POSTER # 42

Successful Expansion and Characterization of Tumor Infiltrating Lymphocytes (TILs) from Non-melanoma Tumors

Jyothi Sethuraman, Joy Prewitt, Alexis Hutchinson, Laurelis Santiago, Jie Qing Chen, Zhimin Dai, Ian Frank, Seth Wardell, James Bender, Michael T Lotze

Lion Biotechnologies, Inc. 999 Skyway Road, Suite 150, San Carlos, CA 94070

Abstract

Background

Adoptive cell therapy (ACT) has shown promise in comparison to other methods of cancer immunotherapy that rely on the active development of anti-tumor T-cells *in vivo* to mediate cancer regression[1]. Administration of autologous TILs in melanoma patients have shown an overall response of 56% at NCI and variable response rates at multiple sites[1]. The durable responses observed in melanoma and cervical patients using ACT has encouraged us to explore broadening their utility in multiple solid tumor settings. Here, we demonstrate the feasibility of growing TILs and to develop TIL therapies for other solid tumors.

Results

To date, we have been able to successfully culture TILs from tumors obtained from bladder, cervical, head and neck, lung, and Triple Negative Breast Cancer (TNBC). The average yield of TILs from pre-REP is listed in **Table 1.** Phenotypic characterization of TILs from bladder, cervical, and lung cancer were >60-70% CD8+ T-cells whereas TILs from head and neck demonstrated variable distribution of CD8+ and CD4+ T-cells. TILs propagated from TNBC were >80% CD4+ T-cells. Regardless of the tumors, most cultures had <20% CD56+ NK-cells.

Table 1. Average yield of TILs obtained from pre-REP in non-melanoma tumors

Tumor	Number of patient tumors	Average yield (and range) of TILs from pre-REP (10^6)
Bladder	2	290 (97-600)
Cervical	4	360 (147-800)
H&N	7	539 (132-738)
Lung	8	688 (50-915)
TNBC	13	429 (81-665)

Methods

Wash tumor in HBSS

↓

Dice into 2-3 mm^3 fragments

↓

Place tumor fragments in G-REX 10 cell culture flasks with medium containing serum and IL-2

↓

Exchange media on D7 and every 4-5 days from D11 until D21

↓

Assess cell count, viability and phenotyping followed by cryopreservation for future purpose

Figure 1. Successful expansion of TILs from non-melanoma tumors

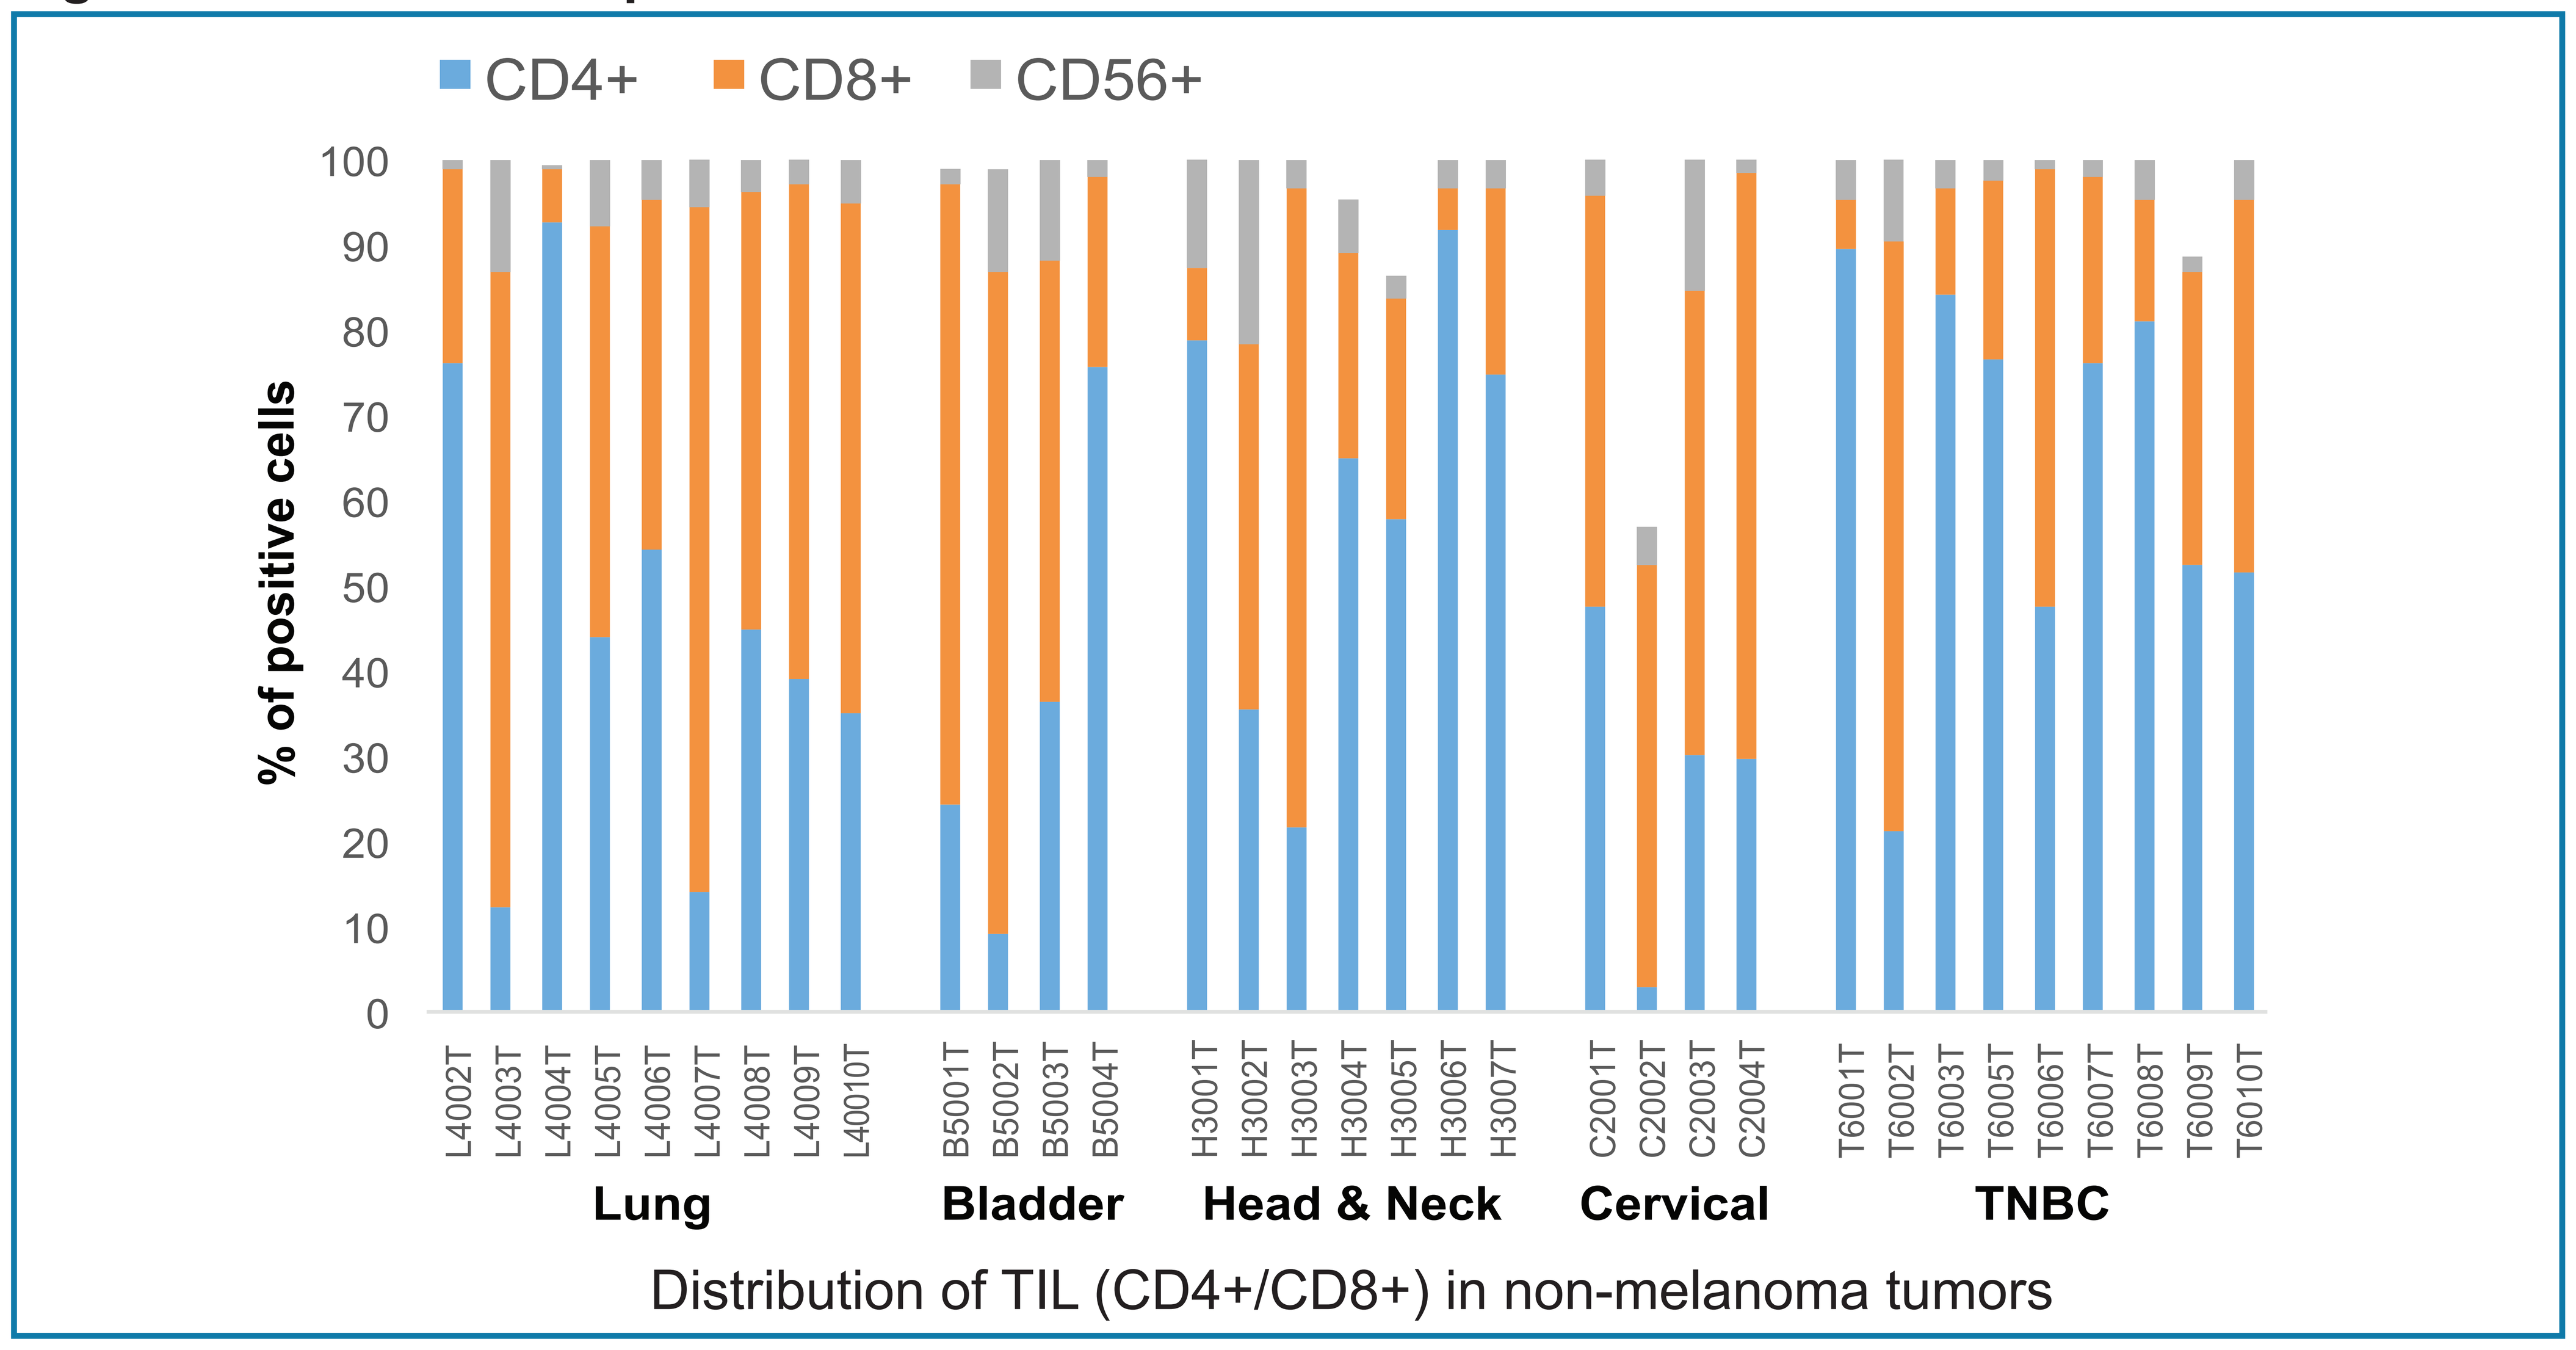
CD4+
CD8+
CD56+
% of positive cells
L4002T L4003T L4004T L4005T L4006T L4007T L4008T L4009T L40010T
B5001T B5002T B5003T B5004T
H3001T H3002T H3003T H3004T H3005T H3006T H3007T
C2001T C2002T C2003T C2004T
T6001T T6002T T6003T T6005T T6006T T6007T T6008T T6009T T6010T
Lung
Bladder
Head & Neck
Cervical
TNBC
Distribution of TIL (CD4+/CD8+) in non-melanoma tumors

Figure 2. Non-melanoma TILs expressed CD27 and CD28, consistent with young TILs

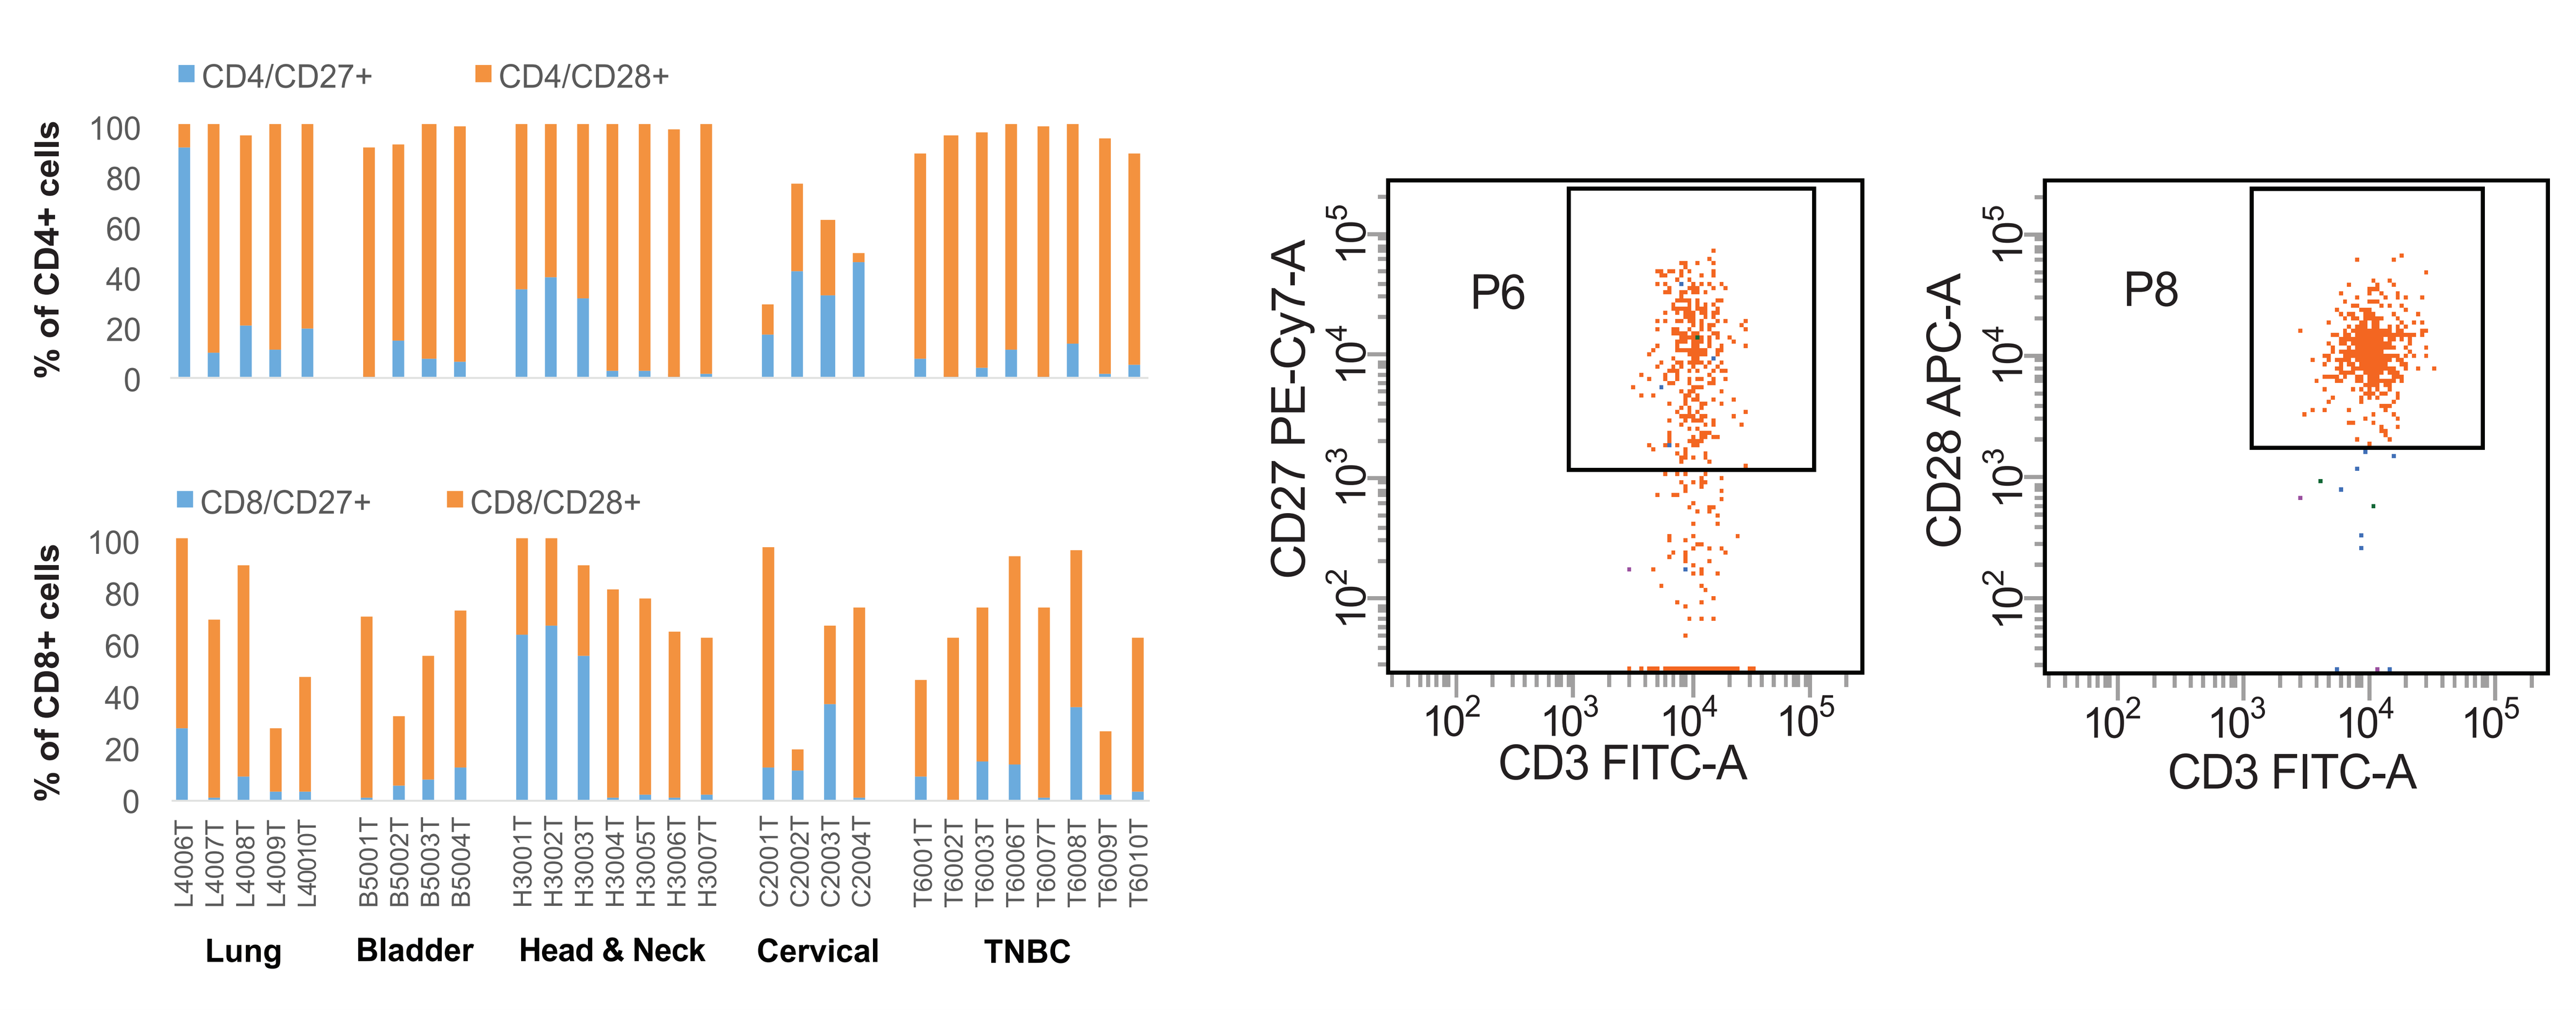
CD4/CD27+
CD4/CD28+
% of CD4+ cells
CD8/CD27+
CD8/CD28+
% of CD8+ cells
Lung
Bladder
Head & Neck
Cervical
TNBC
P6
CD27 PE-Cy7-A
CD3 FITC-A
P8
CD28 APC-A
CD3 FITC-A

Figure 3. Activated TILs skew towards effector memory population

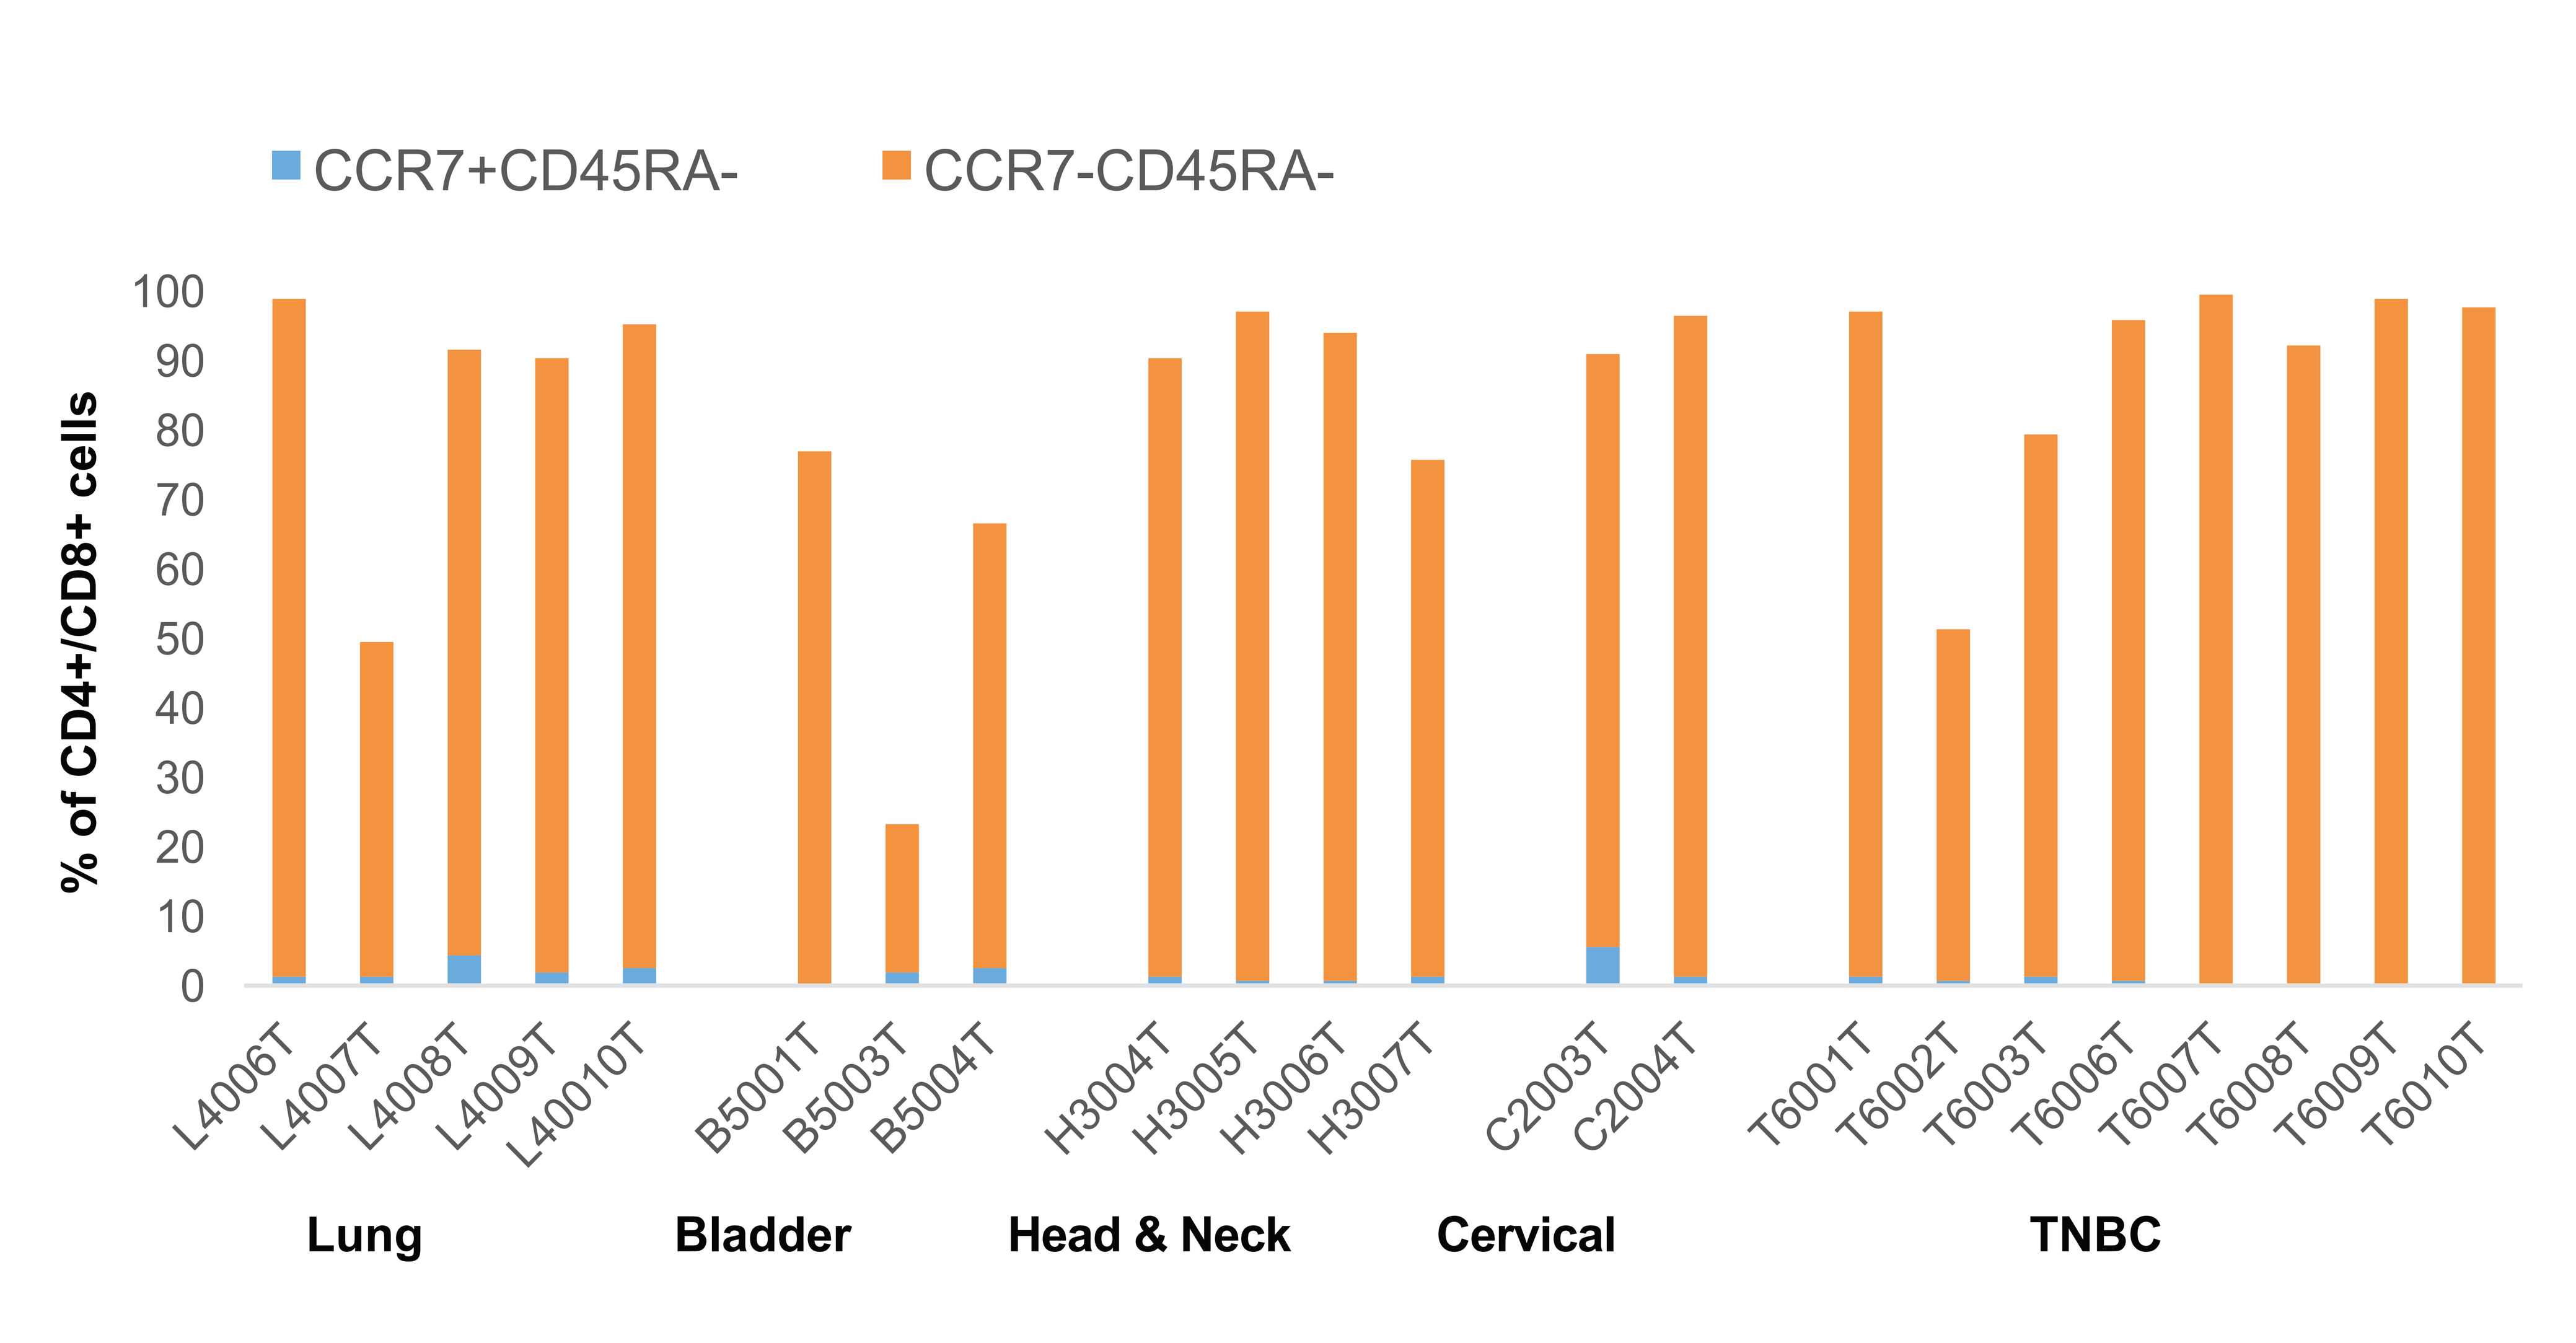
CCR7+CD45RA-
CCR7-CD45RA-
% of CD4+/CD8+ cells
L4006T L4007T L4008T L4009T L40010T
B5001T B5003T B5004T
H3004T H3005T H3006T H3007T
C2003T C2004T
T6001T T6002T T6003T T6006T T6007T T6008T T6009T T6010T
Lung
Bladder
Head & Neck
Cervical
TNBC

Conclusions

- We have been able to successfully grow TILs from lung, bladder, head and neck, cervical, and TNBC patient tumors.
- Lung, bladder, and cervical tumors showed greater proportion of CD8+ TILs, whereas head and neck and TNBC tumors were mostly CD4+ TILs.
- Further characterization of CD4+ and CD8+ TILs demonstrated effector memory phenotypic cells that were also CD27+ and CD28+.

Reference

1. Rosenberg SA and Restifo NP: Adoptive cell transfer as personalized immunotherapy for human cancer. *Science.* 2015, 348:62-8.

Disclosure and Funding Statement

This study was funded by Lion Biotechnologies, Inc. JS, JP, AH, LS, JQC, ZD, IF, SW, JB, and MTL are employees of Lion Biotechnologies, Inc. and have stock options.

Acknowledgment

All listed authors meet the criteria for authorship set forth by the International Committee for Medical Journal Editors. Colleagues from the Moffitt Hospital providing several of these tumors is gratefully acknowledged. Graphic services were provided by AOIC, LLC, and were funded by Lion Biotechnologies, Inc.

Presented at Society for Immunotherapy of Cancer in National Harbor, Maryland, USA, November 9-13, 2016