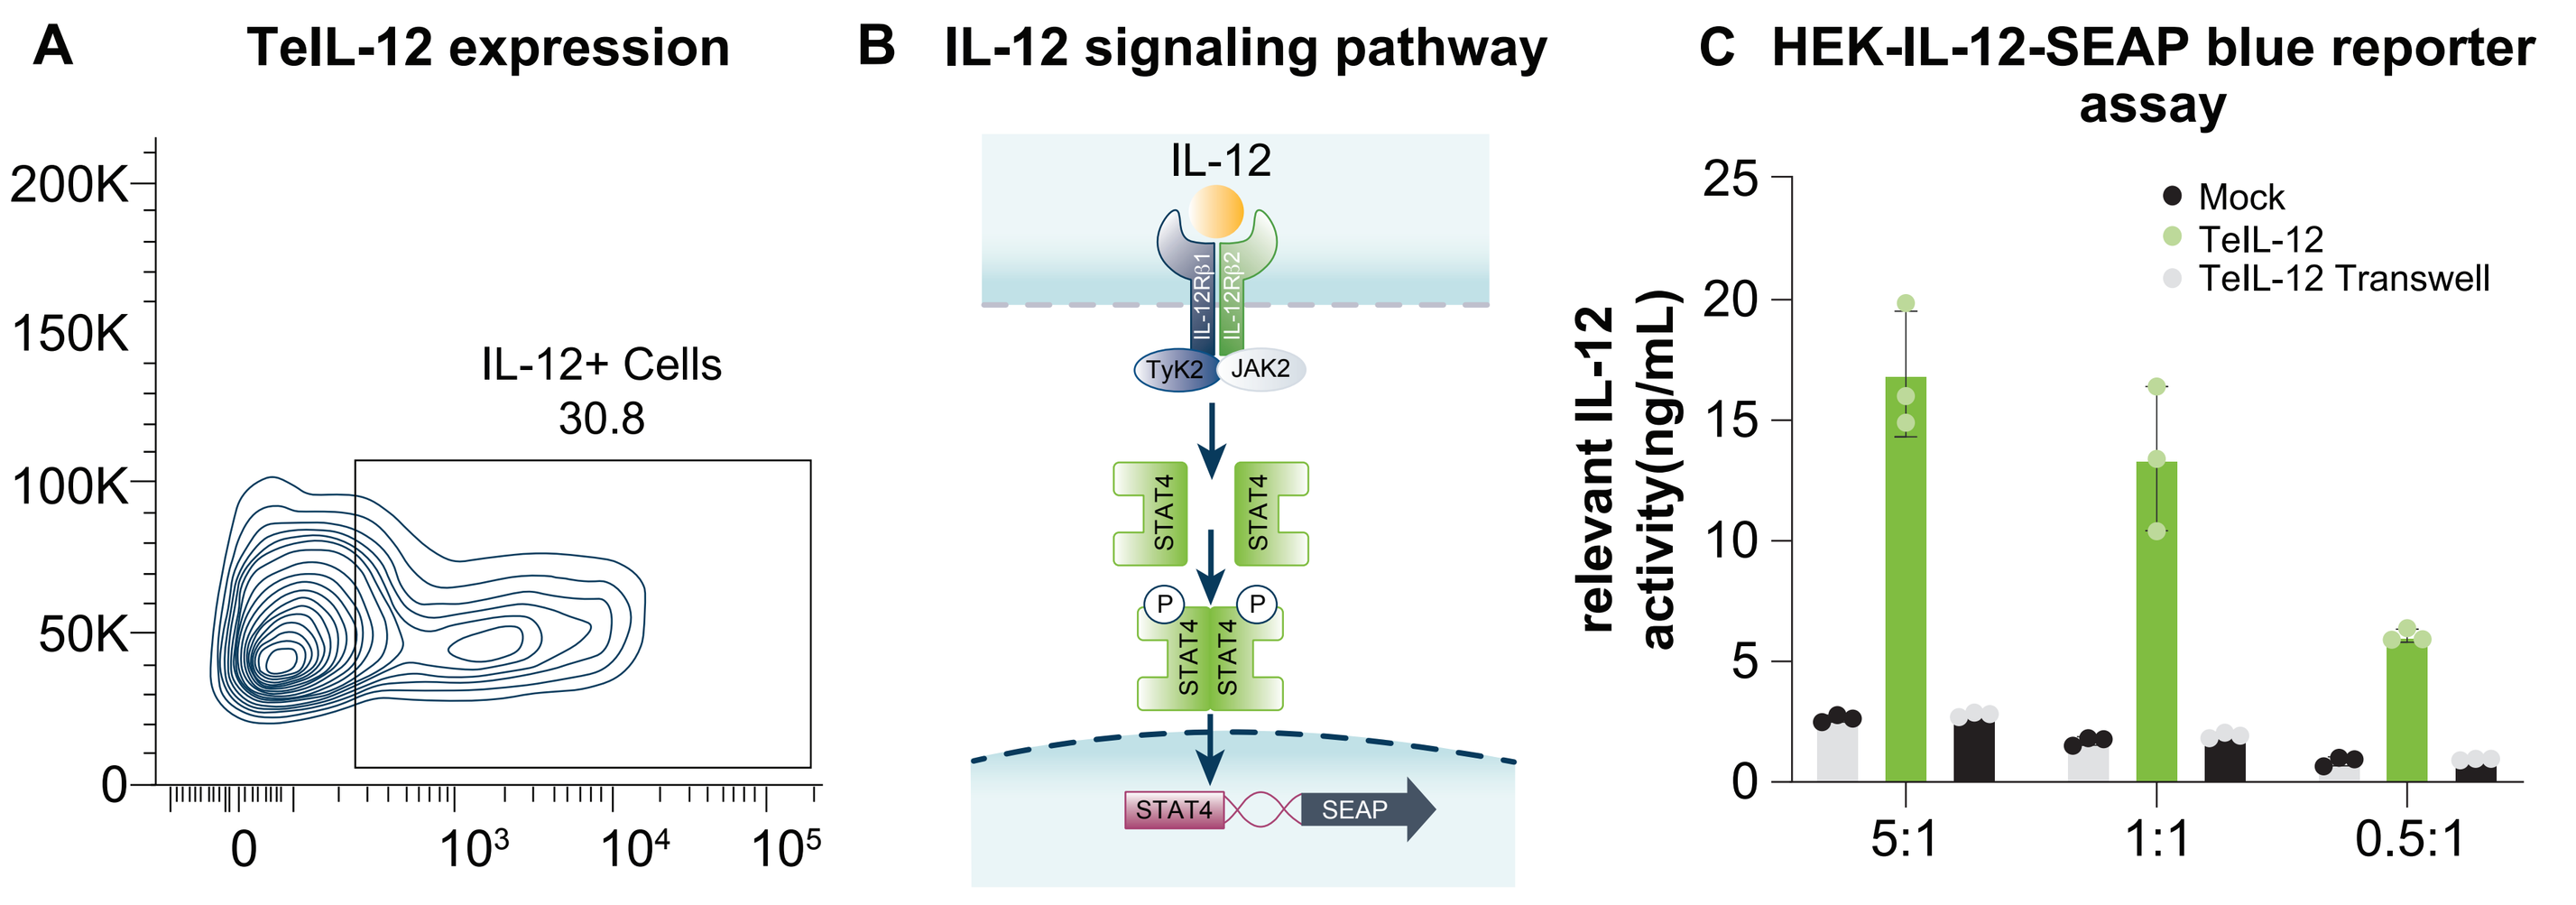
A TeIL-12 expression
IL-12+ Cells 30.8
B IL-12 signaling pathway
IL-12
TyK2 JAK2
STAT4
SEAP
C HEK-IL-12-SEAP blue reporter assay
Mock
TeIL-12
TeIL-12 Transwell
relevant IL-12 activity(ng/mL)
5:1
1:1
0.5:1

A. Surface expression of TeIL-12 on REP-TIL was examined by flow cytometry. **B.** TeIL-12–expressing TIL can activate downstream signaling through IL-12 receptor on **C.** HEK-IL-12-SEAP blue reporter cells, but activation was disrupted when cell-cell interaction was blocked with a transwell, indicating a lack of IL-12 shedding

Figure 3. NFAT promoter drives inducible TeIL-12 (NFAT-TeIL-12) expression upon T-cell receptor stimulation

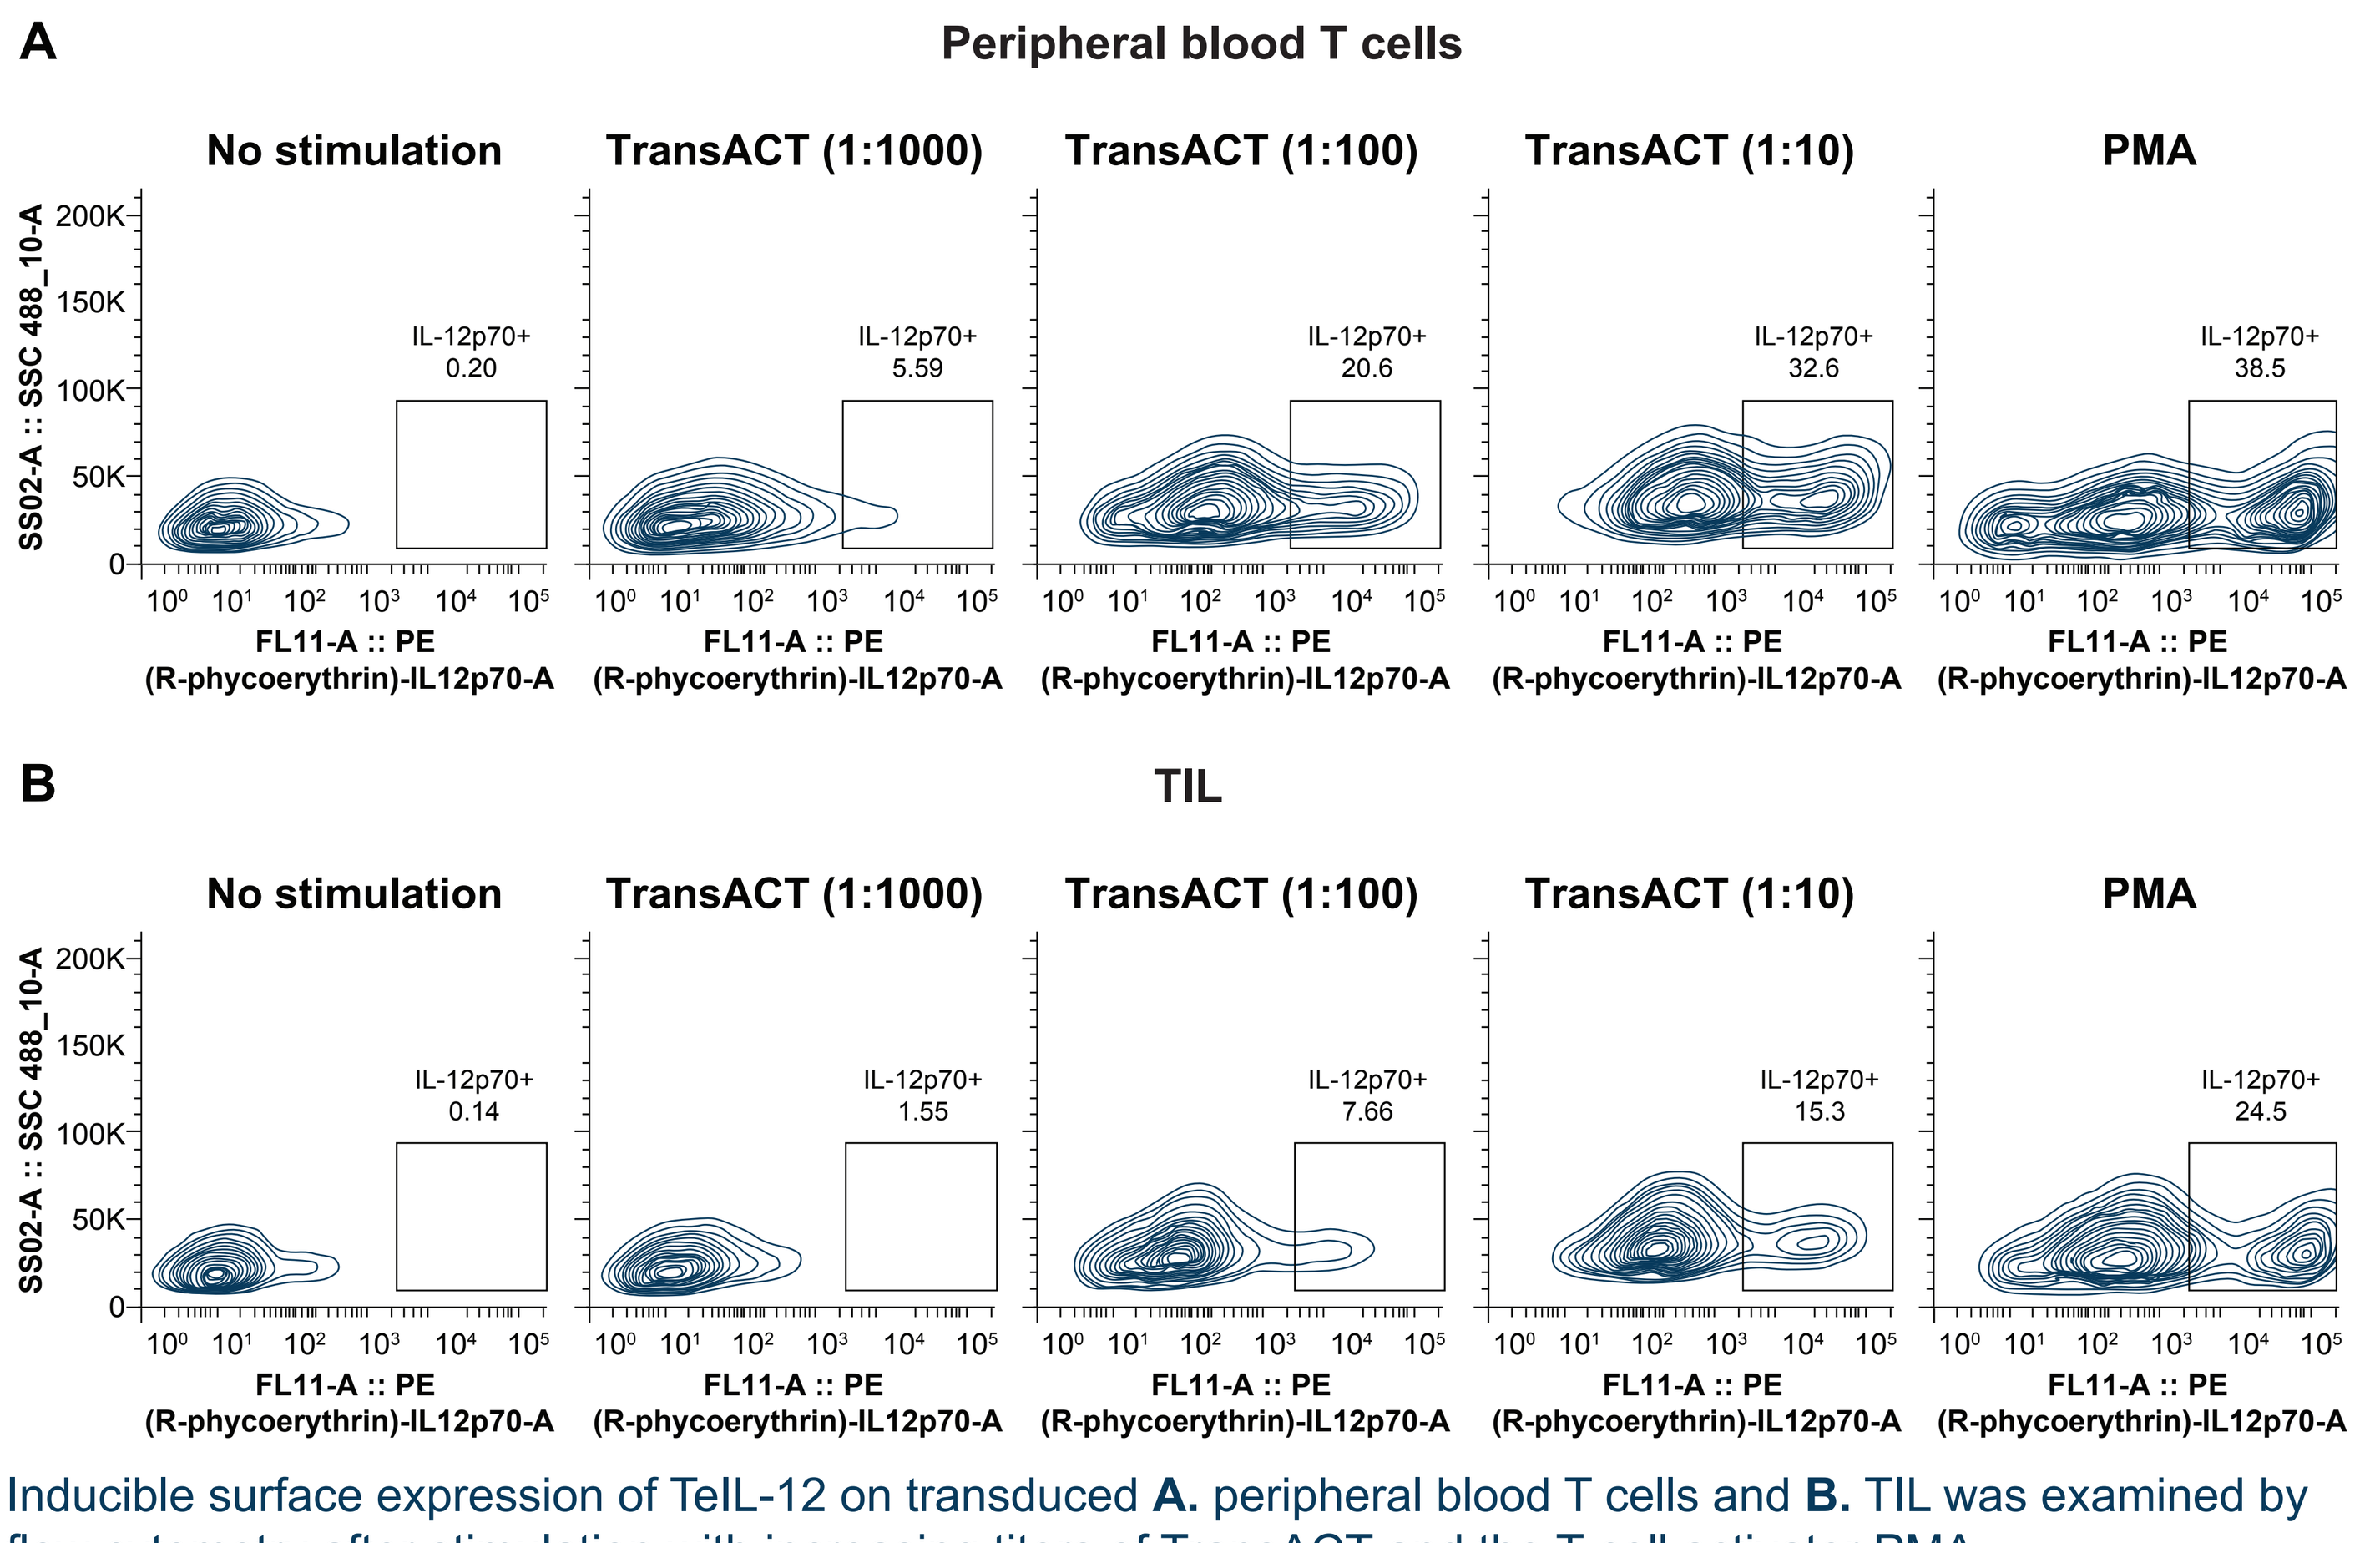
A Peripheral blood T cells
No stimulation
TransACT (1:1000)
TransACT (1:100)
TransACT (1:10)
PMA
IL-12p70+ 0.20
IL-12p70+ 5.59
IL-12p70+ 20.6
IL-12p70+ 32.6
IL-12p70+ 38.5
B TIL
IL-12p70+ 0.14
IL-12p70+ 1.55
IL-12p70+ 7.66
IL-12p70+ 15.3
IL-12p70+ 24.5
SS02-A :: SSC 488_10-A
FL11-A :: PE (R-phycoerythrin)-IL12p70-A

Inducible surface expression of TeIL-12 on transduced **A.** peripheral blood T cells and **B.** TIL was examined by flow cytometry after stimulation with increasing titers of TransACT and the T-cell activator PMA

Figure 4. Expansion, viability, and T-cell subsets in TIL product

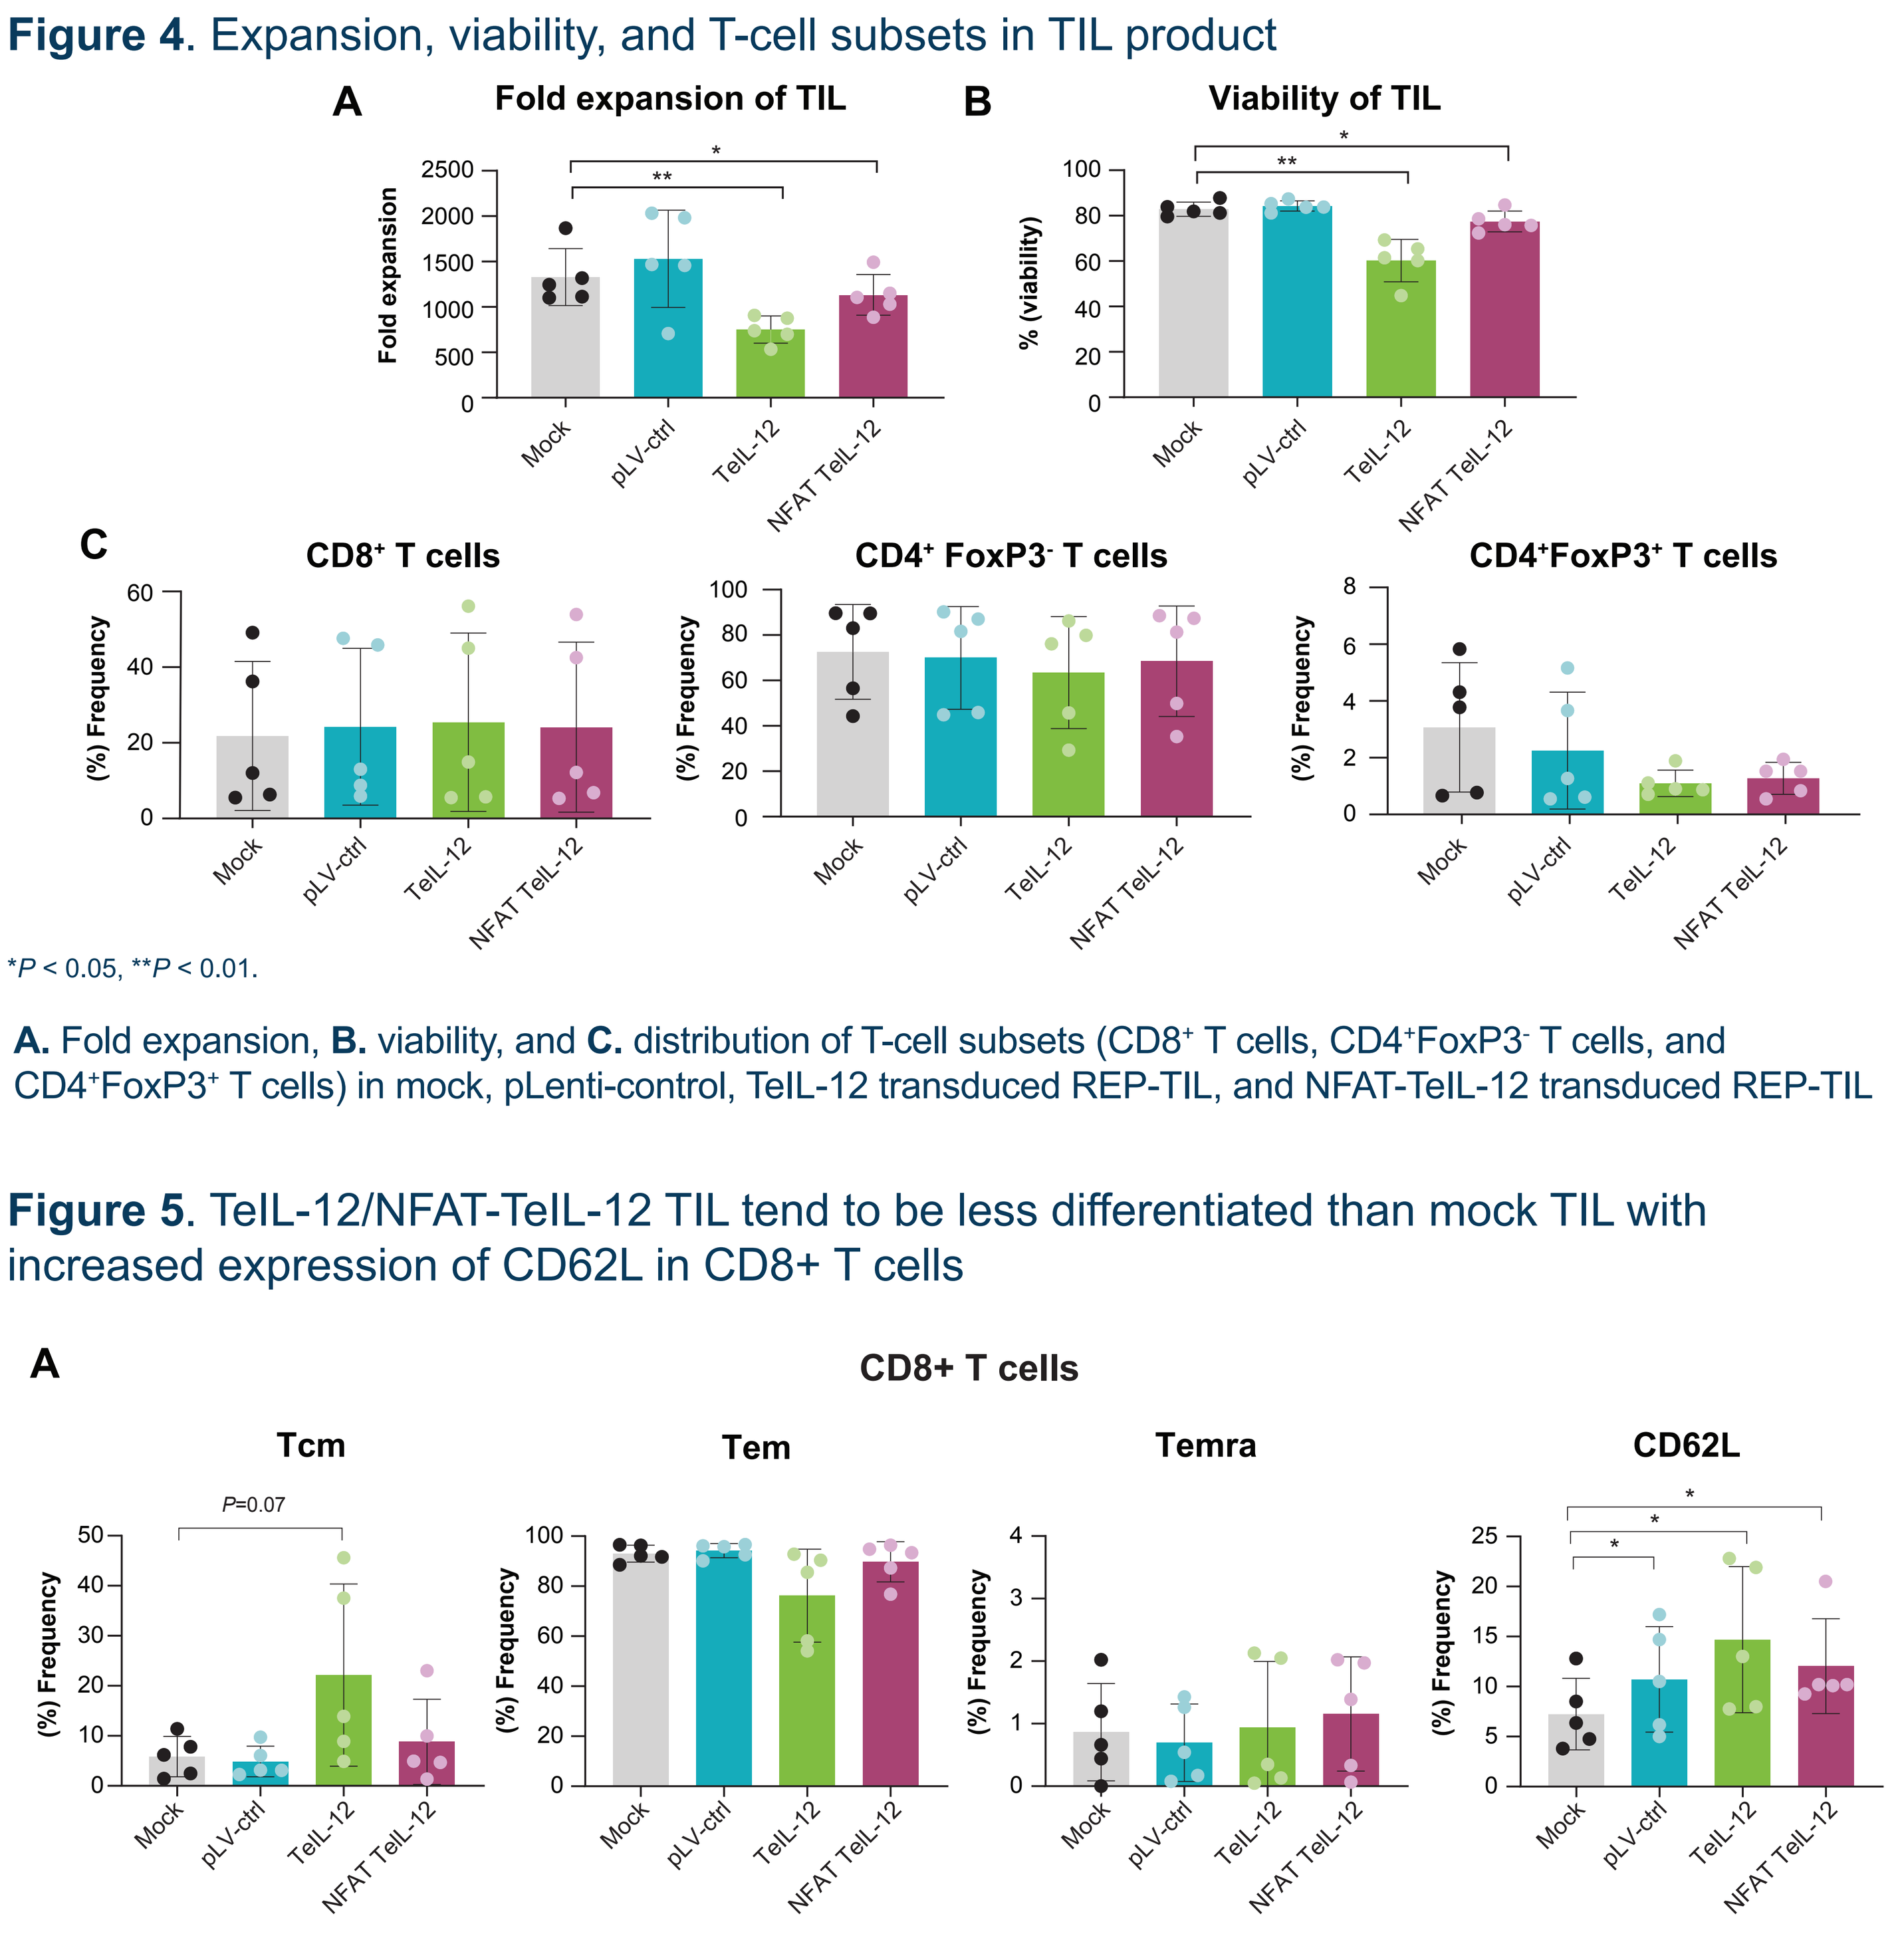
A Fold expansion of TIL
B Viability of TIL
Fold expansion
% (viability)
C CD8+ T cells
CD4+ FoxP3- T cells
CD4+FoxP3+ T cells
(%) Frequency
Mock
pLV-ctrl
TeIL-12
NFAT TeIL-12

*P < 0.05, **P < 0.01.

A. Fold expansion, **B.** viability, and **C.** distribution of T-cell subsets (CD8+ T cells, CD4+FoxP3- T cells, and CD4+FoxP3+ T cells) in mock, pLenti-control, TeIL-12 transduced REP-TIL, and NFAT-TeIL-12 transduced REP-TIL

Figure 5. TeIL-12/NFAT-TeIL-12 TIL tend to be less differentiated than mock TIL with increased expression of CD62L in CD8+ T cells

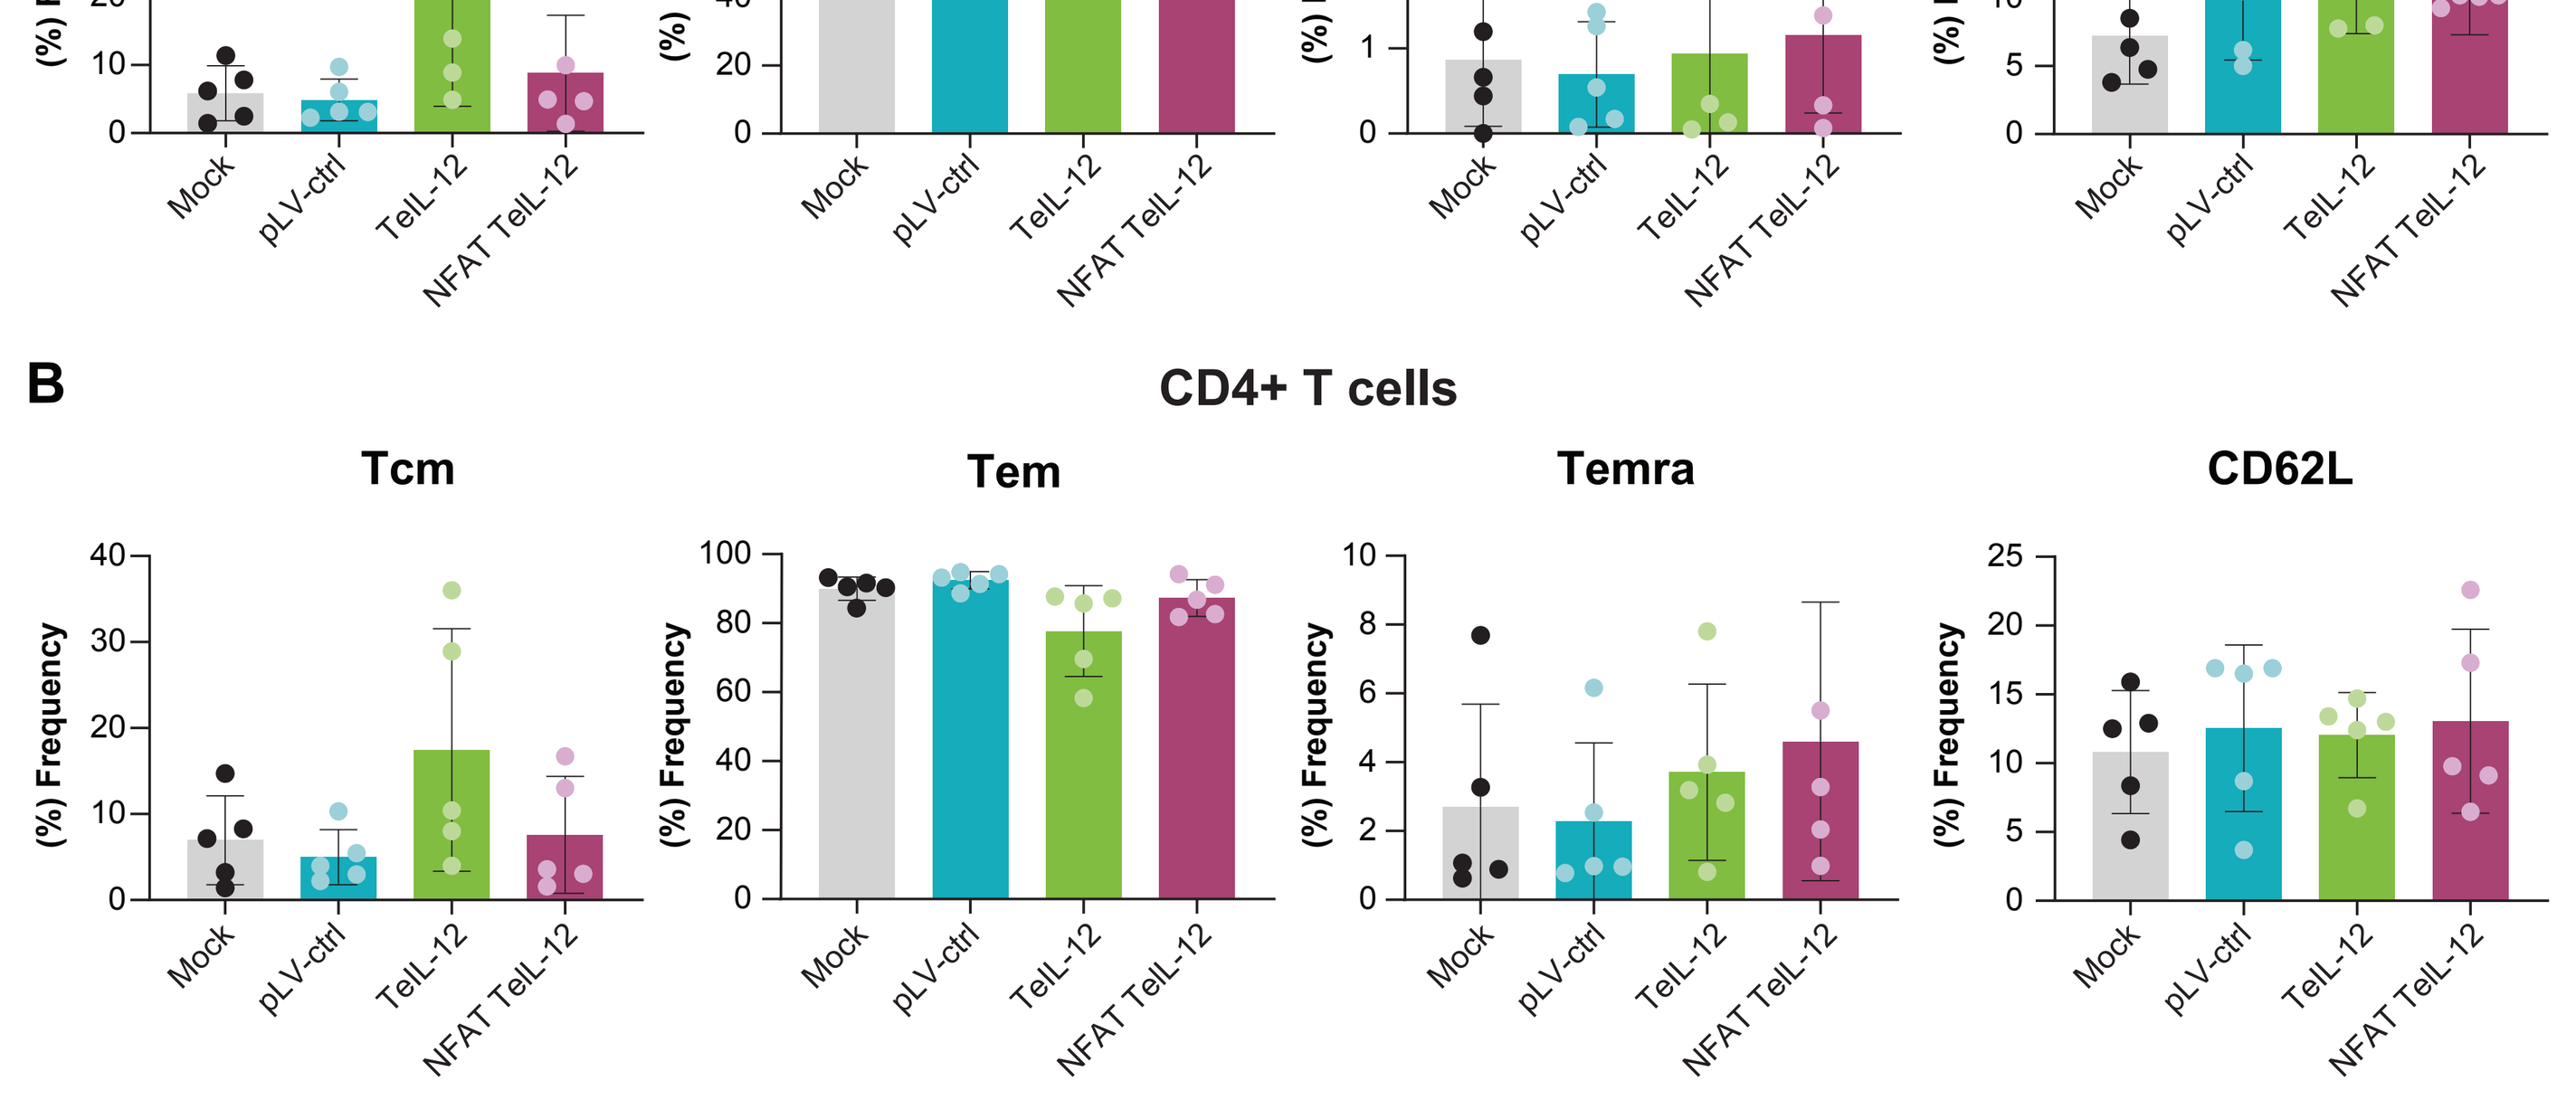
A CD8+ T cells
Tcm
Tem
Temra
CD62L
P=0.07
B CD4+ T cells
(%) Frequency
Mock
pLV-ctrl
TeIL-12
NFAT TeIL-12

*P < 0.05.

T-cell differentiation status was examined in mock, pLenti-control, TeIL-12, and NFAT-TeIL-12 transduced TIL. Frequencies of Tcm (CD45RA-CCR7+), Tem (CD45RA-CCR7-), Temra (CD45+CCR7-), and CD62L+ cells in **A.** CD8+ and **B.** CD4+ TIL are shown

Results

Figure 6. TeIL-12/NFAT-TeIL-12 TIL show reduced TIM-3 and TIGIT expression compared with mock TIL

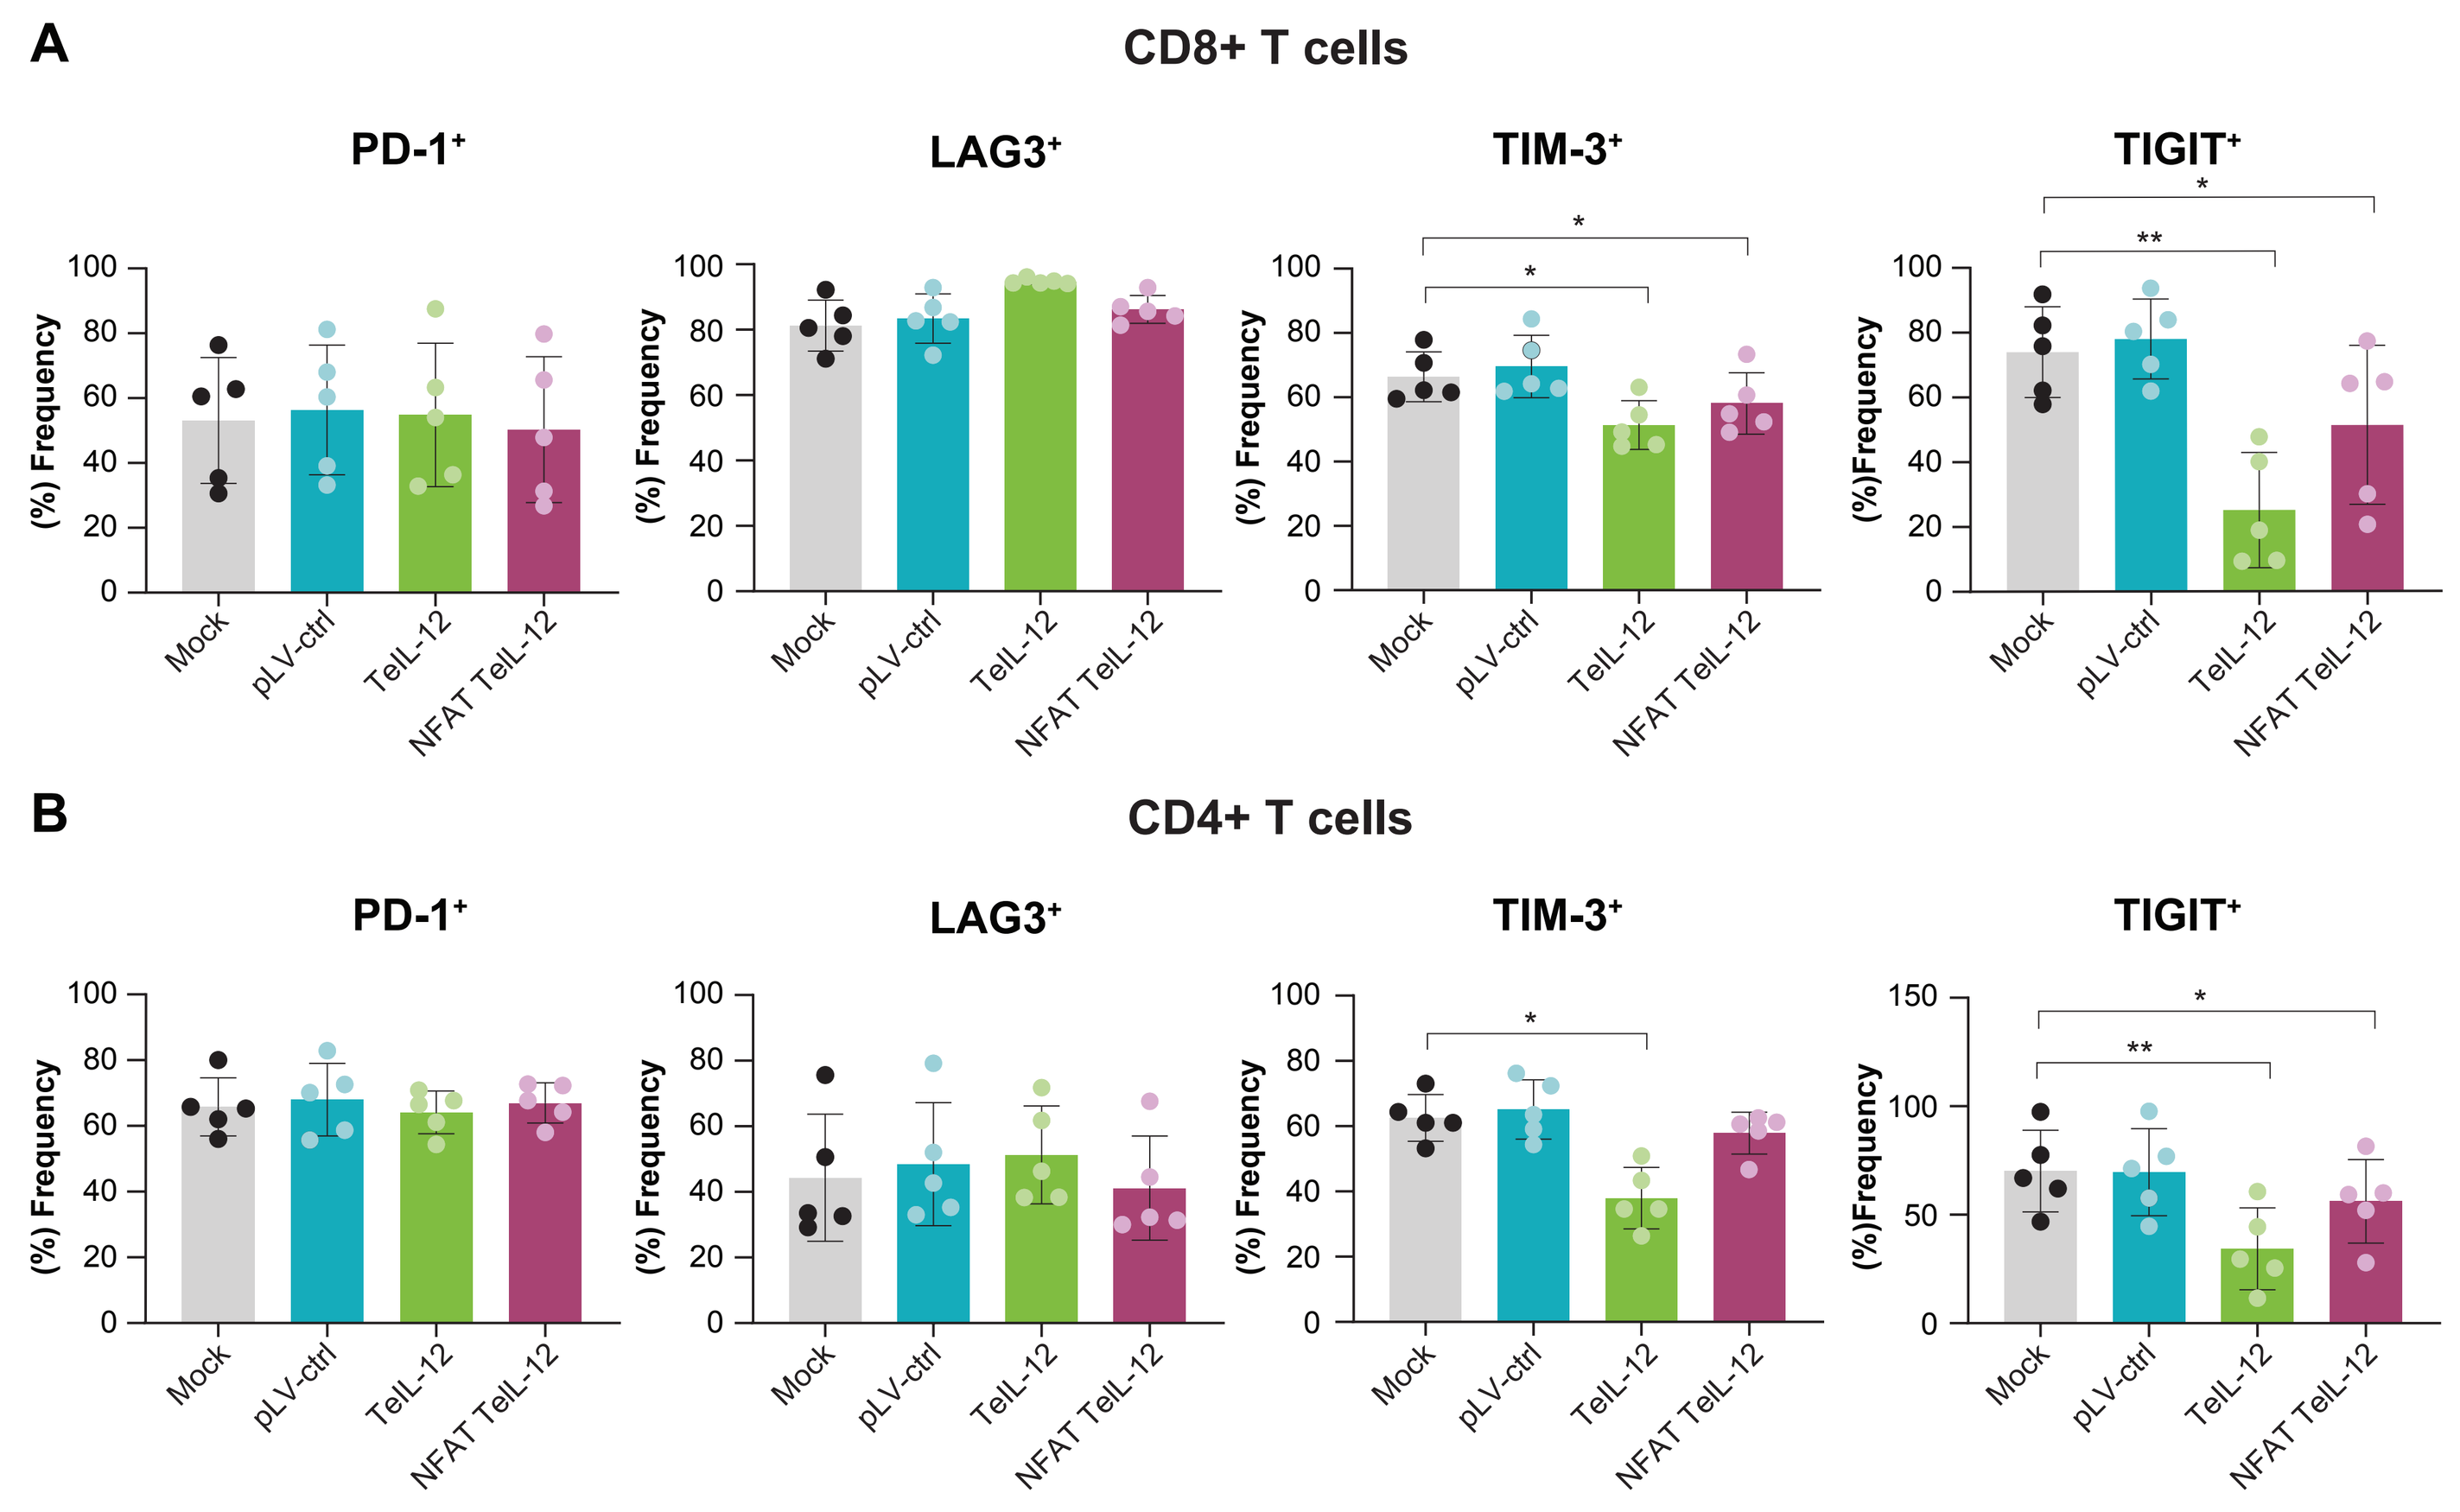
A CD8+ T cells
PD-1+
LAG3+
TIM-3+
TIGIT+
B CD4+ T cells
(%) Frequency
Mock
pLV-ctrl
TeIL-12
NFAT TeIL-12

*P < 0.05, **P < 0.01.

T-cell exhaustion status was examined in mock, pLenti-control, TeIL-12, and NFAT-TeIL-12 transduced TIL Frequencies of PD-1+, LAG3+, TIM-3+, and TIGIT+ cells in **A.** CD8+ and **B.** CD4+ TIL are shown

Figure 7. KILR-THP-1 assay shows enhanced cytotoxic activity of TeIL-12/NFAT-TeIL-12 TIL

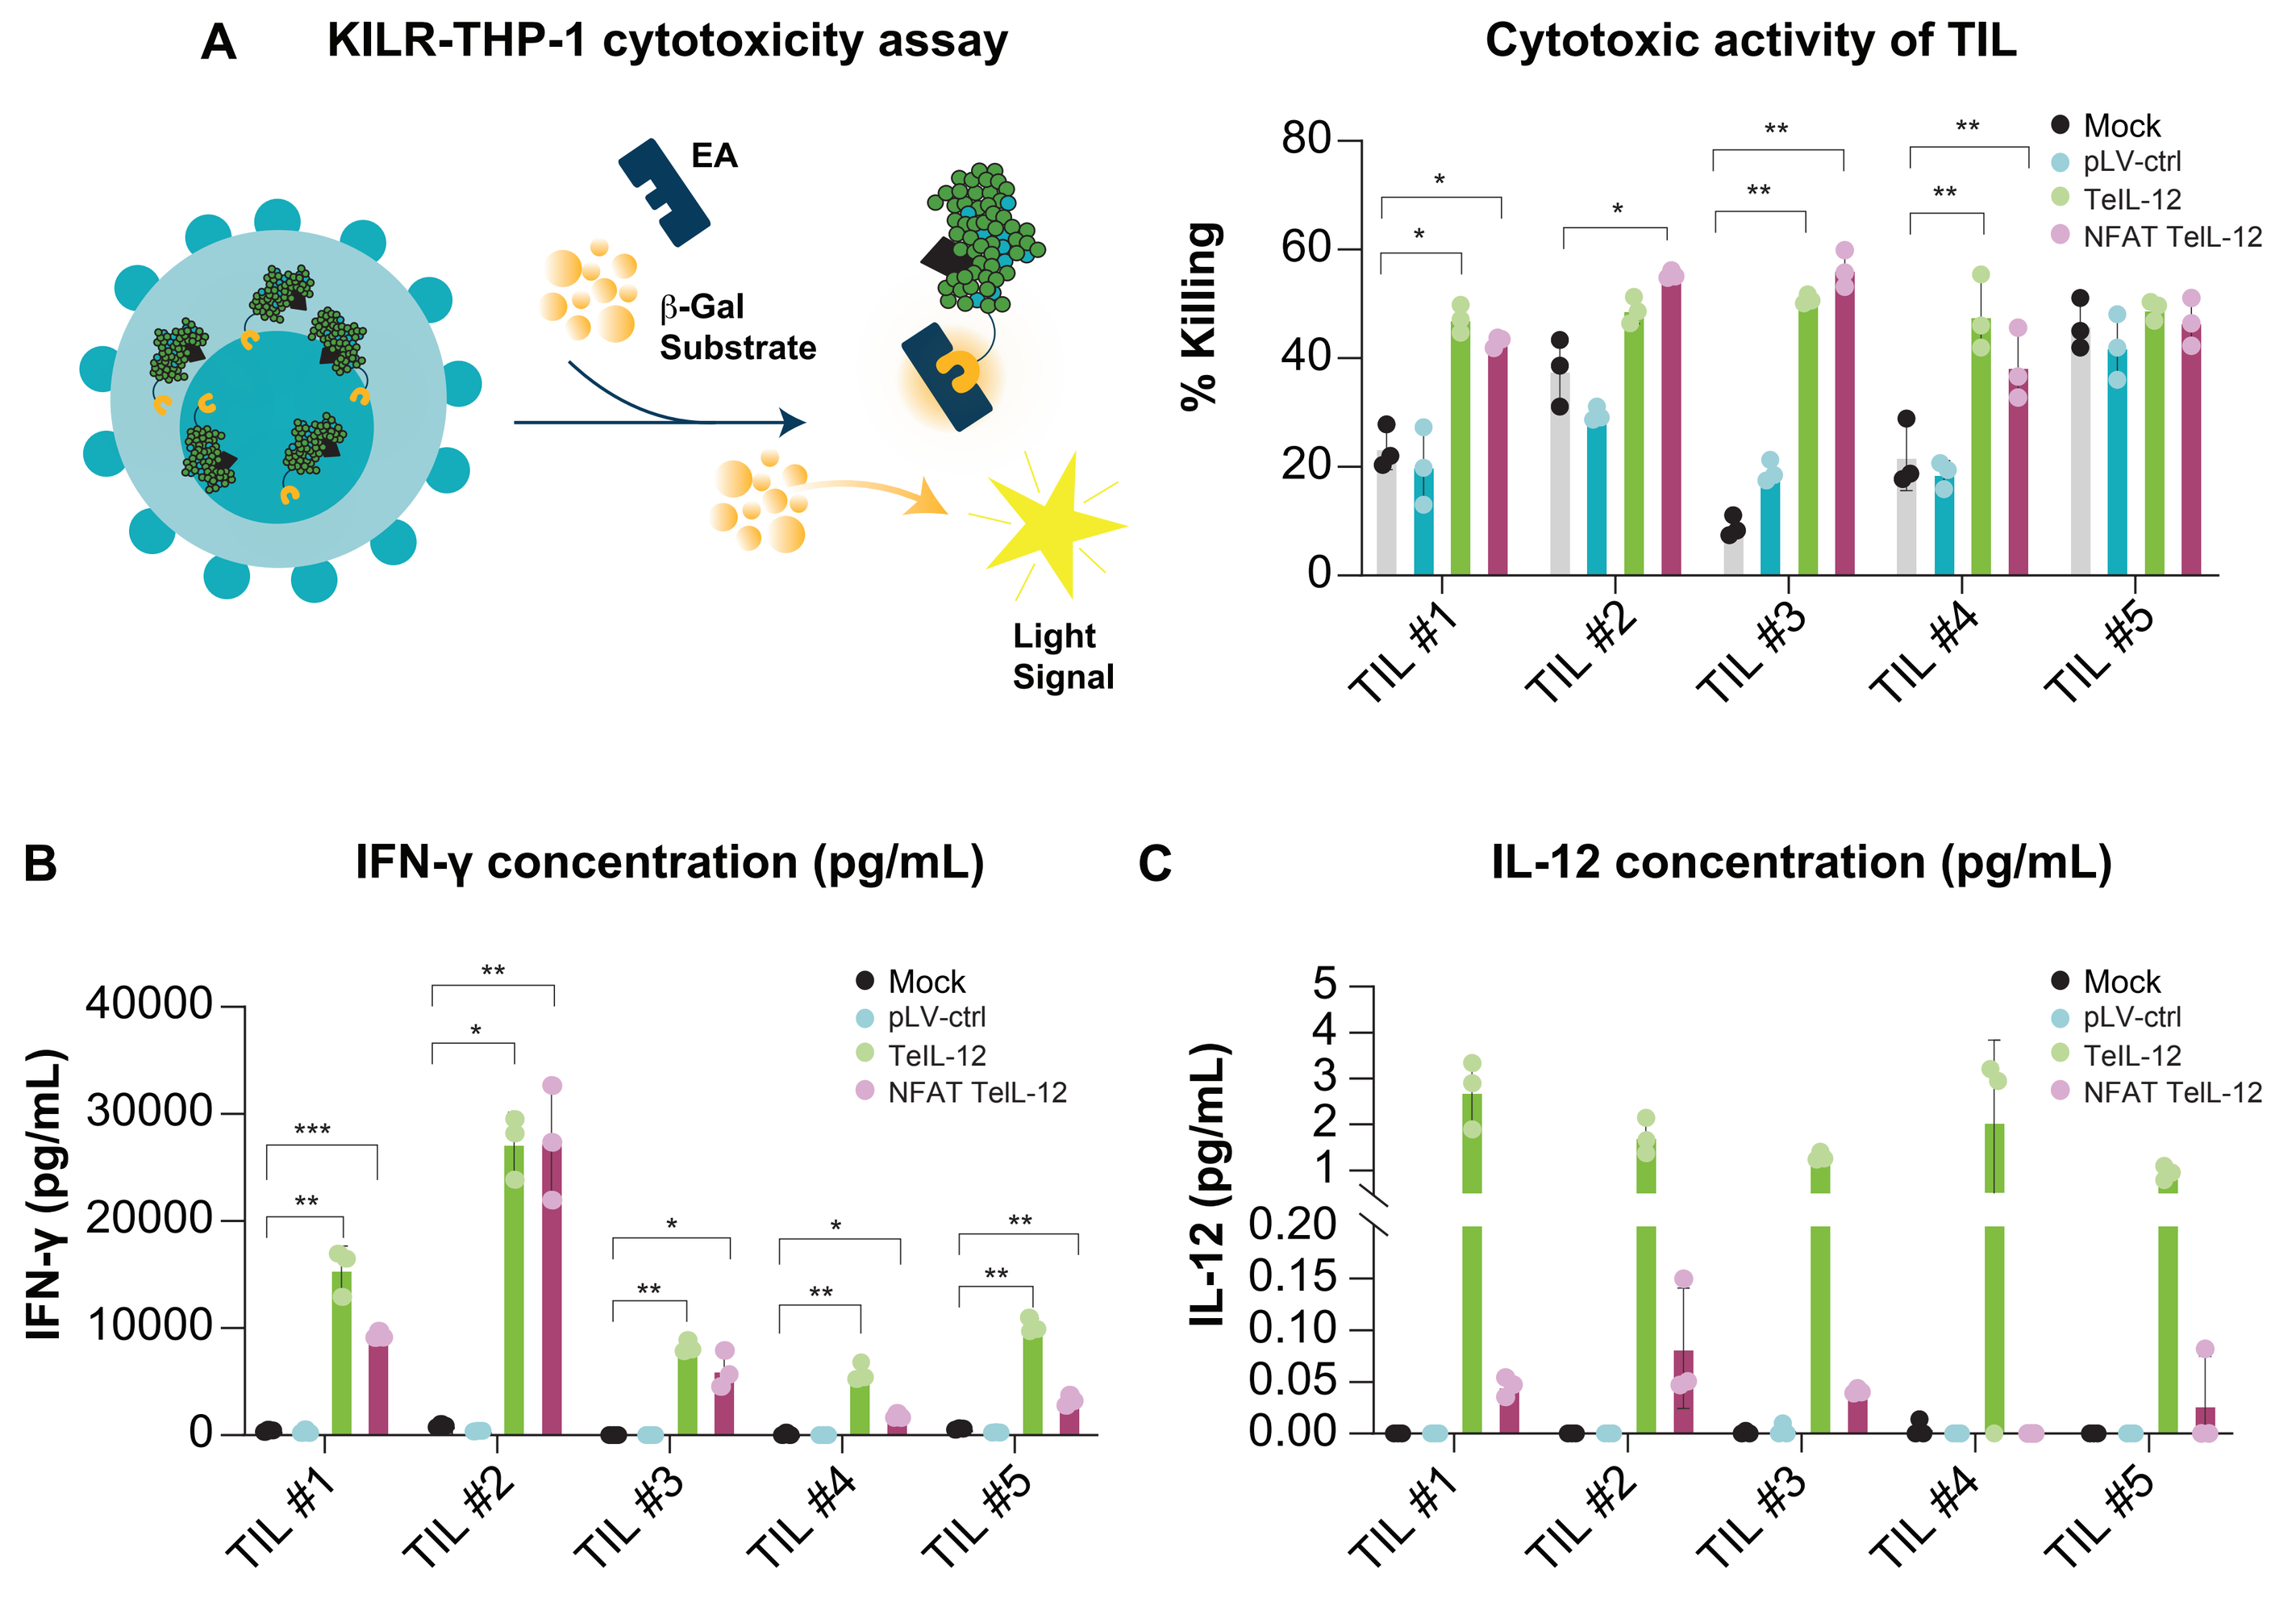
A KILR-THP-1 cytotoxicity assay
EA
β-Gal Substrate
Light Signal
Cytotoxic activity of TIL
% Killing
Mock
pLV-ctrl
TeIL-12
NFAT TeIL-12
TIL #1
TIL #2
TIL #3
TIL #4
TIL #5
B IFN-γ concentration (pg/mL)
IFN-γ (pg/mL)
C IL-12 concentration (pg/mL)
IL-12 (pg/mL)

*P < 0.05, **P < 0.01, ***P < 0.001

A. Cytotoxicity evaluation by the KILR-THP-1 assay. Cryopreserved control and genetically engineered TIL were thawed and cocultured for 24 hours with KILR-THP-1 cells (Eurofins DiscoverX, Fremont, CA, USA) at a 10:1 E:T cell ratio to measure cytotoxicity in an allogeneic setting. Concentrations of **B.** IFN-γ and **C.** soluble IL-12 in the coculture supernatant were examined by ELISA

Figure 8. xCELLigence assay shows enhanced cytotoxic activity of TeIL-12/NFAT-TeIL-12 TIL

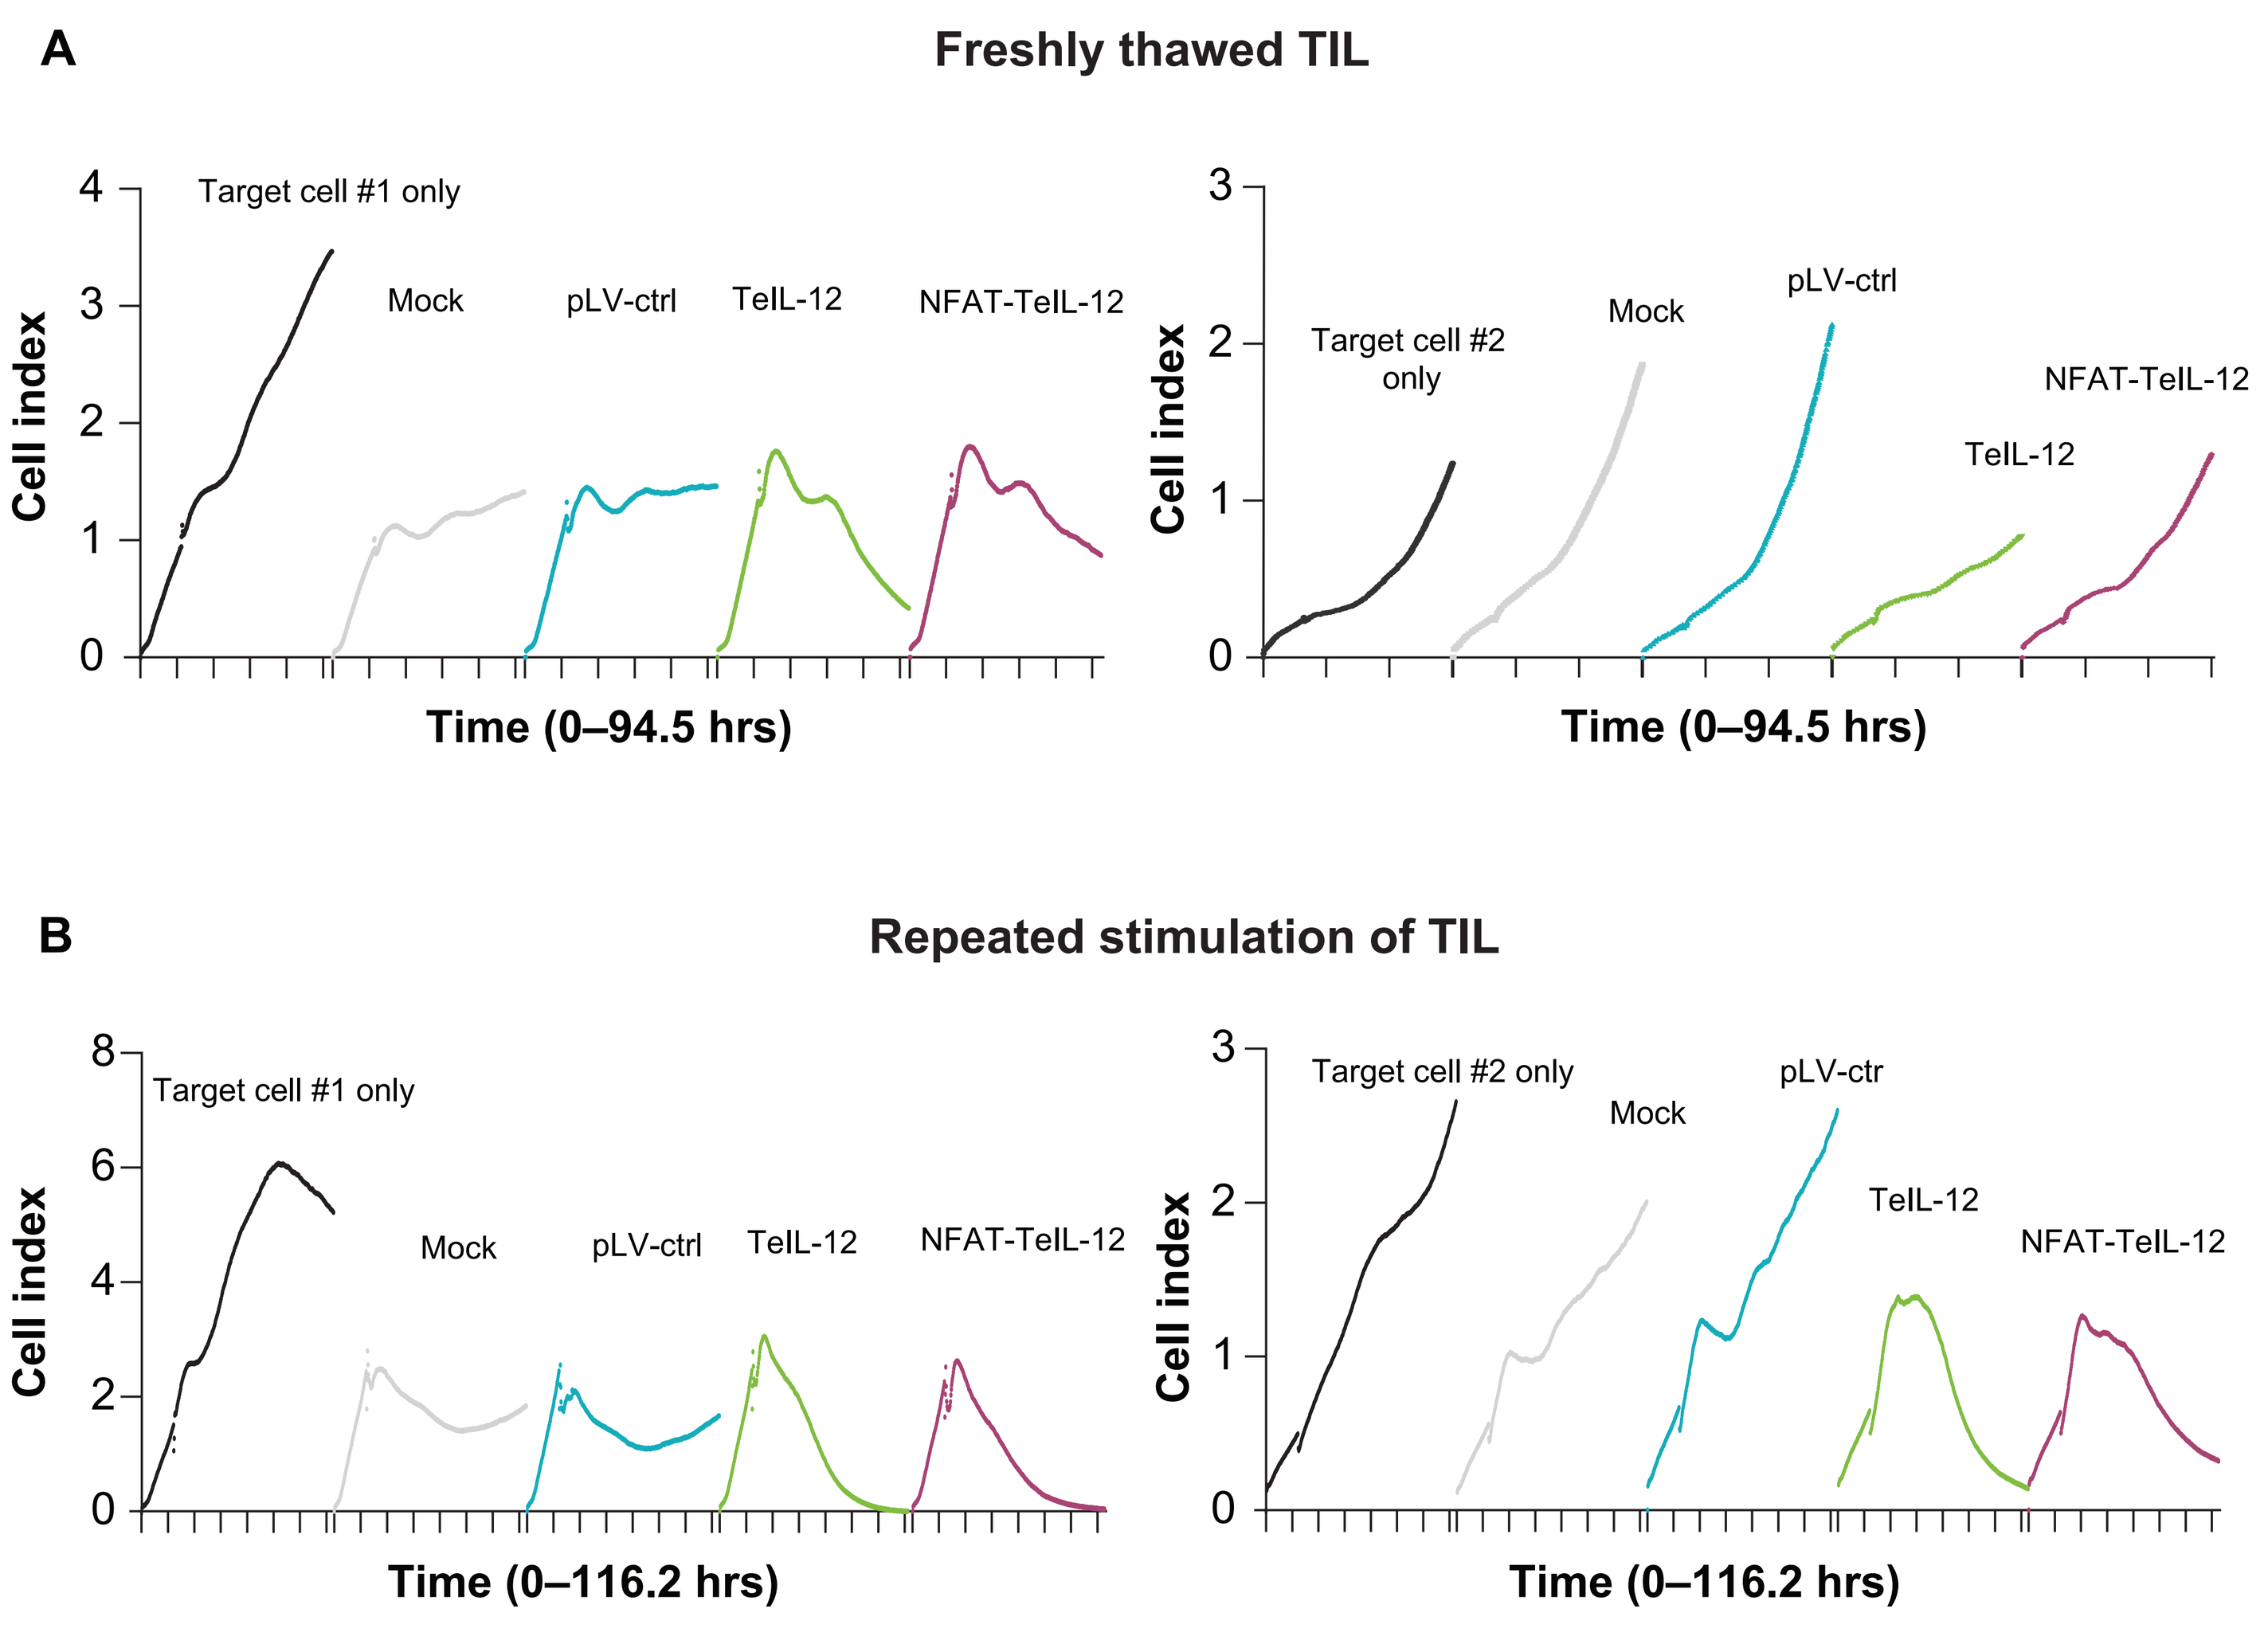
A Freshly thawed TIL
Target cell #1 only
Target cell #2 only
Mock
pLV-ctrl
TeIL-12
NFAT-TeIL-12
Cell index
Time (0–94.5 hrs)
B Repeated stimulation of TIL
pLV-ctr
Time (0–116.2 hrs)

A. Cytotoxicity assessment by the xCELLigence assay. Cryopreserved control and genetically engineered TIL were thawed and added into E-plates, pre-seeded with cells from two different tumor cell lines: Target cells #1 (left) and Target cells #2 (right), at a 3:1 E:T cell ratio to measure cytotoxicity in an allogeneic setting. **B.** To evaluate serial cytotoxic activity, TIL were stimulated every 3 days with TransACT (1:100); 1 day after repeated stimulation (× 3), TIL were washed and assessed for cytotoxic activity

Conclusions

- Both TeIL-12 and NFAT-TeIL-12 show superior *in vitro* cytotoxic activity with pronounced IFN-γ production; superior anti-tumor cytotoxicity was maintained after repeated TransACT stimulation
- Both TeIL-12 and NFAT-TeIL-12 show a favorable T cell phenotype with increased expression of CD62L and decreased expression of TIM-3 and TIGIT
- TeIL-12 shows a good safety profile with minimal shedding of IL-12; however, NFAT-TeIL-12 demonstrates the potential for a better safety profile due to less shedding of IL-12 relative to TeIL-12 in co-culture systems
- Considering the systemic toxicities observed due to secreted IL-12 in a prior NFAT-IL-12 TIL trial with a TIL product manufactured using a different process[5], a tethered IL-12 combined with a NFAT-inducible construct offers a double safety TIL design feature to ensure NFAT-TeIL-12 TIL are safe in future first-in-human clinical trials
- IL-12 expression systems were successfully incorporated into the 22-day TIL manufacturing process
- Overall, the efficacy and safety data support further clinical development of NFAT-TeIL-12 alone or in combination with other approaches to enhance TIL potency

References

1. Chesney J, et al. *J Immunother Cancer*. 2022;10:e005755.
2. Schoenfeld AJ, et al. *J Immunother Cancer*. 2021;9(suppl 2):A486. Abstract 458.
3. Granhøj JS, et al. *Expert Opin Biol Ther*. 2022;22:627–41.
4. Trinchieri G. *Nat Rev Immunol*. 2003;3:133–46.
5. Zhang L, et al. *Clin Cancer Res*. 2015;21:2278–88.
6. Zhang L, et al. *J Immunother Cancer*. 2020;8:e000210.

Abbreviations

CCR7, chemokine (C-C motif) receptor 7; ELISA, enzyme-linked immunosorbent assay; E:T, effector to target ratio; IFN-γ, interferon gamma; IL-12, interleukin 12; LAG3, lymphocyte activation gene 3; NFAT, nuclear factor of activated T cells; PMA, phorbol 12-myristate 13-acetate; PD-1, programmed cell death protein-1; REP, rapid expansion protocol; TIGIT, T-cell immunoreceptor with Ig and ITIM domains; Tcm, central memory T cells; Tem, effector memory T cells; Temra, effector memory RA T cells; TIL, tumor-infiltrating lymphocytes; TME, tumor microenvironment.

Acknowledgments

- The authors thank the patients who provided tumor tissue for this study
- This study was sponsored by Iovance Biotherapeutics, Inc. (San Carlos, CA)
- Editorial support was provided by Peloton Advantage, LLC, an OPEN Health company, and funded by Iovance

Disclosures

Yongliang Zhang: Employment: Iovance Biotherapeutics. Stock or Stock Options: Iovance Biotherapeutics.
Nathan Gilbert: Employment: Iovance Biotherapeutics. Stock or Stock Options: Iovance Biotherapeutics.
Patrick Innamarato: Employment: Iovance Biotherapeutics. Stock or Stock Options: Iovance Biotherapeutics.
Judy Fang: Employment: Iovance Biotherapeutics. Stock or Stock Options: Iovance Biotherapeutics.
Hequn Yin: Employment: Iovance Biotherapeutics. Stock or Stock Options: Iovance Biotherapeutics.

© 2023, Iovance Biotherapeutics